Selective, State-Independent Inhibitors of Nav1.7 are Analgesic in a Non-Human Primate Model of Acute Thermal Pain

Jacob Beckley[1], Anton Delwig[2], David Yeomans[3], Hassan Pajouhesh[2], Dennis Monteleone[2], Xiang Zhou[2], Justin Du Bois[3], John Mulcahy[2]
(1) SiteOne Therapeutics, Bozeman, MT (2) SiteOne Therapeutics, South San Francisco, CA (2) Stanford University, Palo Alto, CA

Poster #: PTH 536

Purpose

- The transmission of nociceptive signaling in response to noxious stimuli requires the voltage gated sodium ion channel isoform Nav1.7.
- Loss-of-function mutations in Nav1.7 result in congenital insensitivity to pain[1], an effect that has been recapitulated in Nav1.7 knockout mice[2].
- A 2 amino acid variation in the extracellular pore region of primate Nav1.7 (including humans) differs from all other primate voltage-gated sodium ion channel isoforms and Nav1.7 in other mammals[3].
- SiteOne Therapeutics has leveraged this variation in the extracellular pore to discover selective inhibitors of Nav1.7.
- During the lead optimization effort, a second series of Nav1.7 inhibitors was discovered that are less selective but equipotent against human and mouse Nav1.7.
- **The objective of this study was to determine the potency and selectivity of SiteOne Therapeutics' Nav1.7 inhibitors and to assess analgesic efficacy in animal models of thermal pain.**
- **All experimental procedures were approved by an Institutional Animal Care and Use Committee in accordance with the National Institutes of Health's Guide for the Care and Use of Laboratory Animals, and conducted at an AAALAC-accredited facility. Efforts were taken to minimize pain and distress of experimental animals.**

Methods

Electrophysiology

Whole cell recordings were carried out on HEK or CHO cells that stably expressed one Nav1.x isoform. For recordings with mouse Nav1.7, CHO cells were transfected with GFP and mouse SCN9A plasmids, and fluorescently-labeled cells were recorded. Cells were patch clamped with pipettes that contained a CsF-based internal solution. Currents were evoked with a voltage step from -110mV to the voltage for maximum activation for 10ms. For determining use-dependent block, the voltage step occurred at 30Hz. For recordings that assessed state-dependent inhibition, the voltage was stepped from -120mV to the voltage of half-inactivation for 8 seconds, then following repolarization to -120mV for 20ms, was stepped to the voltage of max activation.

Once the baseline evoked current was stable in amplitude, the test article was washed onto the cell. Following stable inhibition, a half-log higher concentration was perfused onto the cell. Drug was then washed out until the cell returned to a stable current; these compounds readily wash out back to baseline current values. IC50 was calculated with curve-fitting algorithm in Excel (Microsoft) or Prism software (GraphPad).

Animal Studies

Non-human primate (NHP) noxious heat model: Study was conducted similarly as described[4]. Briefly, four or five male cynomolgus monkeys (*Macaca fascicularis*), aged 4-7 years, were tested individually. The subject was sedated and then maintained in a light anesthetic state with Propofol. The subject was hooked up to an ICU monitor, and heart rate, ECG, pO2, respiratory rate, and body temperature was continuously monitored throughout the experiment.

The hand was heated at either 6.5º or 0.9ºC/s to either elicit an withdrawal induced by A-delta or C fiber activation, respectively. The maximum stimulation for the A-delta fiber activation was between 4 and 6 seconds, while the maximum for C fiber was 20 seconds. Once baseline withdrawal response latency was stable, animals were dosed IV with either Compound A, B, or C. In experiments with escalating doses, each successive dose was given about 30 minutes apart. In the single dose study with Compound C, experimenters were blinded to treatment group, with 2 subjects receiving active compound, and 2 subjects receiving vehicle.

Mouse Hargreaves assay: Adult male C57Bl/6 mice (n=12) were were placed into one of two groups: the first group received saline (SubQ) on the first test session and then Compound D (SubQ, 0.25 mg/kg) on the second session, while the second group had the opposite dosing order. Subjects underwent Hargreaves testing 15 minutes after dosing. Following the Hargreaves testing session, spontaneous locomotion was measured for five minutes in an open arena. Subjects were tested every three days.

Results

Difference in Human Nav1.7 Binding Site

A

Nav Isoform	Domain I	Domain II	Domain III	Domain IV
Nav1.4	RLMTQDYWEN	RILCGEWIET	VATFKGWMDI	ITTSAGWDGL
Nav1.5	RLMTQDCWER	RILCGEWIET	VATFKGWMDI	ITTSAGWDGL
Nav1.6	RLMTQDYWEN	RVLCGEWIET	VATFKGWMDI	ITTSAGWDGL
Nav1.7	RLMTQDYWEN	RVLCGEWIET	VATFKGWTII	ITTSAGWDGL
Cyno Nav1.7	RLMTQDYWEN	RVLCGEWIET	VATFKGWTII	ITTSAGWDGL
Mouse Nav1.7	RLMTQDYWEN	RVLCGEWIET	VATFKGWMDI	ITTSAGWDGL
Rat Nav1.7	RLMTQDYWEN	RVLCGEWIET	VATFKGWMDI	ITTSAGWDGL

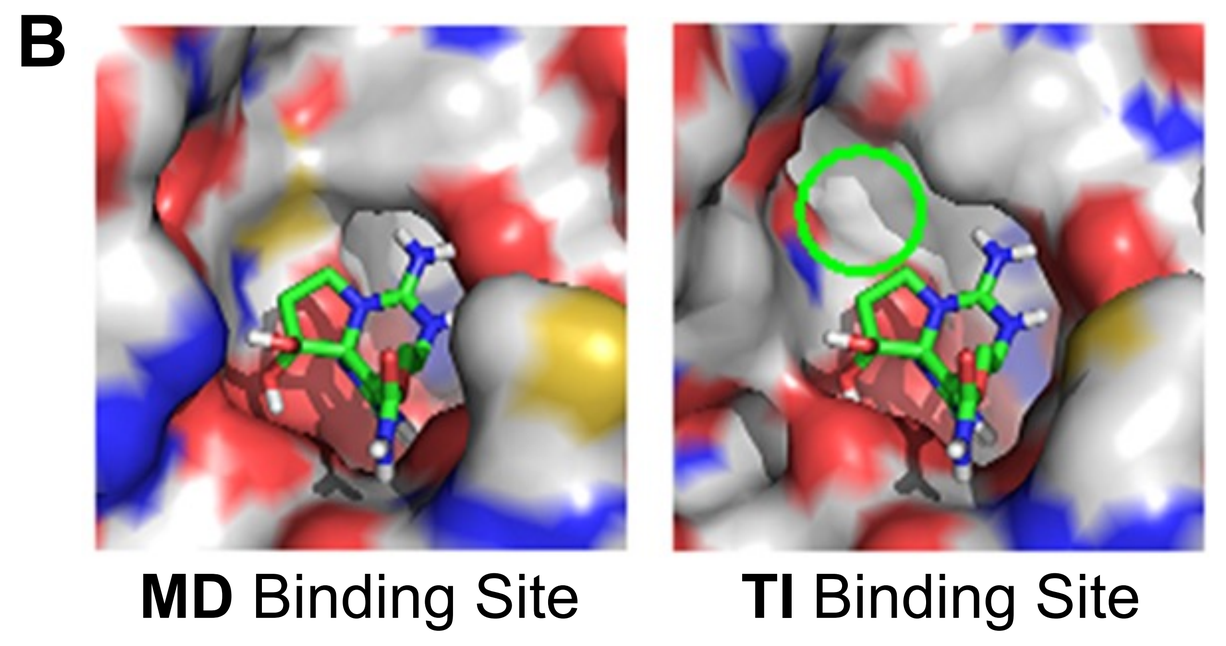
B
MD Binding Site
TI Binding Site

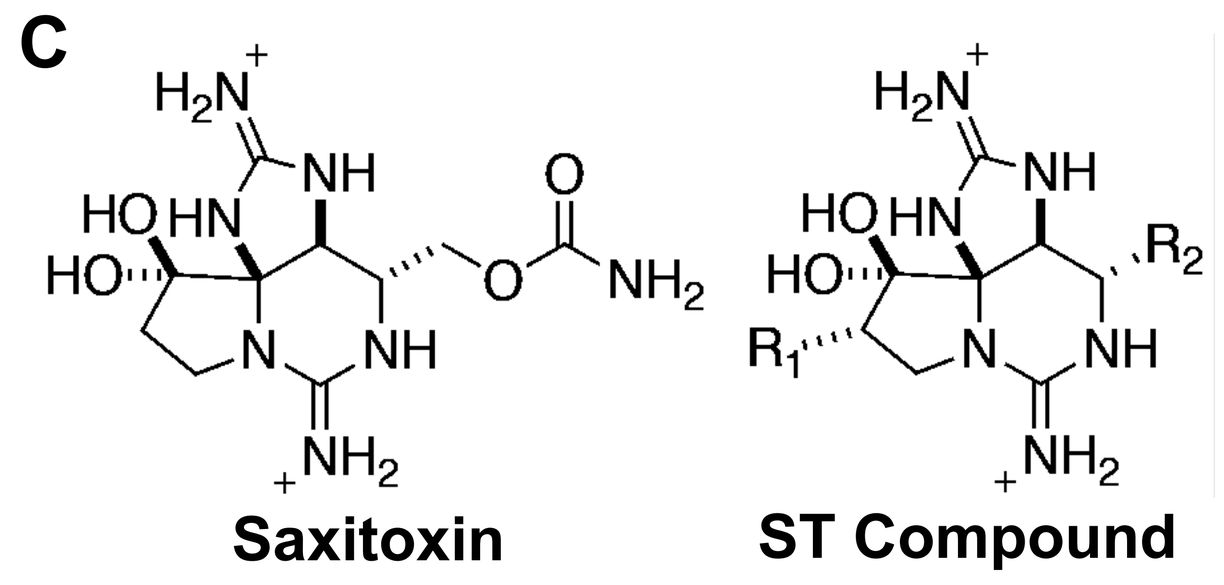
C
Saxitoxin
ST Compound

Figure 1. (A) Primary sequence comparison of the pore loop regions for select human Nav1.x isoforms reveals a two amino acid variation in domain III of the extracellular pore region that is unique to the primate $Na_V1.7$ subtype. Amino acids comprising the DEKA selectivity filter are highlighted in blue. **(B)** Saxitoxin (STX) docked to Nav1.4 (left) and the identical pose in Nav1.7 (right). **(C)** Chemical structures for STX (left) and ST Compounds A-D (Right).

Physiological Characteristics of ST Compounds

Discovery of Highly Potent and Selective Nav1.7 Inhibitors

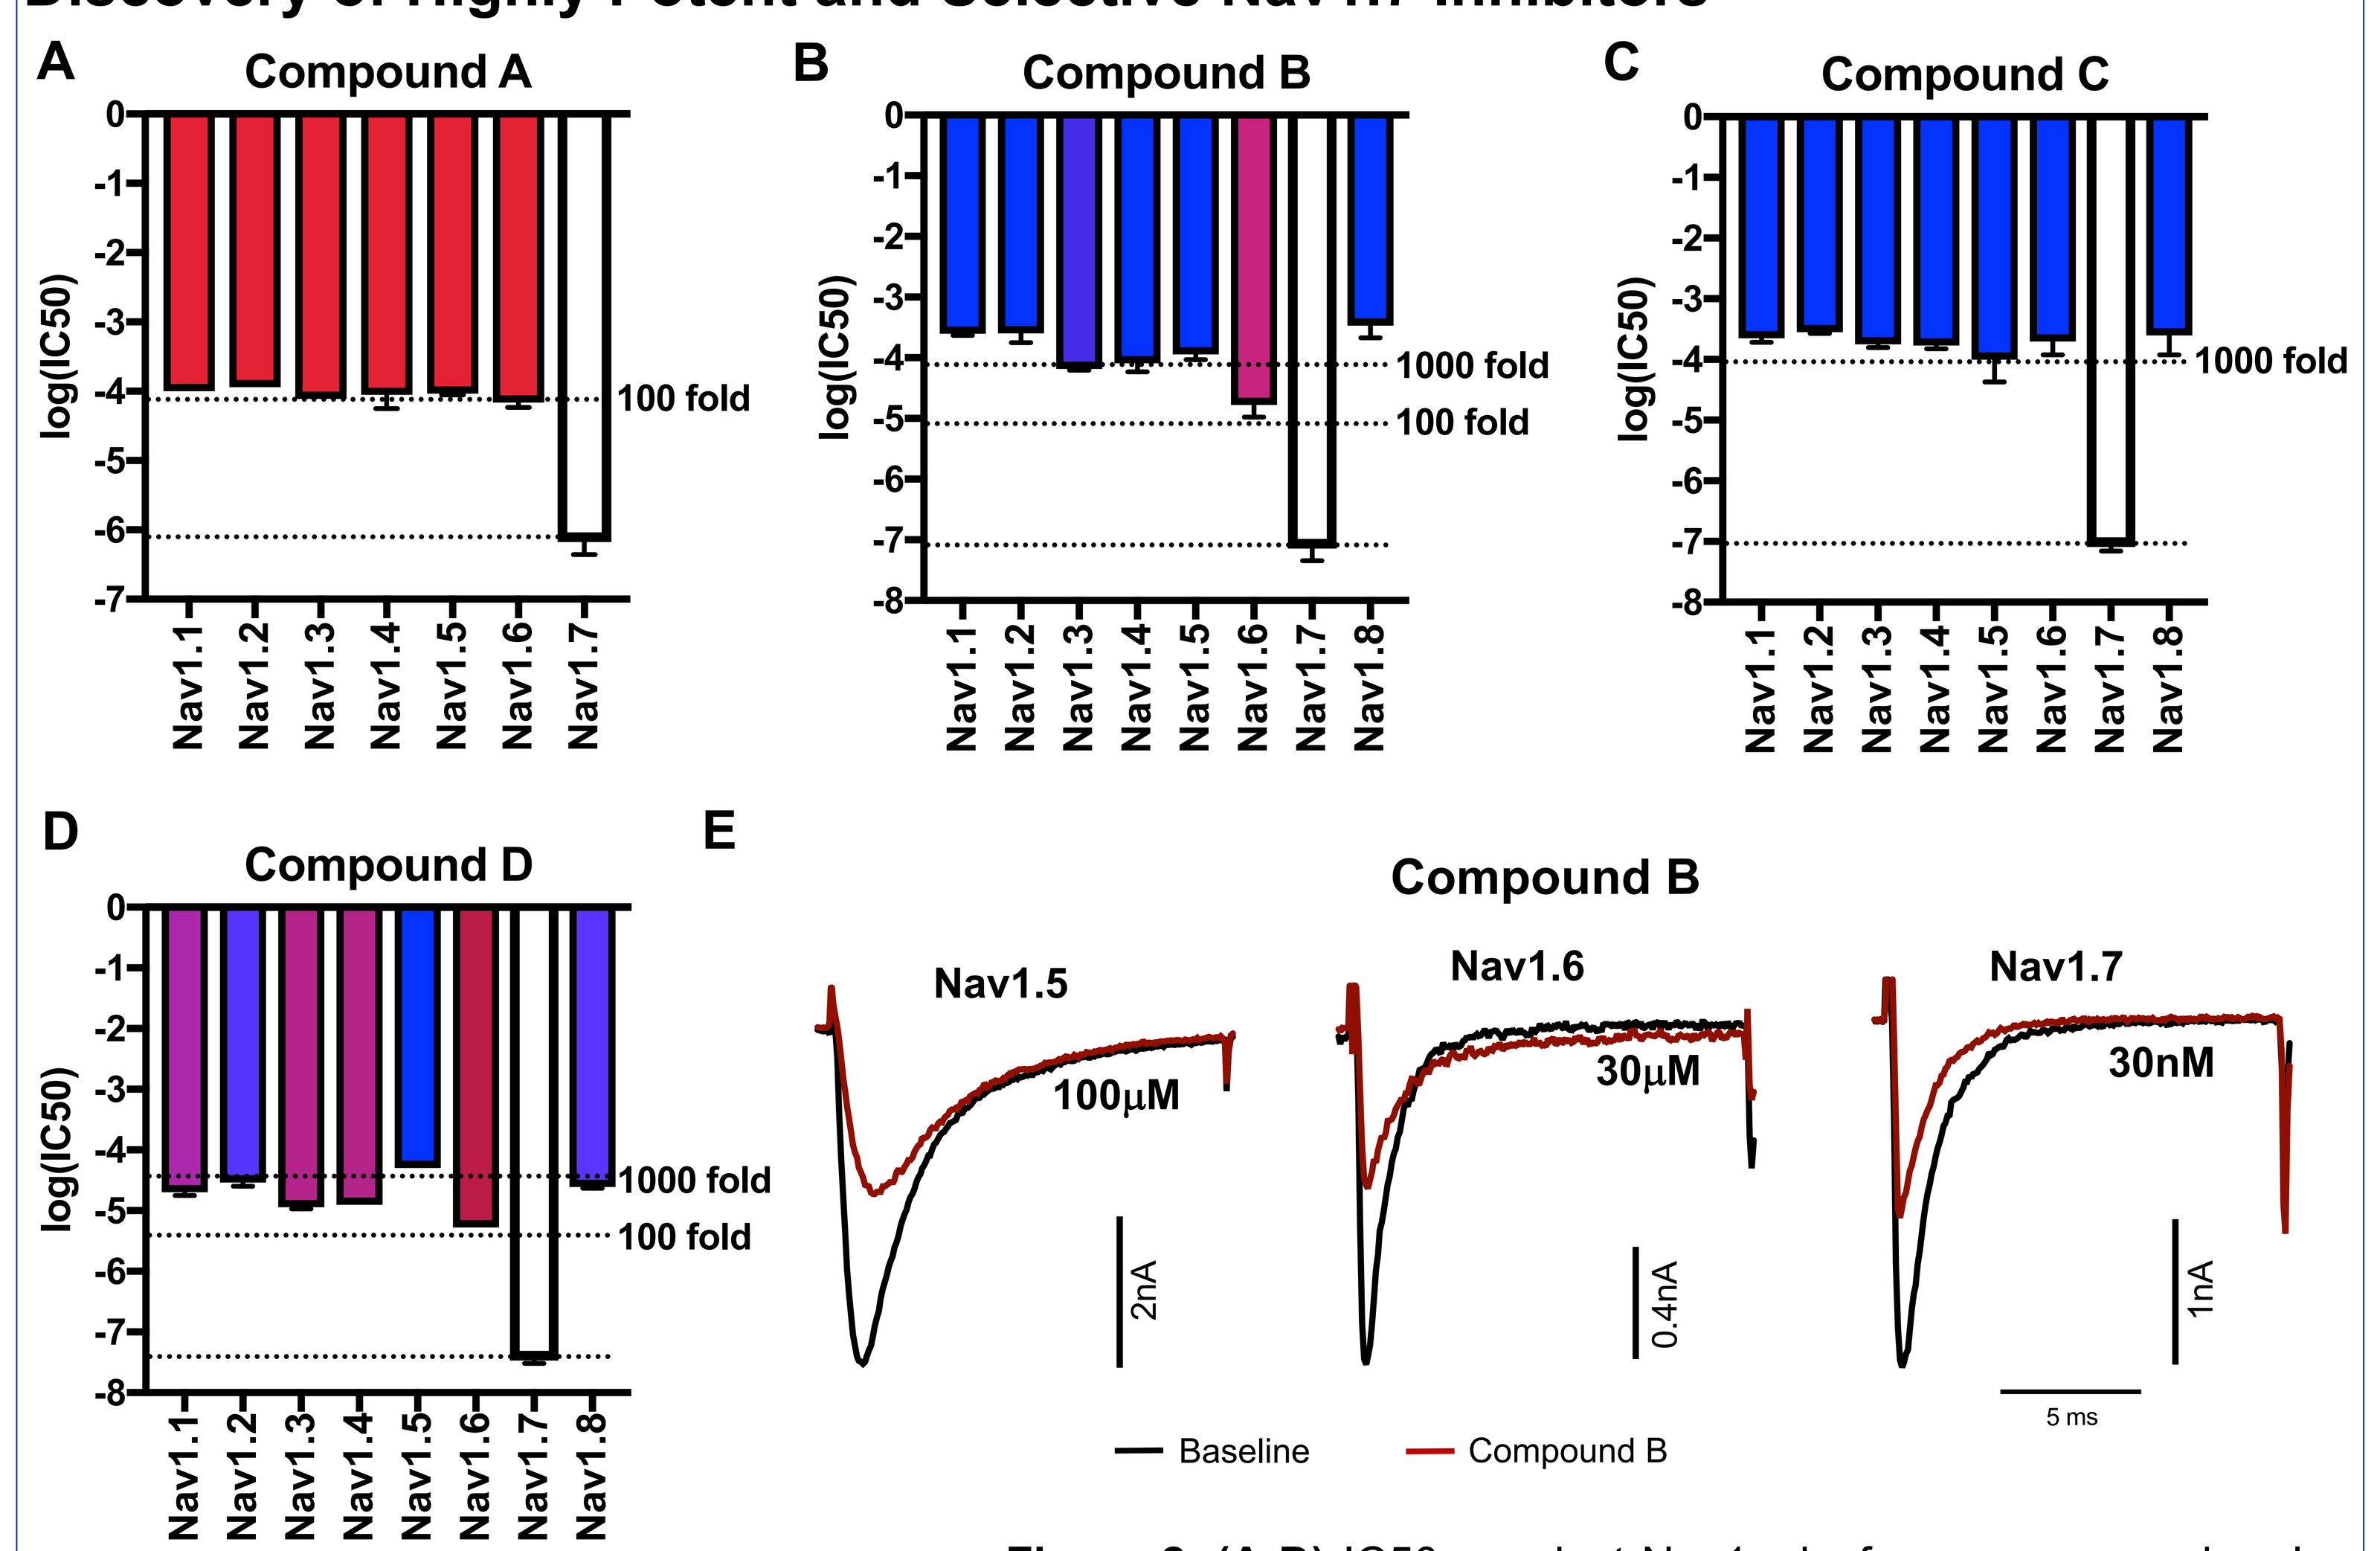
A Compound A
B Compound B
C Compound C
D Compound D
log(IC50)
100 fold
1000 fold
E Compound B
Nav1.5 100μM
Nav1.6 30μM
Nav1.7 30nM
Baseline
Compound B

F

Compound	Nav1.7 IC50(nM)
Compound A	710 ± 282
Compound B	79 ± 38
Compound C	82 ± 13
Compound D	34 ± 4

IC50s are mean ± SD

Figure 2. (A-D) IC50s against Nav1.x isoforms, expressed as log (M), for Compound A-D. Selectivity of Nav1.7 over other Nav1.x isoforms is indicated by the color of the bar associated with each isoform, from red (100x selectivity) to blue (>1000X). Bars are mean ± SD. **(E)** Sample traces for Nav1.5, Nav1.6 and Nav1.7, with a baseline current (black), and current following inhibition due to Compound B (red). Concentrations of Compound B were selected because they are close to the IC50 value – 100μM for Nav1.5,; 30μM for Nav1.6; 30nM for Nav1.7. **(F)** Table with IC50 (nM) values for Compounds A-D against human Nav1.7. Values are mean ± SD.

Evidence of State-Independent Inhibition

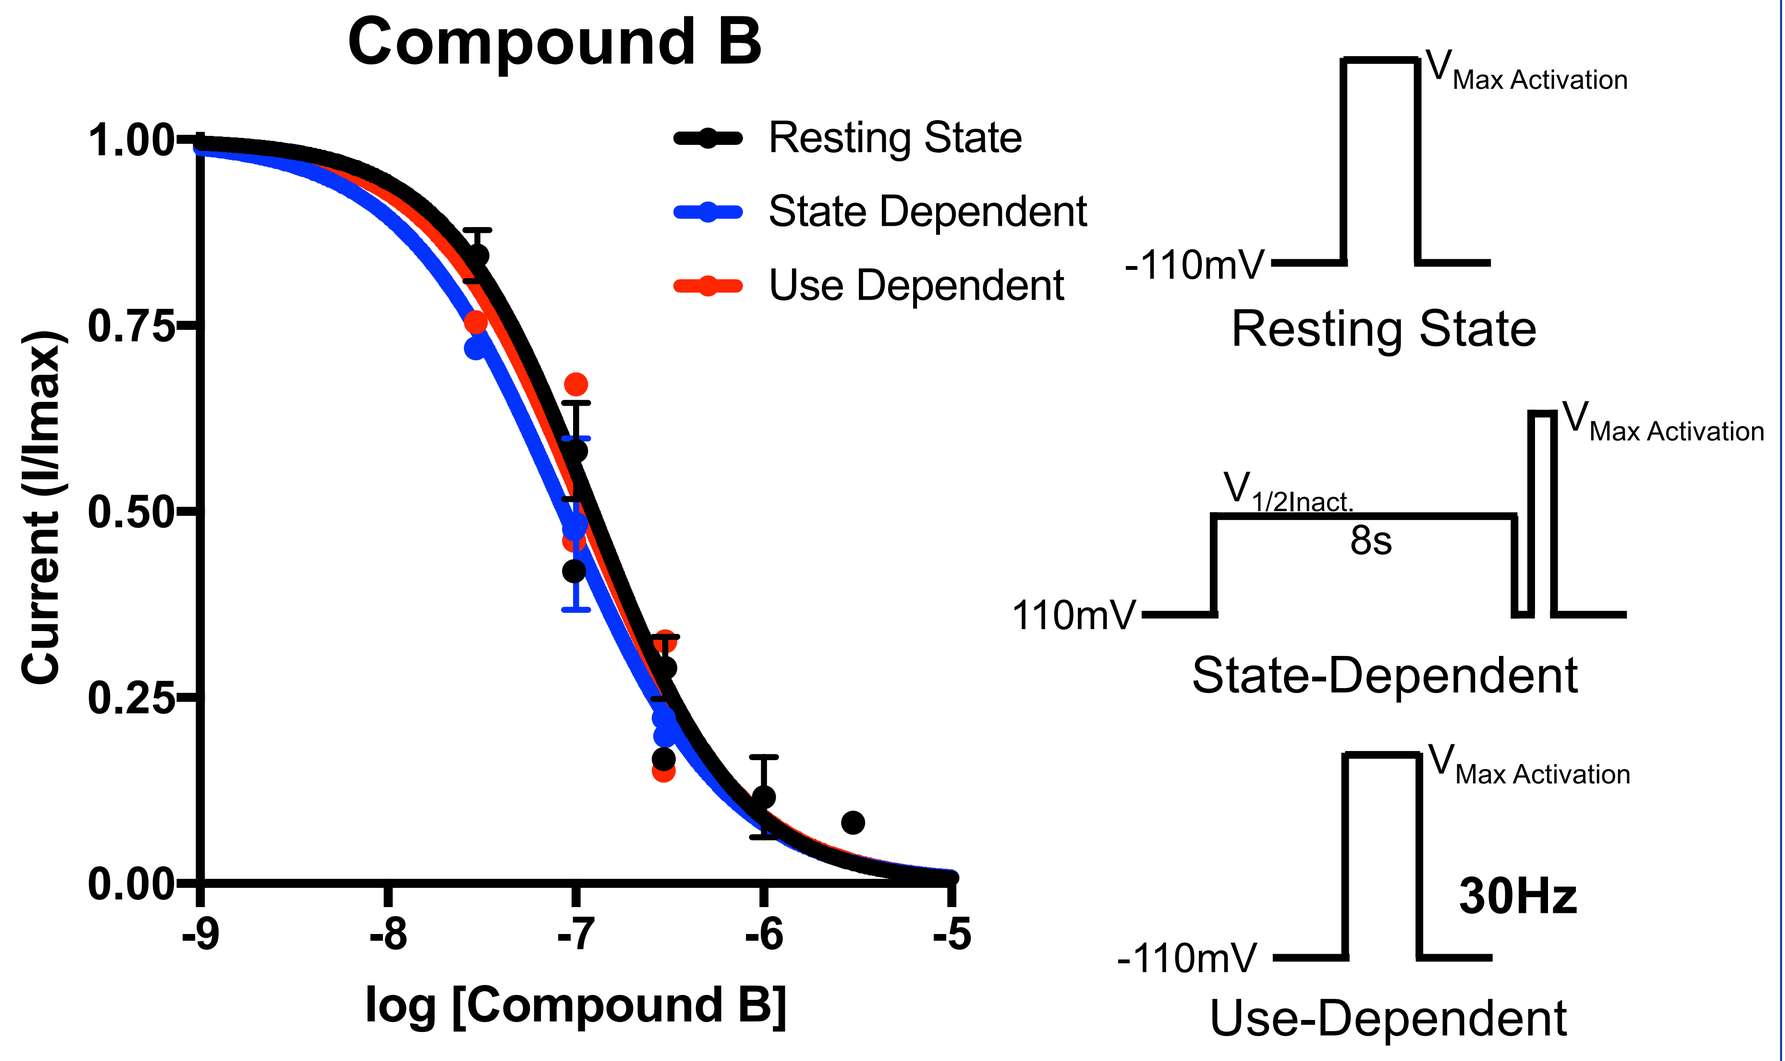
Compound B
Resting State
State Dependent
Use Dependent
Current (I/Imax)
log [Compound B]
Resting State
State-Dependent
Use-Dependent
30Hz

Figure 3. (**Left**) Dose-response relationship for Compound B inhibition of Nav1.7 when current is evoked using different protocols. Points are either a single replicate or mean ± SD. The stimulation patterns – resting state (black), state-dependent (blue), and use-dependent (red), are shown on the **right**.

Not All Compounds are Less Potent against Mouse Nav1.7

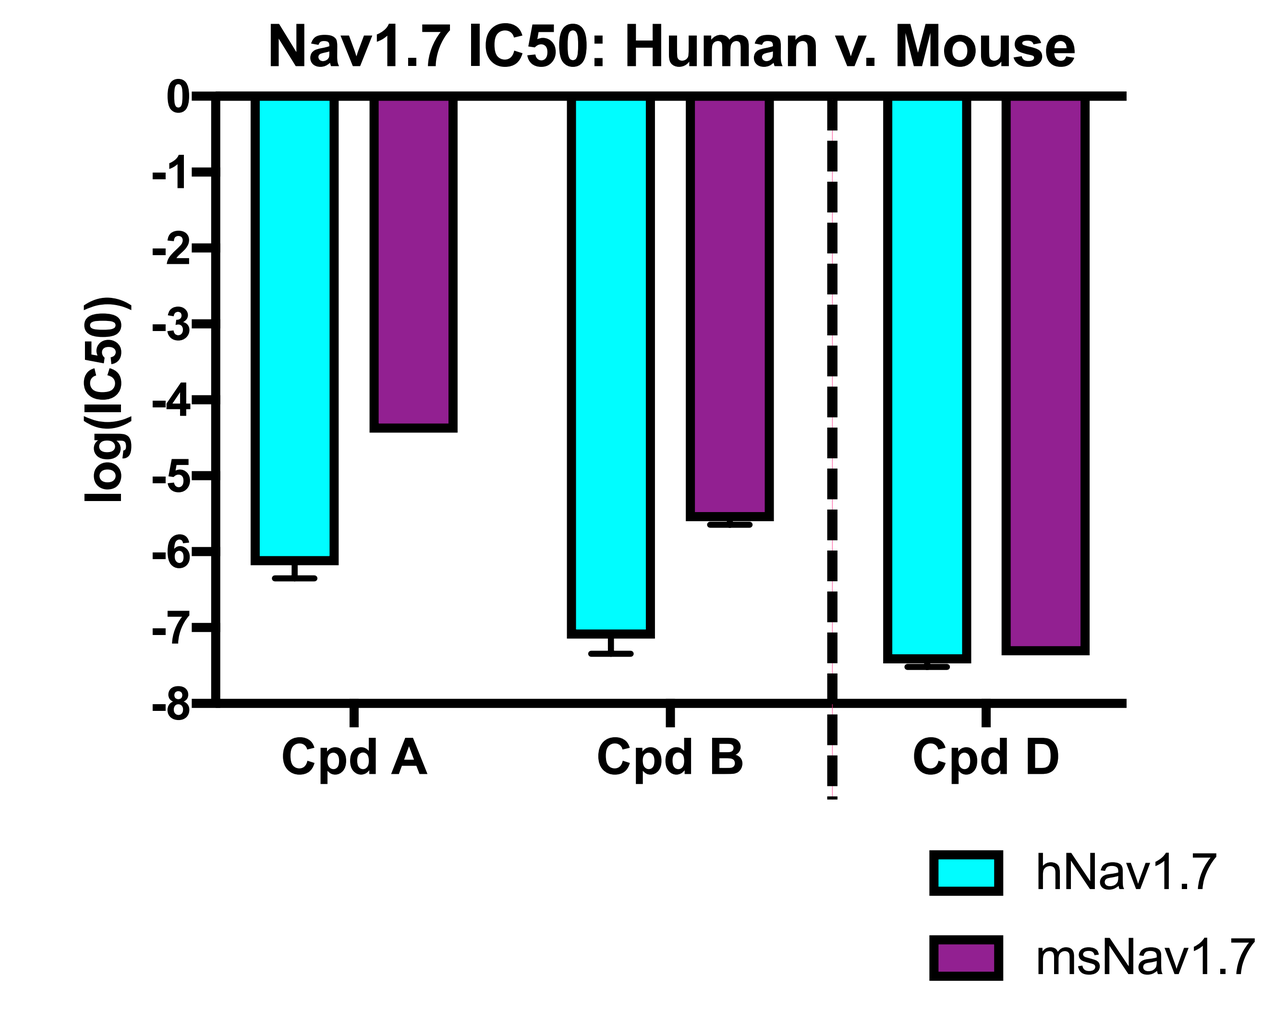
Nav1.7 IC50: Human v. Mouse
log(IC50)
Cpd A
Cpd B
Cpd D
hNav1.7
msNav1.7

Figure 4. IC50s, expressed in log (M), for human (teal) or mouse (purple) Nav1.7. Bars are mean ± SD.

Analgesic efficacy in a cynomolgus monkey model

Compound A

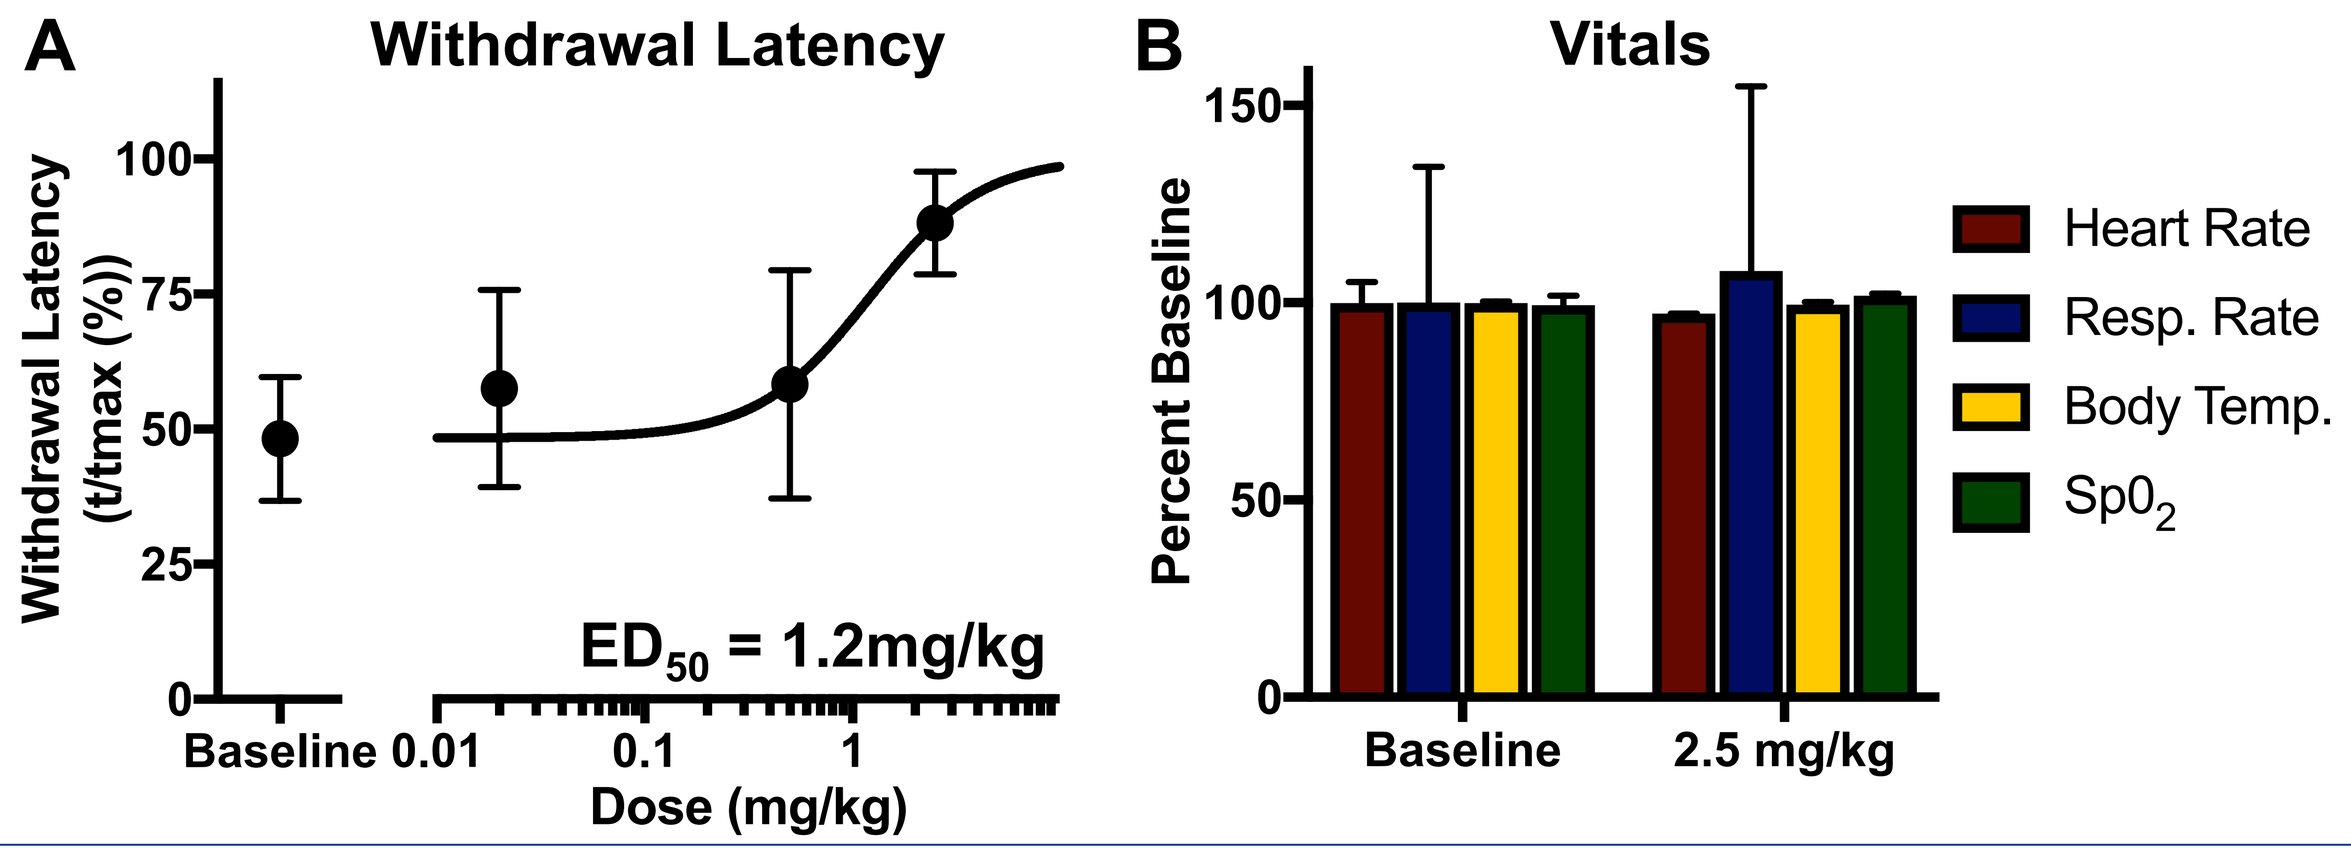
A Withdrawal Latency
ED50 = 1.2mg/kg
Dose (mg/kg)
B Vitals
Heart Rate
Resp. Rate
Body Temp.
Sp02

Figure 5. (A) Dose-response relationship for withdrawal latency to a thermal stimulus (A-delta). Points are expressed as mean ± SD. **(B)** Summary of effect of high dose of Compound B (2.5 mg/kg, IV) on measured vital signs, expressed as a percent of the baseline value.

Compound B

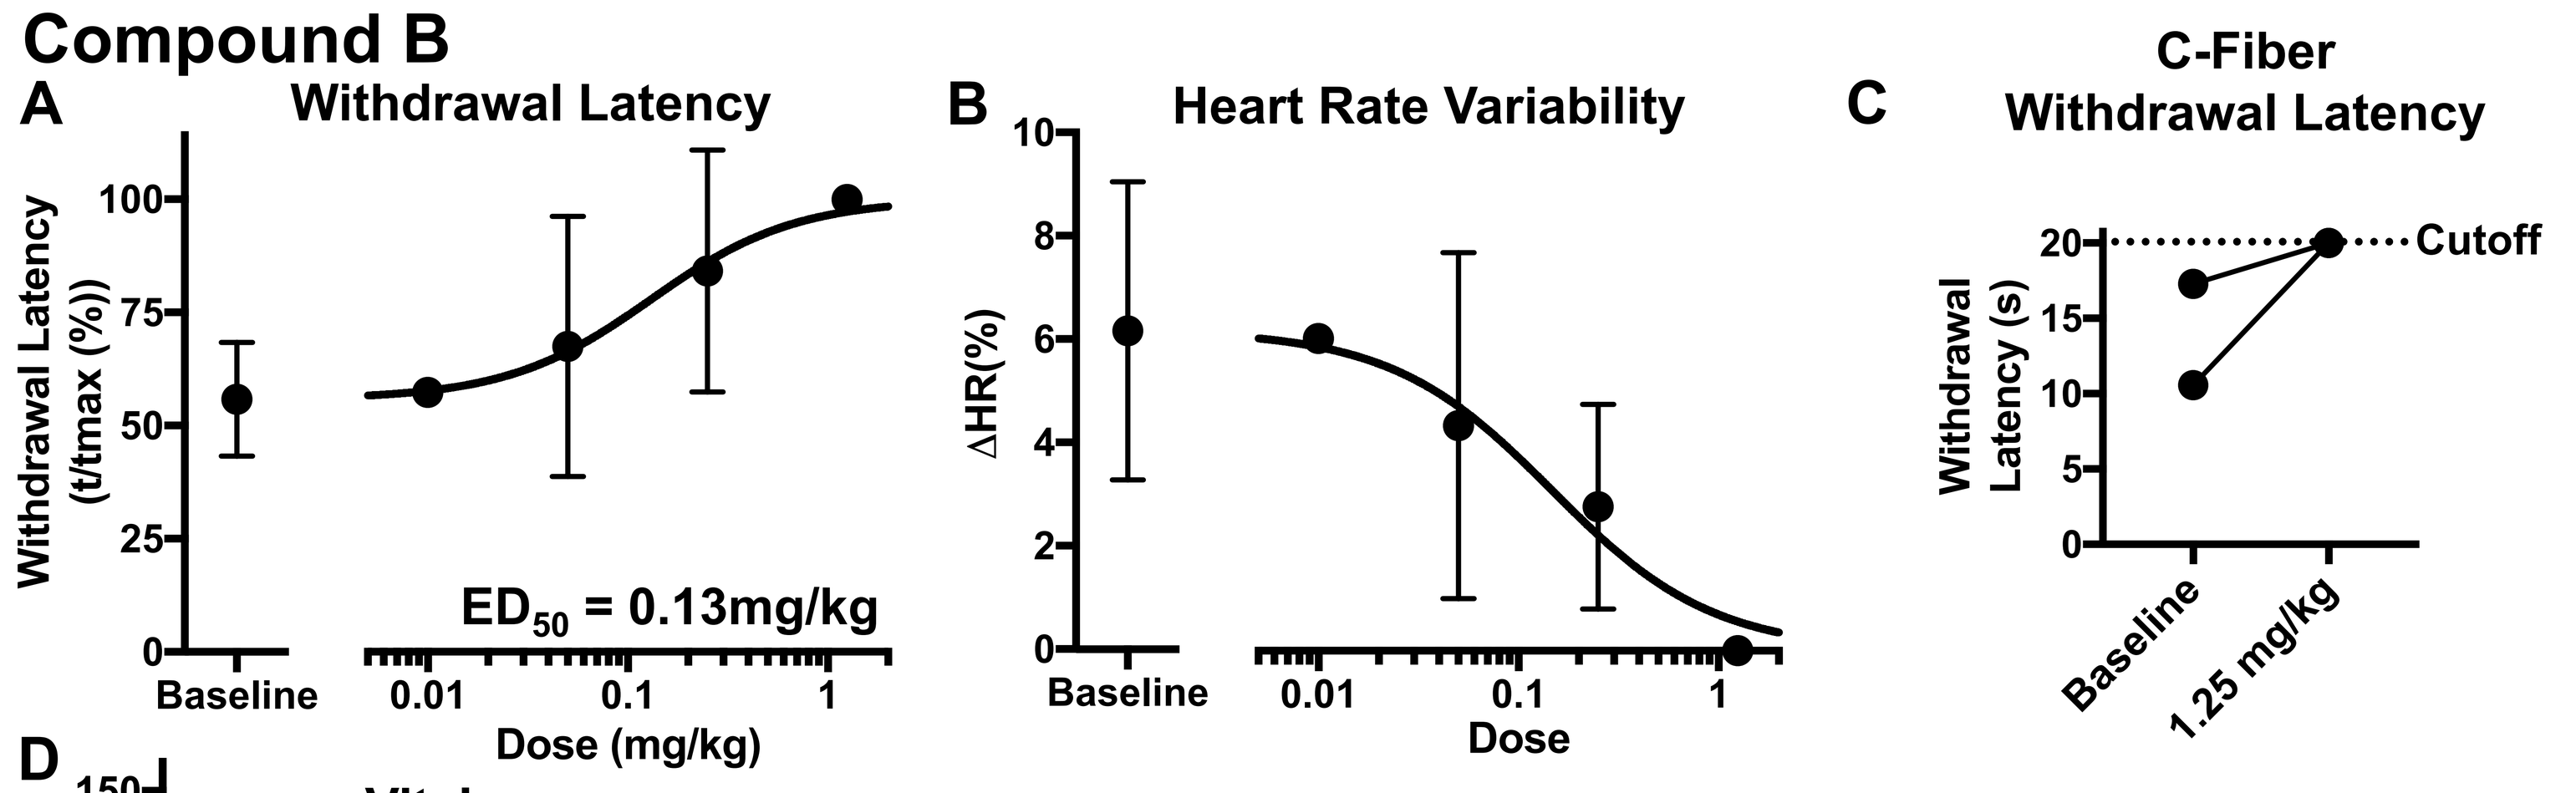
A Withdrawal Latency
ED50 = 0.13mg/kg
B Heart Rate Variability
C C-Fiber Withdrawal Latency
Cutoff

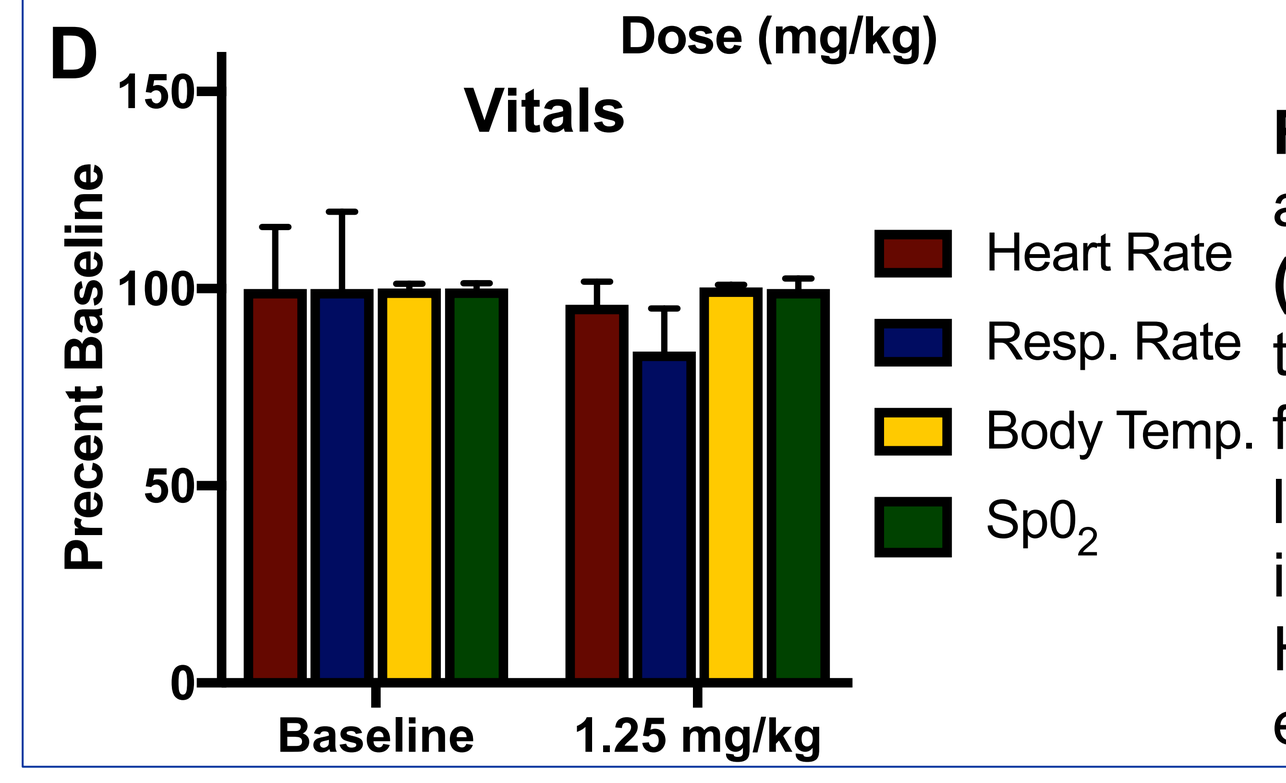
D Vitals
Heart Rate
Resp. Rate
Body Temp.
Sp02

Figure 6. (A) Dose-response relationship for withdrawal latency to a thermal stimulus (A-delta). Points are expressed as mean ± SD. **(B)** Dose-response relationship for change in heart rate due to the thermal stimulus. Heart rate at end of stimulus was subtracted from rate immediately prior to stimulus. **(C)** Change in withdrawal latency due to the slow ramp, C-fiber activation stimulus in 2 individual subjects that received the high dose (1.25 mg/kg, IV). Horizontal dashed line represents the cut-off time. **(D)** Summary of effect of high dose of Compound B on measured vital signs.

Compound C

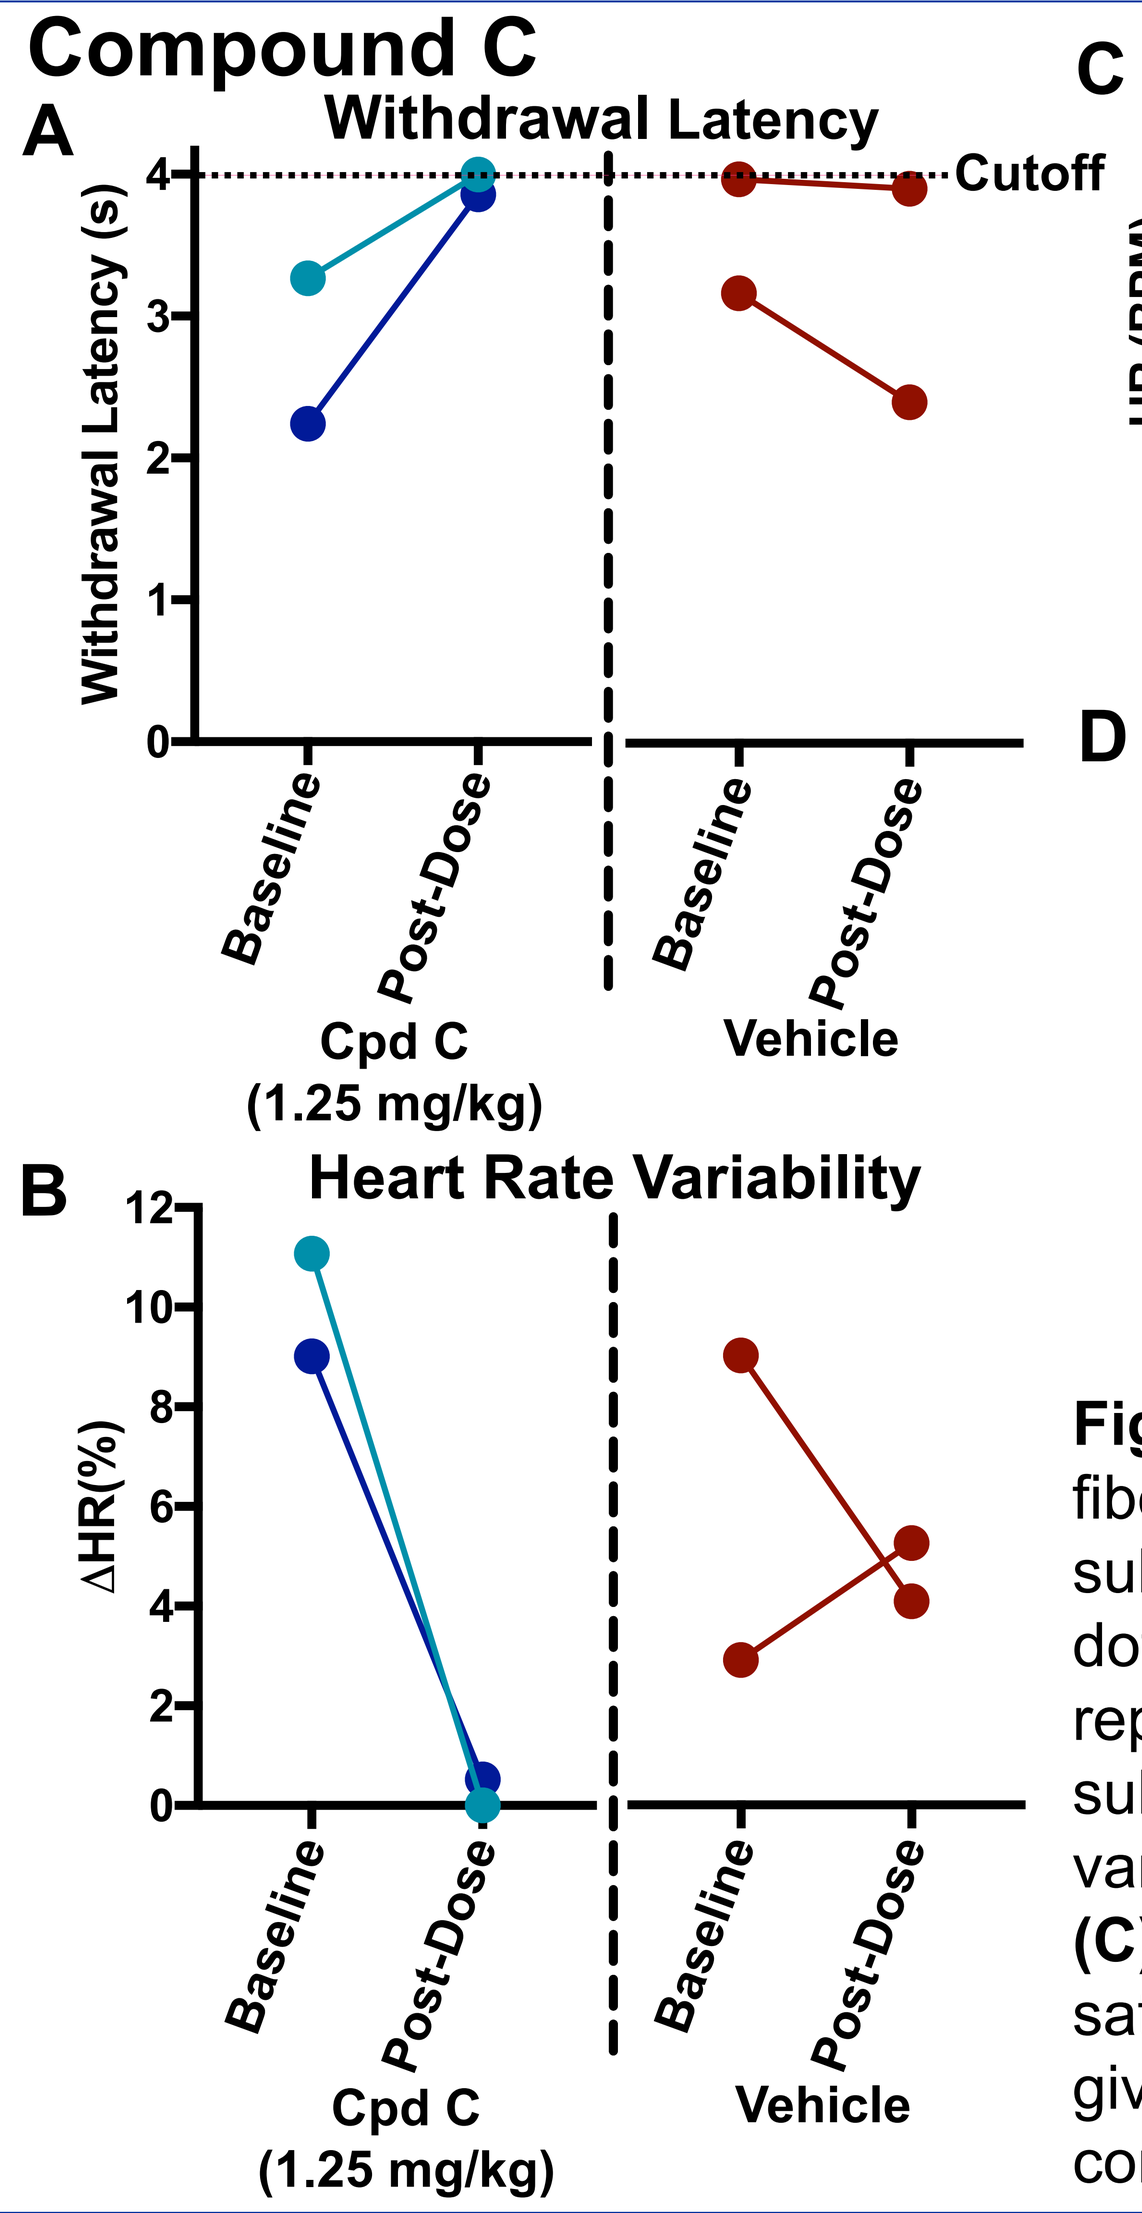
A Withdrawal Latency
Cutoff
Cpd C (1.25 mg/kg)
Vehicle
B Heart Rate Variability

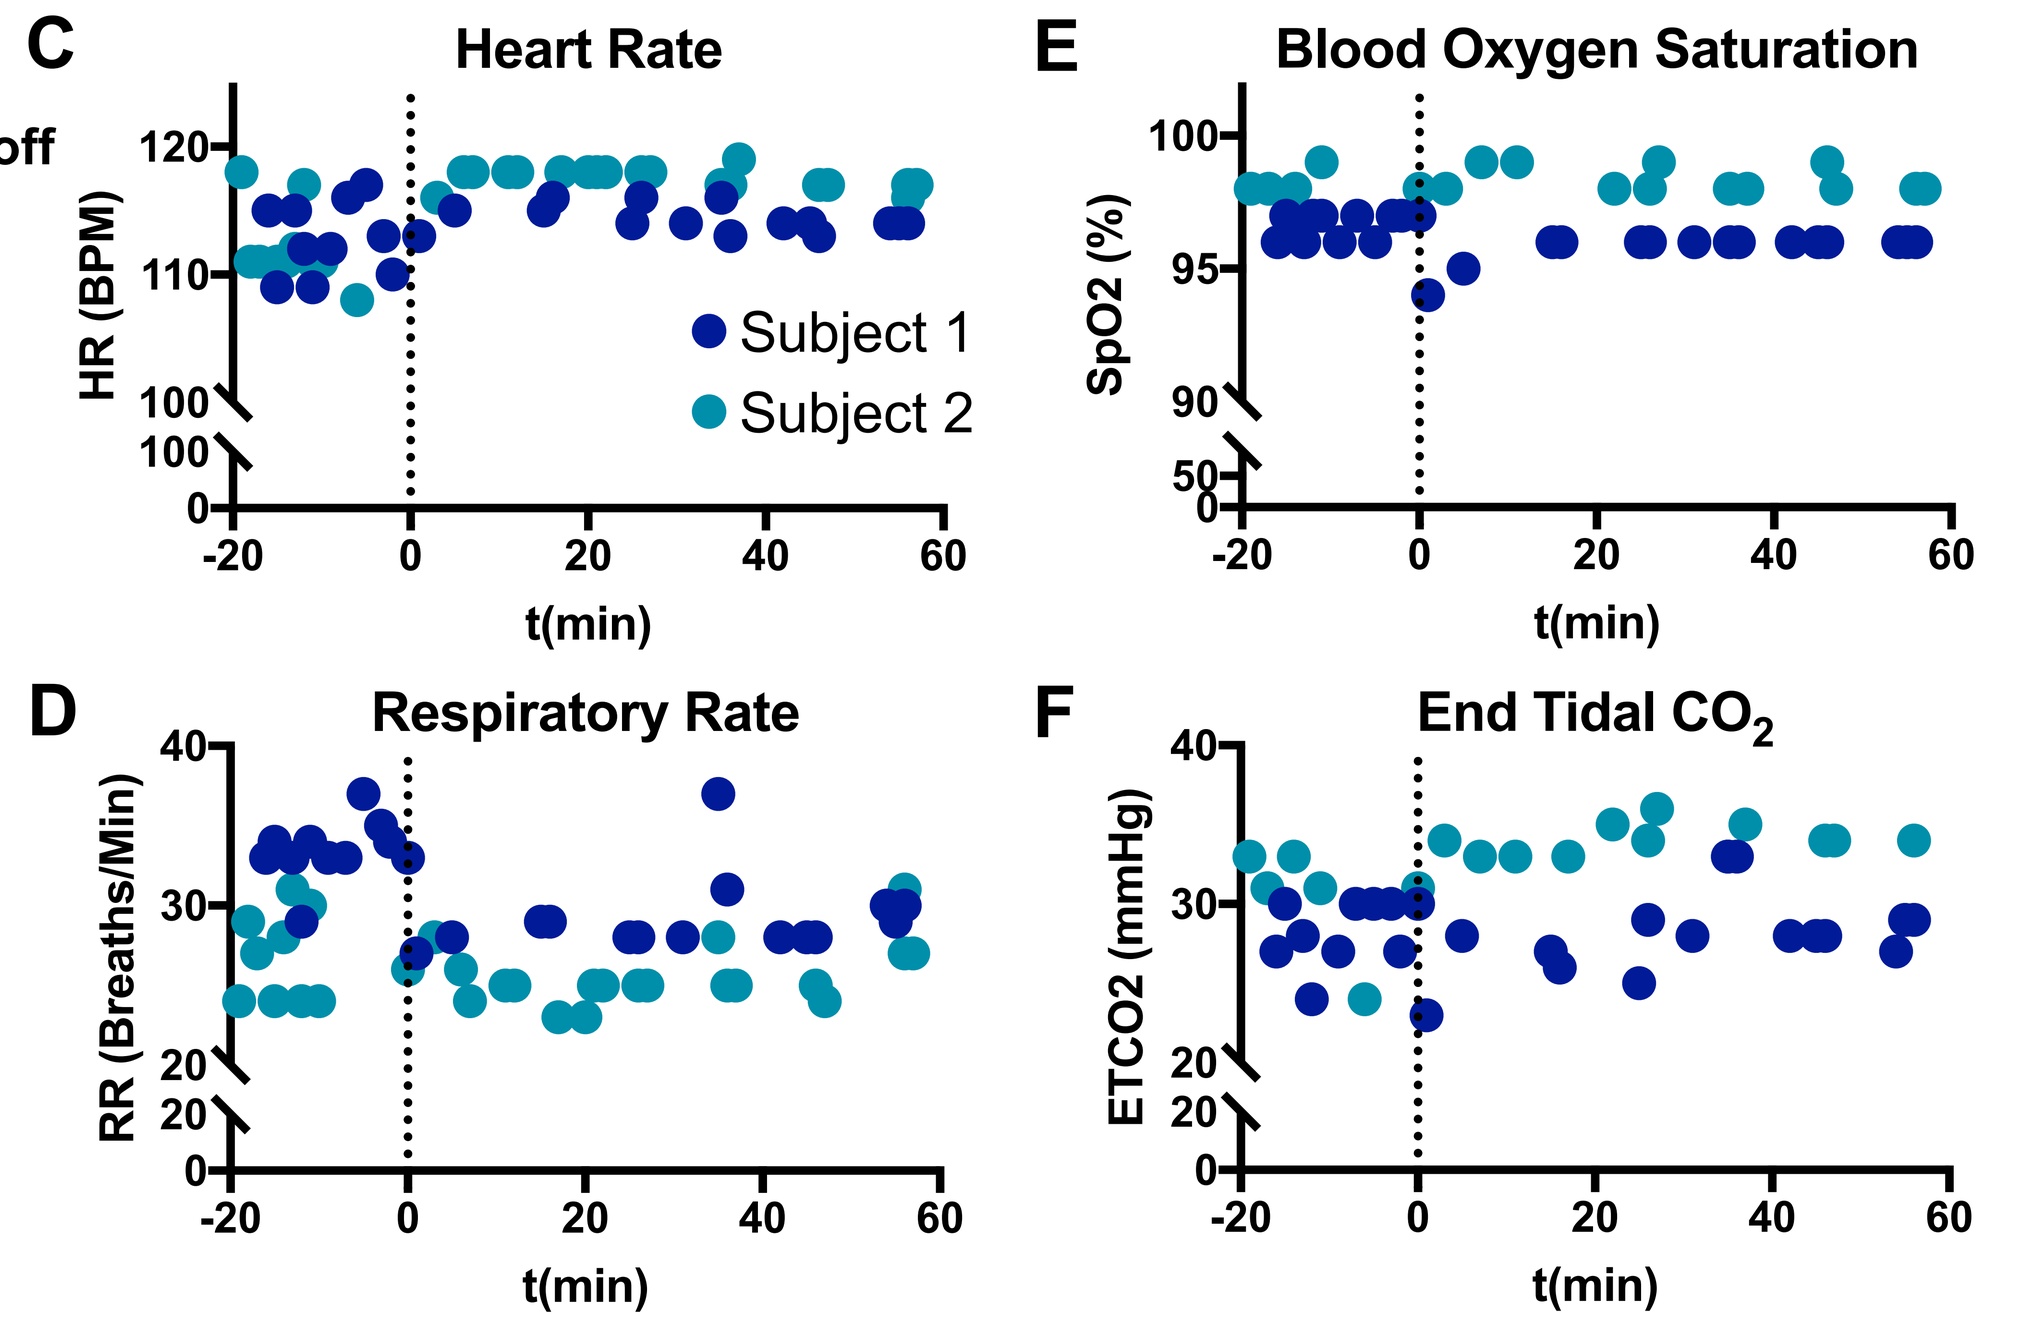
C Heart Rate
Subject 1
Subject 2
D Respiratory Rate
E Blood Oxygen Saturation
F End Tidal CO2
t(min)

Figure 7. (A) Change in withdrawal latency due to the fast ramp, A-delta fiber stimulus in individual subjects. The blue dots represent individual subjects given the active compound (Cpd C, 1.25 mg/kg, IV) and the red dots are subjects that were given vehicle control. Horizontal dashed line represents the cut-off time. The two shades of blue correspond to two subjects (dark blue – Subject 1; light blue – Subject 2). **(B)** Heart rate variability in subjects that received either active (blue) or vehicle (red). **(C)** Heart rate (BPM), **(D)** Respiratory rate (breaths/min), **(E)** oxygen saturation (% SpO_2), and **(F)** End tidal CO2 (mmHg) for the two subjects given Compound C. The vertical line indicates the time that the compound was administered (t=0min).

Compound D Reduces Thermal Sensitivity in a Mouse Model

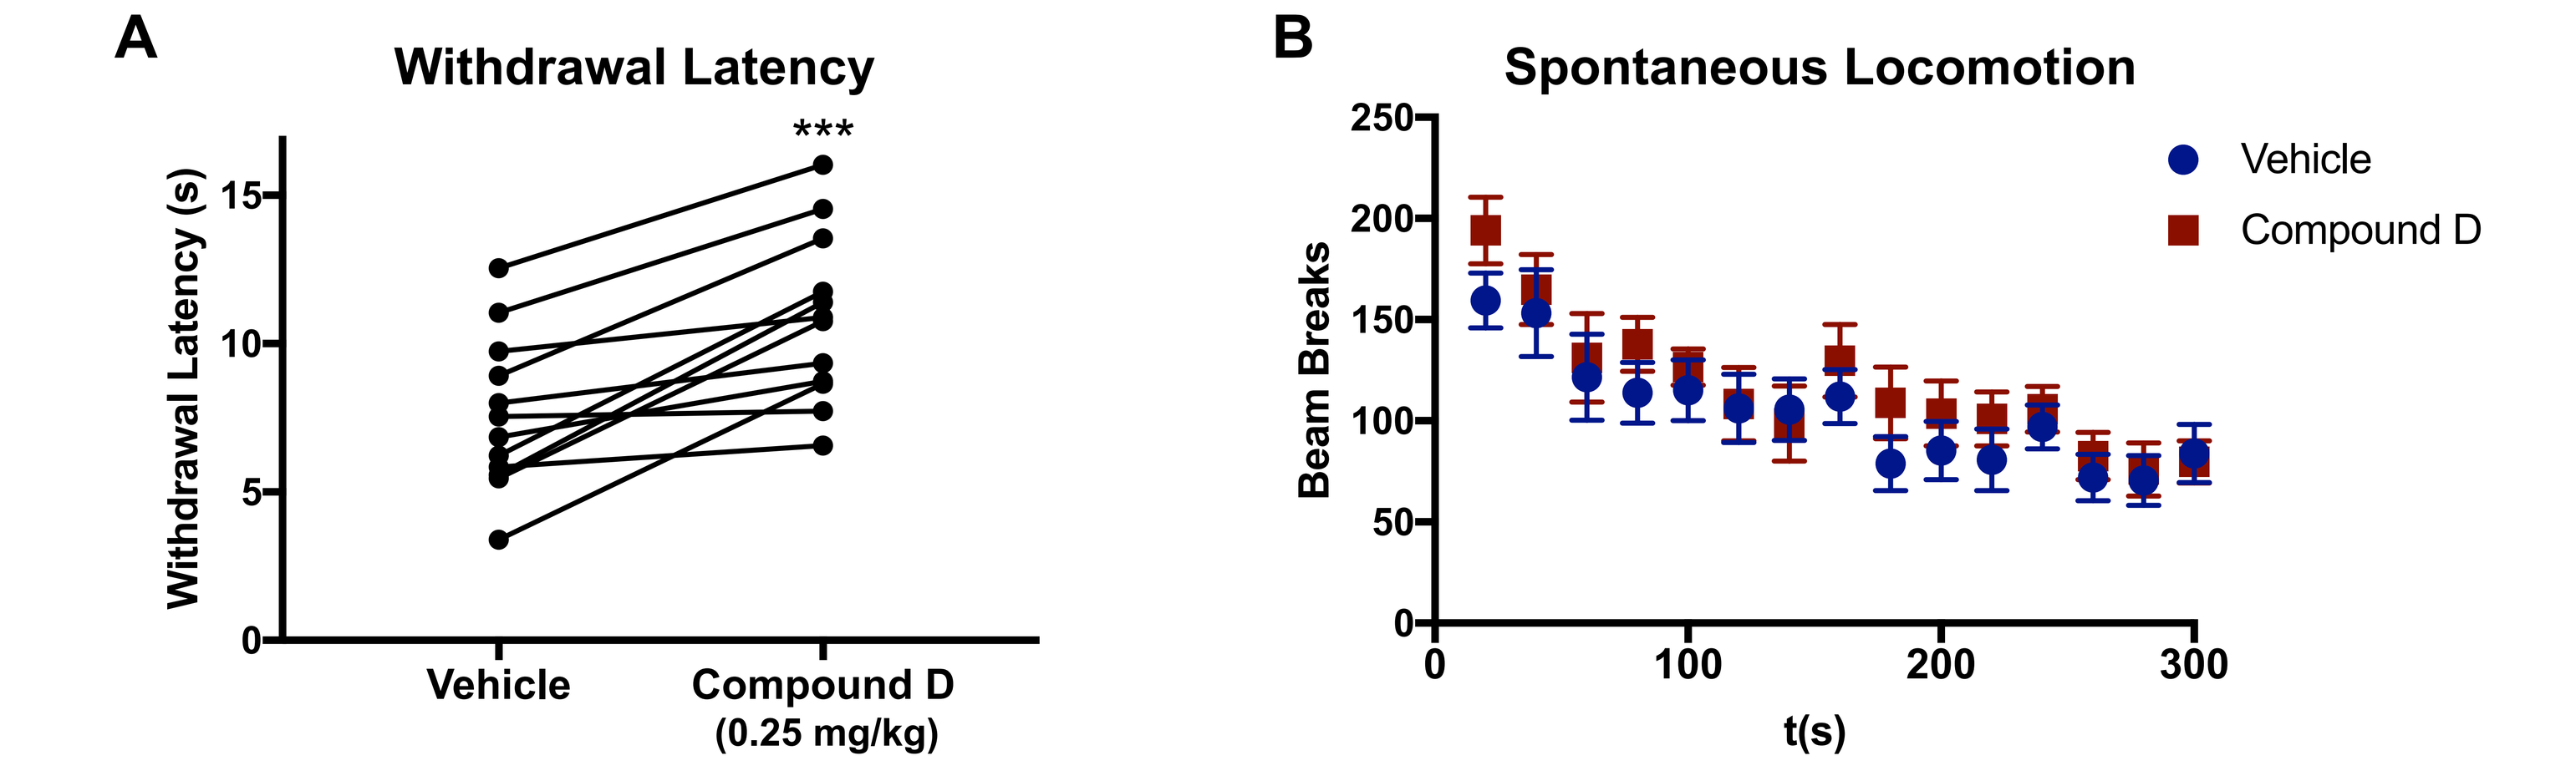
A Withdrawal Latency

Vehicle
Compound D (0.25 mg/kg)
B Spontaneous Locomotion
Vehicle
Compound D
Beam Breaks
t(s)

Figure 8. (A) Withdrawal latency in mice that were treated with SubQ vehicle or Compound D (0.25 mg/kg) 15 minutes prior to testing. *** - p<0.001. **(B)** Number of beam breaks over 5 minutes, for subjects following treatment with vehicle (blue) or Compound D (red.)

Conclusion

- SiteOne Therapeutics has discovered highly potent and selective state-independent Nav1.7 inhibitors.
- Compound C exhibited a Nav1.7 potency of under 100nM, and **selectivity of greater than 1000X over all other Nav1.x isoforms tested.**
- Surprisingly, Compound D was equipotent against mouse and human Nav1.7, suggesting that its potency and selectivity may be derived from an unknown binding pocket.
- Compounds A-C demonstrated evidence of analgesic efficacy in a non-human primate model of thermal pain. The compounds had no effect on heart rate, respiratory rate, body temperature, or blood oxygen saturation at any dose tested.
- In a preliminary study, Compound D decreased sensitivity to an aversive thermal stimulus in mice, while having no effect on spontaneous locomotion.
- **Evidence from these experiments support the hypothesis that selective small-molecule state-independent Nav1.7 inhibitors may be efficacious as pain therapeutics.**

About SiteOne Therapeutics

SiteOne Therapeutics is an early-stage biotechnology company founded in 2010 by Stanford University researchers and a biotech entrepreneur-scientist. SiteOne Therapeutics is headquartered in Bozeman, Montana with a research laboratory in the South San Francisco, California. Since its inception, SiteOne has been dedicated to developing novel pain therapeutics to safely, effectively and efficiently treat acute and chronic pain without the limitations of existing pain therapies, such as NSAIDs or opioids. The company's therapeutic candidates are highly selective sodium channel isoform 1.7 (Nav1.7) inhibitors based on naturally occurring small molecules. Given the urgent need for new, non-opioid solutions for managing pain, SiteOne is focused on advancing its lead product candidates for multiple therapeutic applications.

More information about SiteOne Therapeutics can be found at https://www.siteonetherapeutics.com/.

References

1) Cox JJ, Reimann F, Nicholas AK, et al (2006) An SCN9A channelopathy causes congenital inability to experience pain. **Nature** 444(7121):894-8.
2) Gingras J, Smith S, Matson DJ, et al (2014) Global Nav1.7 knockout mice recapitulate the phenotype of human congenital indifference to pain. **PLoS One** 9(9):e105895.
3) Walker JR, Novick PA, Parsons WH, et al (2012) Marked difference in saxitoxin and tetrodotoxin affinity for the human nociceptive voltage-gated sodium channel (Nav1.7). **Proc Natl Acad Sci USA** 109(44):18102-7.
4) Yeomans DC, Lu Y, Laurito CE, et al (2006) Recombinant herpes vector-mediated analgesia in a primate model of hyperalgesia. **Mol Ther** 13(3):589-97.

Disclosures

JB, AD, HP, DM, XZ, JM: SiteOne employees
DY, JDB: SiteOne cofounders and consultants

Acknowledgements

Grant Funding: NIH NINDS 5R44NS081887
USAMRAA W81XWH-17-1-072

We would like to thank Dr. Jennifer Lane, attending veterinarian, and the animal research technicians at Montana State University's Animal Resources Center for their assistance in the primate studies. We'd also like to thank Dr. Sheri Klas, Director of Nonclinical Sciences for SiteOne Therapeutics, for help on the behavioral studies with compounds C and D.