#514: Phase I Study of Pazopanib in Combination with the Investigational Hypoxia-targeted Drug TH-302

Duke Cancer Institute
A National Cancer Institute-designated Comprehensive Cancer Center

Kellen L. Meadows, Paula H. Lee, Richard F. Riedel, Michael A. Morse, Hope E. Uronis, Gerard C. Blobe, Daniel J. George, Jeffrey Crawford, Herbert I. Hurwitz

Duke Cancer Institute, Duke University, Durham, North Carolina, USA

Introduction

- Preclinical data demonstrate anti-VEGF agents can induce hypoxia, which may mediate resistance and induce increased tumor invasiveness.[1]
- TH-302 is an investigational hypoxia-targeted drug. Under markedly hypoxic conditions, TH-302 releases the DNA alkylator bromo-isophosphoramide mustard (Br-IPM).[2,3]
- In several preclinical models, TH-302 plus an anti-VEGF therapy has greater anti-tumor activity than either treatment alone and this activity was associated with anti-VEGF induced hypoxia.[4]
- In this study, the multi-kinase VEGF inhibitor, pazopanib (Paz) is evaluated in combination with TH-302 in a phase I dose-escalation study followed by an expanded cohort to better define the safety, tolerability and preliminary activity of the combination.

Objectives

Primary

- Define the maximal tolerated dose (MTD) and the recommended phase II dose (RPTD) for pazopanib plus TH-302 in patients with advanced solid tumors.

Secondary

- To describe any dose-limiting and non dose-limiting toxicities of this drug regimen.

Materials & Methods

Trial Design

Stage 1: Dose escalation: cohort size 3-6

Stage 2: Expansion cohort at MTD/RPTD, approximately 16-18

- 28-day cycle

Table 1. Dosing Schema

Cohort	TH-302 IV, Days 1, 8 15 mg/m²	Pazopanib p.o., daily mg
1	340 mg/m²	800 mg
2	480 mg/m²	800 mg

Assessments

- Adverse events (AEs) are graded according to the NCI Common Toxicity Criteria version 4.0.
- Efficacy [stable disease (SD), partial response (PR)] is assessed every 2 cycles with computed tomography (CT) using Response Evaluation Criteria in Solid Tumors (RECIST1.1).

Eligibility

Key inclusion criteria:

- Histologically confirmed solid tumor malignancy for which standard therapy or palliative measures do not exist or are no longer effective.
- Disease measurable by RECIST.
- Age ≥18 years.
- Karnofsky performance status >70%.
- Life expectancy of at least 3 months.
- Adequate organ and marrow function.

Key exclusion criteria:

- Previous hypersensitivity reactions to pazopanib and/or a structural compound, biological agent, or formulation.
- Proteinuria at screening demonstrated by urine analysis (UA) > 1+ or 24 hour urine protein ≥ 1 gram/24 hours.
- Severely impaired lung function (e.g. use of home O_2, history of Idiopathic Lung Disease (ILD), any evidence of ILD on scan.
- Poorly controlled hypertension [defined as systolic blood pressure of >140 mmHg or diastolic blood pressure of >90 mmHg].
- History or abdominal fistula or perforation, impairment of gastrointestinal function or gastrointestinal disease within 6 months of study drug.
- Significant or poorly controlled cardiovascular disease or vascular disease within 6 months of study drug.
- History of significant bleeding with 6 months of study drug.
- Concomitant use of CYP3A4 inducers, strong inhibitors or substrates with a narrow therapeutic window.
- Use or need for full dose anticoagulation other than low molecular weight heparin.
- Serious, non-healing wound, active ulcer, or untreated bone fracture as judged by treating physician.

DLT definitions

- Hematologic toxicity: Any grade 4 neutropenia, thrombocytopenia or anemia or grade ≥ 3 neutropenia or thrombocytopenia lasting over 7 days.
- Any grade 3 thrombocytopenia associated with bleeding.
- Neutropenic fever.
- Nausea, vomiting or diarrhea grade ≥3 and lasting ≥ 4 days despite adequate supportive measures.
- Grade ≥ 3 ALT or AST elevation > 7 days.
- Other non-hematologic toxicity grade ≥3, excluding alopecia, anorexia, fatigue, hypertension, isolated lab abnormalities (not clinically significant) and rare, idiosyncratic reactions to any of the study drugs. Anorexia, fatigue and hypertension will be considered as DLT only if they reach grade 4 or are considered unmanageable.
- Treatment delay of ≥ 14 days for cycle 2 due to unresolved toxicity.
- Any treatment-related death or treatment-related hospitalization.

Results

As of September 26, 2013 :

Enrollment is complete at 30; 13 in dose escalation, 17 in the expanded cohort

Table 2. Baseline Characteristics

Characteristic	Patients (n = 30)
Median age, years (range)	56, (26 – 79)
Female:male, no. (%)	9 (30): 21 (70)
Median number of prior treatments	1 (range 0-8)
Type of primary tumor, no. (%)	
Sarcoma (solitary fibrous tumor, osteosarcoma, chondrosarcoma, pleomorphic sarcoma, leiomyosarcoma, Ewing's sarcoma, clear cell sarcoma, liposarcoma)	12 (40)
Colorectal	4 (13)
Bladder/Urothelial/Renal	4 (13)
Neuroendocrine	3 (10)
NSCLC	2 (7)
Pancreatic (adenocarcinoma)	2 (7)
Other*	3 (10)

*Other includes: gastroesophageal junction, ovarian, hurthle cell

Determination of MTD/RPTD

13 treated, 11 evaluable for DLT toxicity

Table 3. DLT per cohort

Cohort	Patients	DLT Toxicity
1 (TH-302 @ 340 mg/m²)	7*	• Grade 3 vaginitis
2 (TH302 @480 mg/m²)	6*	• Grade 3 neutropenia/thrombocytopenia • Grade 2 intolerable esophagitis†

*One patient in each cohort was inevaluable for DLT due to intercurrent illness.

† Patient had two dose holds and one dose reduction in cycle one.

Efficacy

- 25 have at least one restaging scan; 3 came off for toxicity before restaging, 1 clinically progressed and 1 has not yet been restaged.
- Response rate for restaged patients: 12% PR, 64% SD and 24% PD.
- 24% experienced disease control for ≥ 6 months.

Figure 1. Duration of disease control

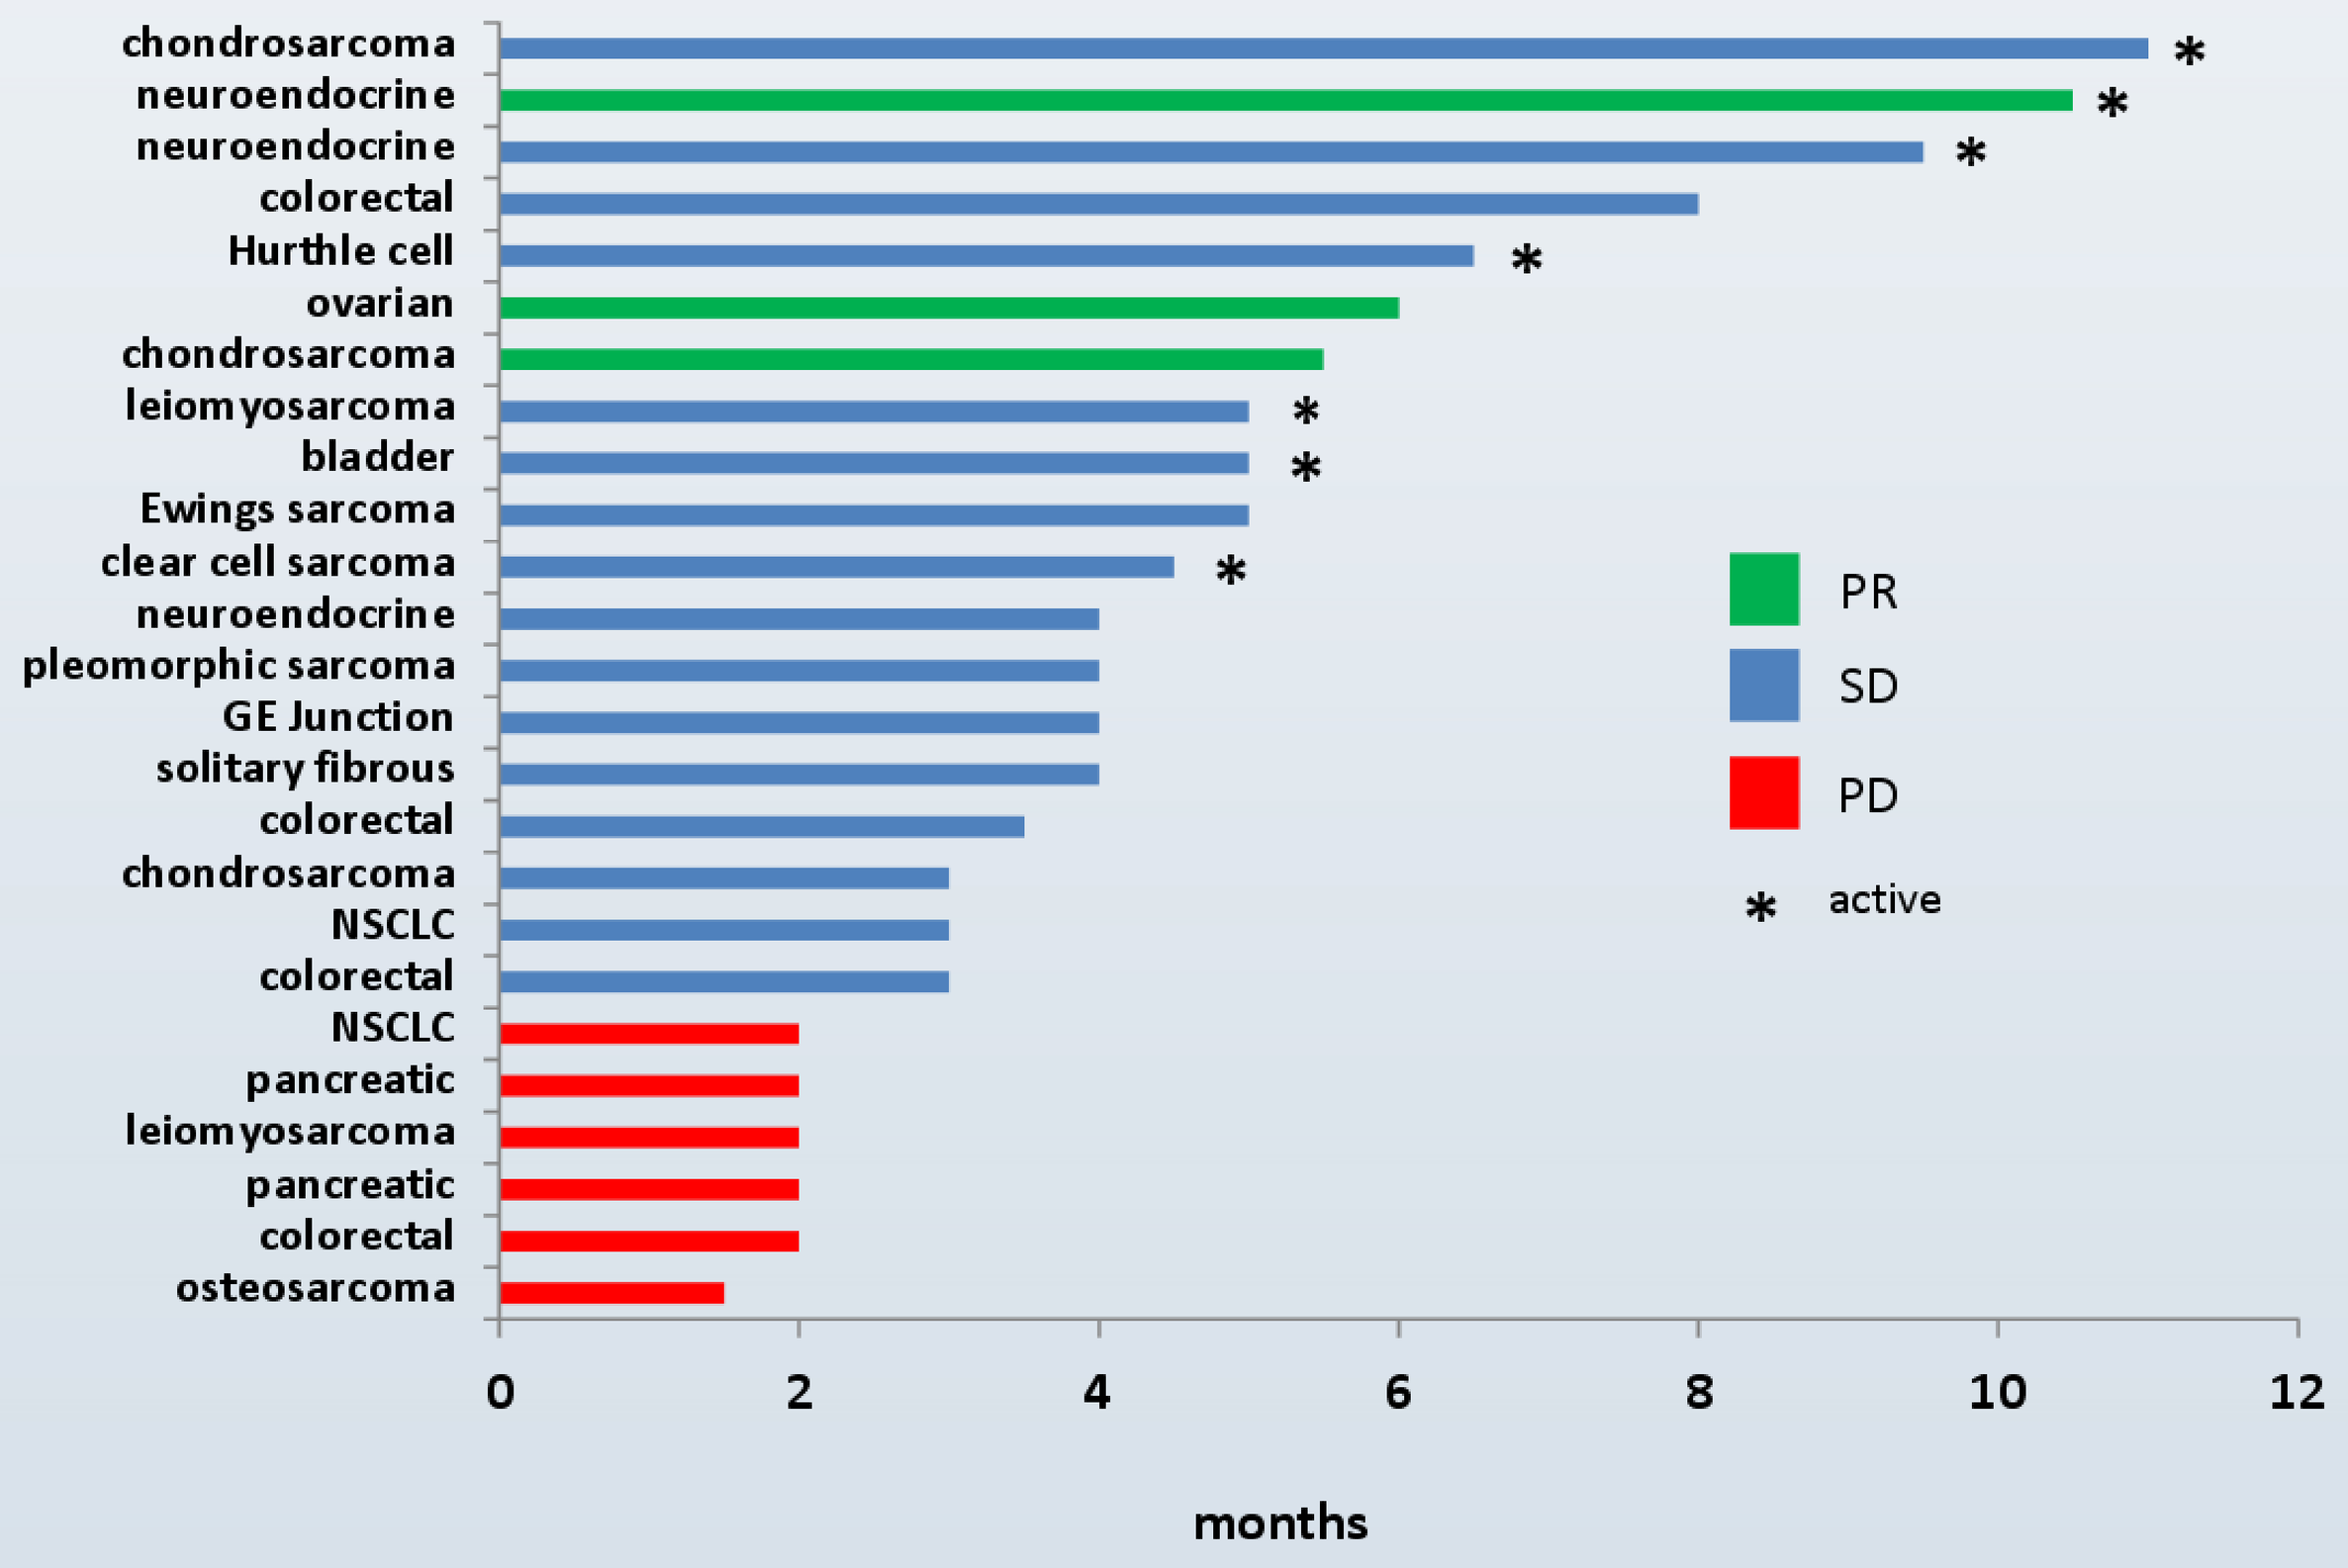
chondrosarcoma
neuroendocrine
neuroendocrine
colorectal
Hurthle cell
ovarian
chondrosarcoma
leiomyosarcoma
bladder
Ewings sarcoma
clear cell sarcoma
neuroendocrine
pleomorphic sarcoma
GE Junction
solitary fibrous
colorectal
chondrosarcoma
NSCLC
colorectal
NSCLC
pancreatic
leiomyosarcoma
pancreatic
colorectal
osteosarcoma
0
2
4
6
8
10
12
months
PR
SD
PD
* active

Table 4. Treatment-related Grade ≥2 AEs any time on study

Toxicity, highest grade per patient (N= 30 patients)	Grade 2	Grade 3	Grade 4/5
Hematologic			
Neutropenia	8	2	0
Thrombocytopenia	3	2	0
Anemia	4	4	0
Nonhematologic			
Vomiting/Nausea/Diarrhea	7	2	0
Fatigue	4	2	0
ALT/AST	3	1	0
Hyponatremia	0	2	0
Stroke/Cerebral Bleed	1	1	0
Pain	6	1	0
Mucositis (esophagitis, proctitis, vaginitis)	7	2	0
Headache	0	1	0
Hand-foot Syndrome	8	0	0
Hypertension	8	0	0

Conclusions

- The RPTD for this novel combination is pazopanib at 800 mg daily plus TH-302 at 340 mg/m² on days 1, 8 and 15 of each 28-day cycle.
- Toxicities for this combination are generally managable with appropriate dose holds, reductions and supportive care as clinically indicated.
- Preliminary activity is seen in treatment-refractory cancers and support potential value of co-targeting tumor angiogenesis and tumor hypoxia.

References

1. Paez-Ribes M, Allen E, Hudock J, et al. (2009) Antiangiogenic therapy elicits malignant progression of tumors to increased local invasion and distant metastasis. Cancer Cell, 15:220-31

2. Meng F, Evans JW, Bhupathi D, et al. (2012) Molecular and cellular pharmacology of the hypoxia-activated prodrug TH-302. Mol Cancer Ther, 201:740-51.

3. Sun JD, Liu Q, Wang J, Ahluwalia D, et al. (2012) Selective tumor hypoxia targeting by hypoxia-activated prodrug TH-302 inhibits tumor growth in preclinical models of cancer. Clin Cancer Res,18:758-70

4. Threshold Pharmaceutricals, Inc, unpublished data.

Acknowledgements

We are thankful to the patients and their families and the DCI Phase I study team with special acknowledgement to Faith Gilchrist for data assistance.

This study was approved and funded by the National Comprehensive Cancer Network (NCCN) from general research support provided by GlaxoSmithKline and research supported by Threshold Pharmaceuticals, Inc.

Disclosures

Drs. George and Riedel have research support from GSK.

Drs. Hurwitz and Uronis have research support from Threshold. Dr. Hurwitz has stock in Threshold.