Abstract TPS2613
2015 ASCO
Chicago, IL

A phase 1, dose-escalation, safety, pharmacokinetic, pharmacodynamic study of thioureidobutyronitrile, a novel p53 targeted therapy, in patients with advanced solid tumors

Geoffrey I. Shapiro[a], James W. Mier[b], John F. Hilton[a], Leena Gandhi[a], Nicole G. Chau[a], Andrea J. Bullock[b], Jeffrey G. Supko[c], Sigitas Jonas Verselis[a], Kayla Murgo[a], Cameron Sze[a], Susan Gotthardt[b], Andrew Wolanski[a], W. James Alexander[d], Ashok Kumar[d], Sylvia A. Holden[d], Karima Chafai-Fadela[d], Siya Ram[d], Krishna Menon[d]

[a]Dana-Farber Cancer Institute, Boston, MA; [b]Beth Israel Deaconess Medical Center, Boston, MA; [c]Massachusetts General Hospital, Boston, MA; [d]Cellceutix Corporation, Beverly, MA

INTRODUCTION

Background: Thioureidobutyronitrile, Kevetrin, induced apoptosis in wild type p53, mutant p53 and p53 null cell lines (*Shapiro 2013*). In A549 lung carcinoma cells, wild type p53 was stabilized by Kevetrin. Kevetrin induced non-genotoxic activation of the p53 signaling pathway (*Kumar 2012*). Kevetrin also induced p21 and PUMA, known transcriptional targets of p53 (*Kumar 2011*). Kevetrin caused accumulation of monoubiquitinated p53 and induced transcriptional independent apoptosis. In p53 mutant breast carcinoma cells (MDA-MB-231), Kevetrin induced degradation of hyperstable oncogenic mutant p53 and induced apoptotic cell death (*Kumar 2011*). Apoptotic cell death was also induced in K-562, a p53 null CML cell line. Consistent with *in vitro* data, Kevetrin showed potent antitumor activity in wild type p53 (A549), mutant p53 (MDA-MB-231), and p53 null (K-562) human tumor xenograft models (*Chafai-Fadela 2010*). Kevetrin has the unique ability to target both wild type and mutant p53 tumors controlling tumor growth in various preclinical tumor models (*Kumar 2012, Shapiro 2013*).

Based on the pre-clinical data, a Phase I study was initiated with Kevetrin for solid carcinomas at Dana-Farber/Harvard Cancer Center in 2012. Participating sites include Dana Farber Cancer Institute and Beth Israel Deaconess Medical Center.

Methods: Adults with refractory locally advanced or metastatic solid tumors, acceptable liver, kidney function, and hematologic status were eligible. Objectives include determination of DLT, MTD, pharmacokinetics, pharmacodynamics, and evaluating preliminary evidence of antitumor activity.

Kevetrin is given as an intravenous infusion once weekly for 3 weeks in a 28-day cycle. The starting dose was 10 mg/m^2. In a 3+3 design, groups of 3-6 subjects are evaluated for toxicity at each dose level. Dose escalation is based upon the number and intensity of adverse events in cycle 1. Kevetrin PK is characterized for the first and last doses given in cycle 1.

Kevetrin induced increases in p21 levels in lymphocytes in nonclinical studies *(Kumar 2011)*; therefore p21 expression in peripheral blood mononuclear cells is measured as a pharmacodynamic biomarker. Antitumor activity by RECIST 1.1 criteria and serum tumor markers are assessed. The p53 status of tumors of selected subjects is determined.

Phase 1 study CTIX-0000

Dana-Farber/Harvard Cancer Center

ClinicalTrials.gov Identifier: NCT01664000

Sites:
- Dana-Farber Cancer Institute
- Beth Israel Deaconess Medical Center

Objectives:
- Evaluate the safety, tolerability, maximum tolerated dose (MTD)
- Determine recommended Phase 2 dose
- Characterize pharmacokinetic (PK) profiles
- Evaluate preliminary evidence of antitumor activity
- Explore potential biomarkers of tumor response

Study Design:
- Intravenous infusion once weekly for 3 weeks in a 28-day cycle
- Starting dose was 10 mg/m^2
- Dose escalation in a 3+3 design, groups of 3-6 subjects are evaluated for toxicity at each dose level
- Dose escalation is based upon the number and intensity of adverse events in Cycle 1
- Once the MTD is established, up to 12 additional subjects may be enrolled at the MTD dose level
- PK is characterized for the first and last doses given in Cycle 1
- Antitumor activity by RECIST 1.1 criteria and serum tumor markers is assessed
- p53 status of tumors of selected subjects will be determined
- The total number of subjects planned is 60

Pharmacodynamic Biomarker:
- p21 expression in peripheral blood mononuclear cells

Key Eligibility Criteria:
- Adults with refractory locally advanced or metastatic solid tumors
- Acceptable liver and kidney function, and hematological status

Phase 1 study CTIX-0000

STATUS:

The Phase 1 trial began November 7, 2012

A total of 40 subjects have been enrolled to date, 34 have completed

Completed: Cohort 0 (10 mg/m^2) through Cohort 9 (450 mg/m^2)

Cohort 10: 750 mg/m^2 is ongoing

1 DLT occurred to date

The MTD has not yet been reached

SUBJECTS:

Average age: 63 ± 9 years

Age range: 42 to 85 years

Females / Males: 26 / 14

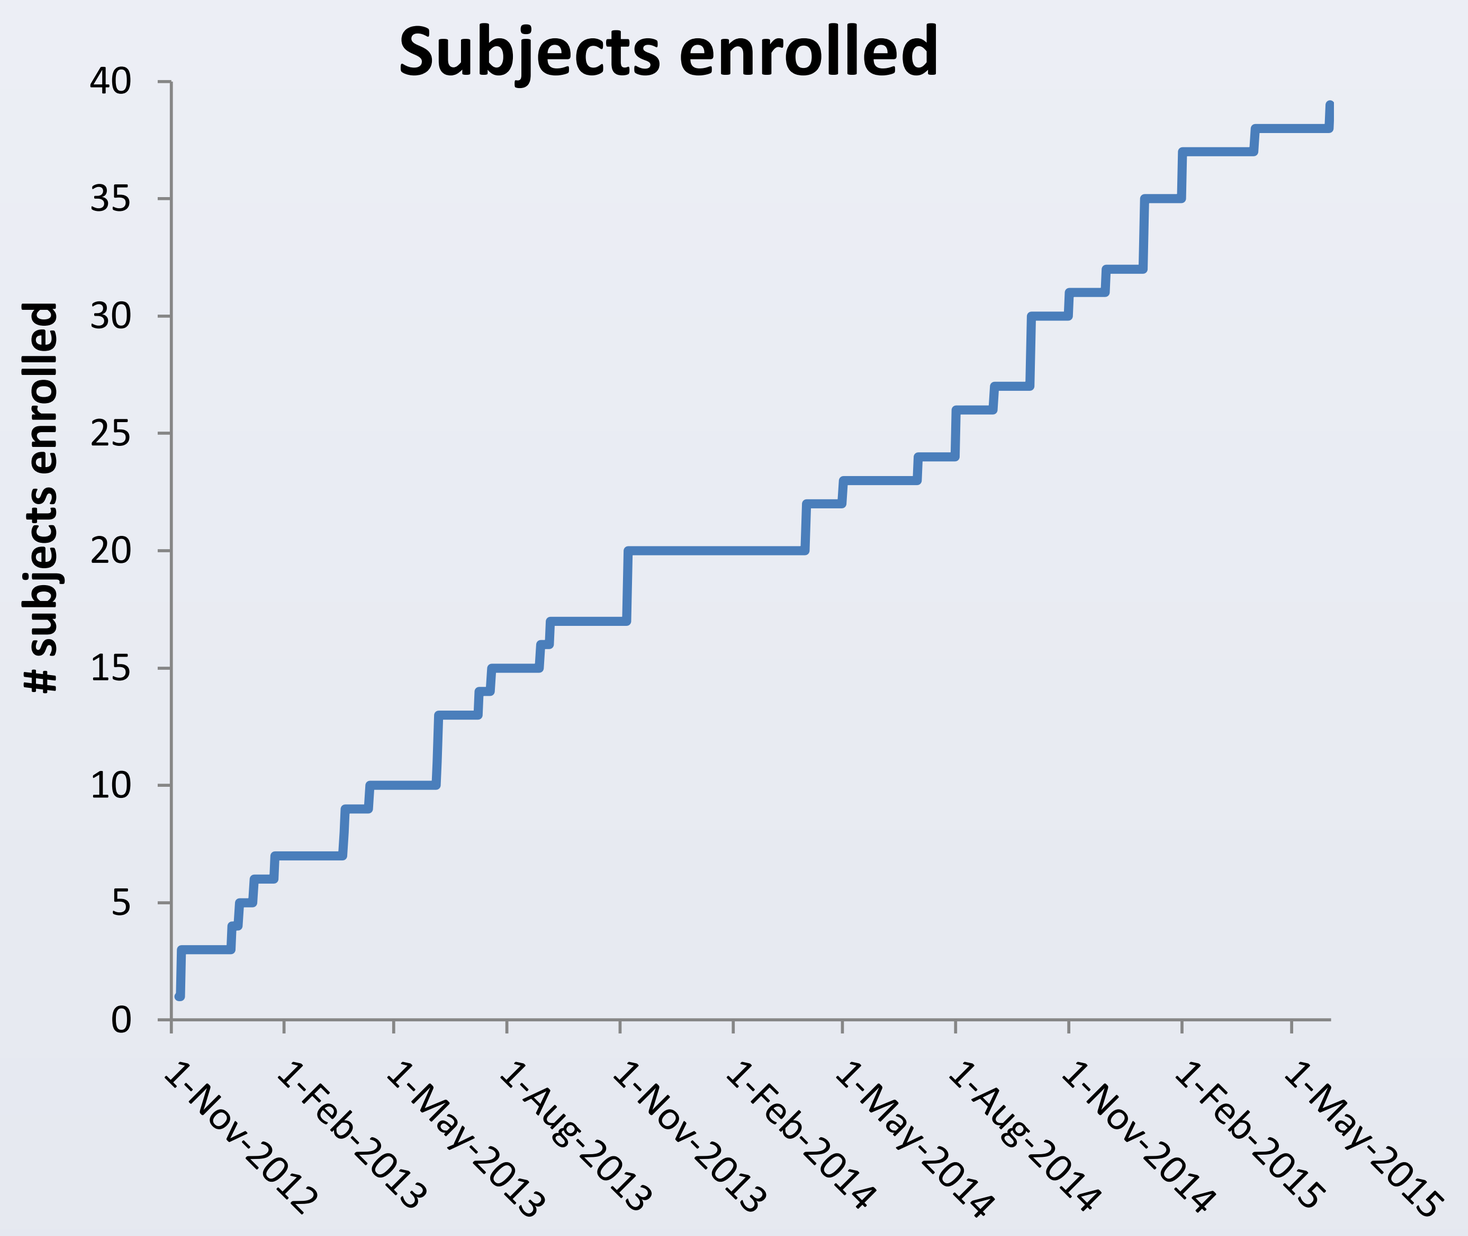
Subjects enrolled
subjects enrolled
40
35
30
25
20
15
10
5
0
1-Nov-2012
1-Feb-2013
1-May-2013
1-Aug-2013
1-Nov-2013
1-Feb-2014
1-May-2014
1-Aug-2014
1-Nov-2014
1-Feb-2015
1-May-2015

Cancer types of participating subjects

17	gynecological cancers
6	head & neck cancers
5	lung cancers
4	renal /urothelial / bladder cancers
3	skin cancers
2	prostate cancers
2	pancreatic / biliary cancers
1	liposarcoma

Cohort	Dose (mg/m^2)	Subject	Age	Sex	Cancer Type
9	450	**132**	74	female	ovarian
4	75	**114**	66	female	ovarian
3	50	**111**	59	female	ovarian
8	350	**126**	57	female	ovarian
0	10	**103**	79	female	ovarian stromal
8	350	**206**	58	female	metastatic ovarian
1	20	**104**	73	female	metastatic ovarian
8	350	**124**	59	female	papillary serous ovarian
8	350	**125**	57	female	endometrial
8	350	**130**	65	female	endometrial
8	350	**131**	66	female	endometrial
7	215	**123**	61	female	endometrial
1	20	**106**	71	female	metastatic endometrial
4	75	**115**	47	female	cervical
5	110	**116**	61	female	metastatic cervical
5	110	**117**	76	female	cervical mesonephric tumor
8	350	**127**	63	female	metastatic mullerian serous adeno
9	450	**207**	69	female	SCLC
1	20	**201**	69	female	SCLC
7	215	**122**	67	male	NSCLC
6	165	**204**	73	male	NSCLC
10	750	**208**	63	male	metastatic pulmonary basal cell
0	10	**102**	53	male	metastatic squamous cell carcinoma of the head and neck
9	450	**133**	54	male	laryngeal adenoid cystic
6	165	**119**	42	male	parotid myoepithelial
5	110	**118**	58	male	parotid
6	165	**120**	54	female	metastatic adenoid cystic ca of trachea
8	350	**129**	48	male	thymoma
3	50	**110**	62	female	metastatic renal cell carcinoma
3	50	**203**	52	female	urothelial
3	50	**112**	68	male	urothelial
2	30	**108**	62	male	metastatic bladder
4	75	**113**	59	male	prostate
2	30	**107**	56	male	prostate
7	215	**121**	85	male	atypical intradermal spindle cell neoplasm of scalp
1	20	**105**	75	female	metastatic melanoma
0	10	**101**	45	female	metastatic squamous cell carcinoma of the skin
2	30	**202**	69	female	metastic pancreatic
7	215	**205**	63	male	cholangiocarcinoma
3	50	**109**	62	female	metastatic liposarcoma

Biomarker

Kevetrin induces cell cycle arrest and apoptosis through activation and stabilization of wild type p53, resulting in increased expression of target genes p21 and PUMA *(Kumar 2011)*.

In a preclinical mouse model, Kevetrin induced p21 in lymphocytes *(Kumar 2011)*; therefore p21 expression in peripheral blood mononuclear cells is measured at 7 and 24 hours after initiation of dosing as a pharmacodynamic biomarker in this trial.

Of 40 subjects enrolled to date, 31 were evaluable for changes in p21 biomarker (pre-dose and at least 1 time point post-dose blood sample obtained after first dose of Kevetrin).

Of the 31 evaluable subjects :

68 % had an increase in p21 expression in a range of 3% to 205%

48 % had an increase in p21 expression of ≥ 10%

These results suggest that Kevetrin activates p53 by inducing p21 gene expression.

SUMMARY

The Phase 1 study with Kevetrin, CTIX-0000, is in progress at Dana-Farber/Harvard Cancer Center in subjects with various solid carcinomas; the majority of which are gynecological cancers.

10 cohorts of subjects have been completed; the 11th cohort is ongoing. Only 1 DLT has been observed to date, but the MTD has not yet been reached. The current dose is 750 mg/m^2, 75-fold greater than the starting dose.

Kevetrin was shown to activate wild type p53 and degrade mutant p53. Since Kevetrin activates both transcriptional-dependent and transcriptional-independent pathways to promote apoptosis through wild type p53 activation and degrades oncogenic mutant p53, Kevetrin can function as a major inducer of apoptosis in many types of tumors independent of p53 mutation status.

In this Phase 1 study, the biomarker, p21 expression levels in peripheral blood, were increased in 68% of subjects and 48% had an increase in p21 expression at a level of ≥10%. These results suggest that Kevetrin activates p53 by inducing p21 gene expression.

REFERENCES

Chafai-Fadela, Hiran T, Badger S, Holden SA, Brackett S, Menon K. Novel small molecule has potent antitumor activity in drug-resistant human carcinoma xenograft models. *Cancer Research* (2010) 70:LB-283.

Kumar A, Hiran T, Holden SA, Chafai-Fadela K, Rogers S, Ram S, Menon K. Kevetrin, a novel small molecule, activates p53, enhances expression of p21, induces cell cycle arrest and apoptosis in a human cancer cell line. *Cancer Research* (2011) 71:4470.

Kumar A, Holden SA, Chafai-Fadela K, Ram S, Menon K. Kevetrin targets both MDM2-p53 and Rb-E2F pathways in tumor suppression. *Cancer Research* (2012) 72:2874

Shapiro G, Supko JG, Cho D, Hilton J, Hadfield M, Pruitt-Thompson S, Bordoli-Trachsel E, Zvereva N, Wolanski A, Sato-DiLorenzo A, Gotthardt Sm Fox EA, Verselis SJ, Kumar A, Holden SA, Ram S, Chafai-Fadela D, Harding K, Menon K. A phase 1, dose-escalation, safety, pharmacokinetic, pharmaco-dynamic study of thioureidobutyronitrile, a novel p53 targeted therapy, in patients with advanced solid tumors. *American Society for Clinical Oncology* (2013) Abstract #113150.

For further information

Please contact Cellceutix Corporation at 978-236-8717 or info@cellceutix.com
More information on this and related projects can be obtained at www.cellceutix.com

RESEARCH POSTER PRESENTATION DESIGN © 2012
www.PosterPresentations.com