VGL101 RESCUES CSF1R DYSFUNCTION IN HUMAN MICROGLIA AND MACROPHAGES: EVALUATION OF IN VITRO TREM2 AGONISM IN MODELS OF A CSF1R-DEPENDENT LEUKODYSTROPHY

Daria Tchessalova, Kelley C. Larson, Frederick W. Gergits, Abigail Renoux, Matthew Figley, Christian Mirescu

Vigil Neuroscience, Cambridge, MA, USA

INTRODUCTION

Adult-onset leukoencephalopahy with axonal spheroids and pigmented glia (ALSP) is a rare, autosomal dominant disorder that is caused by loss of function mutations in the gene encoding for CSF1R (see poster from Papapetropoulos et al.).

These mutations result in microglial dysfunction and cell death. CSF1R and TREM2 share a common, convergent signaling pathway through phosphorylation of SYK.

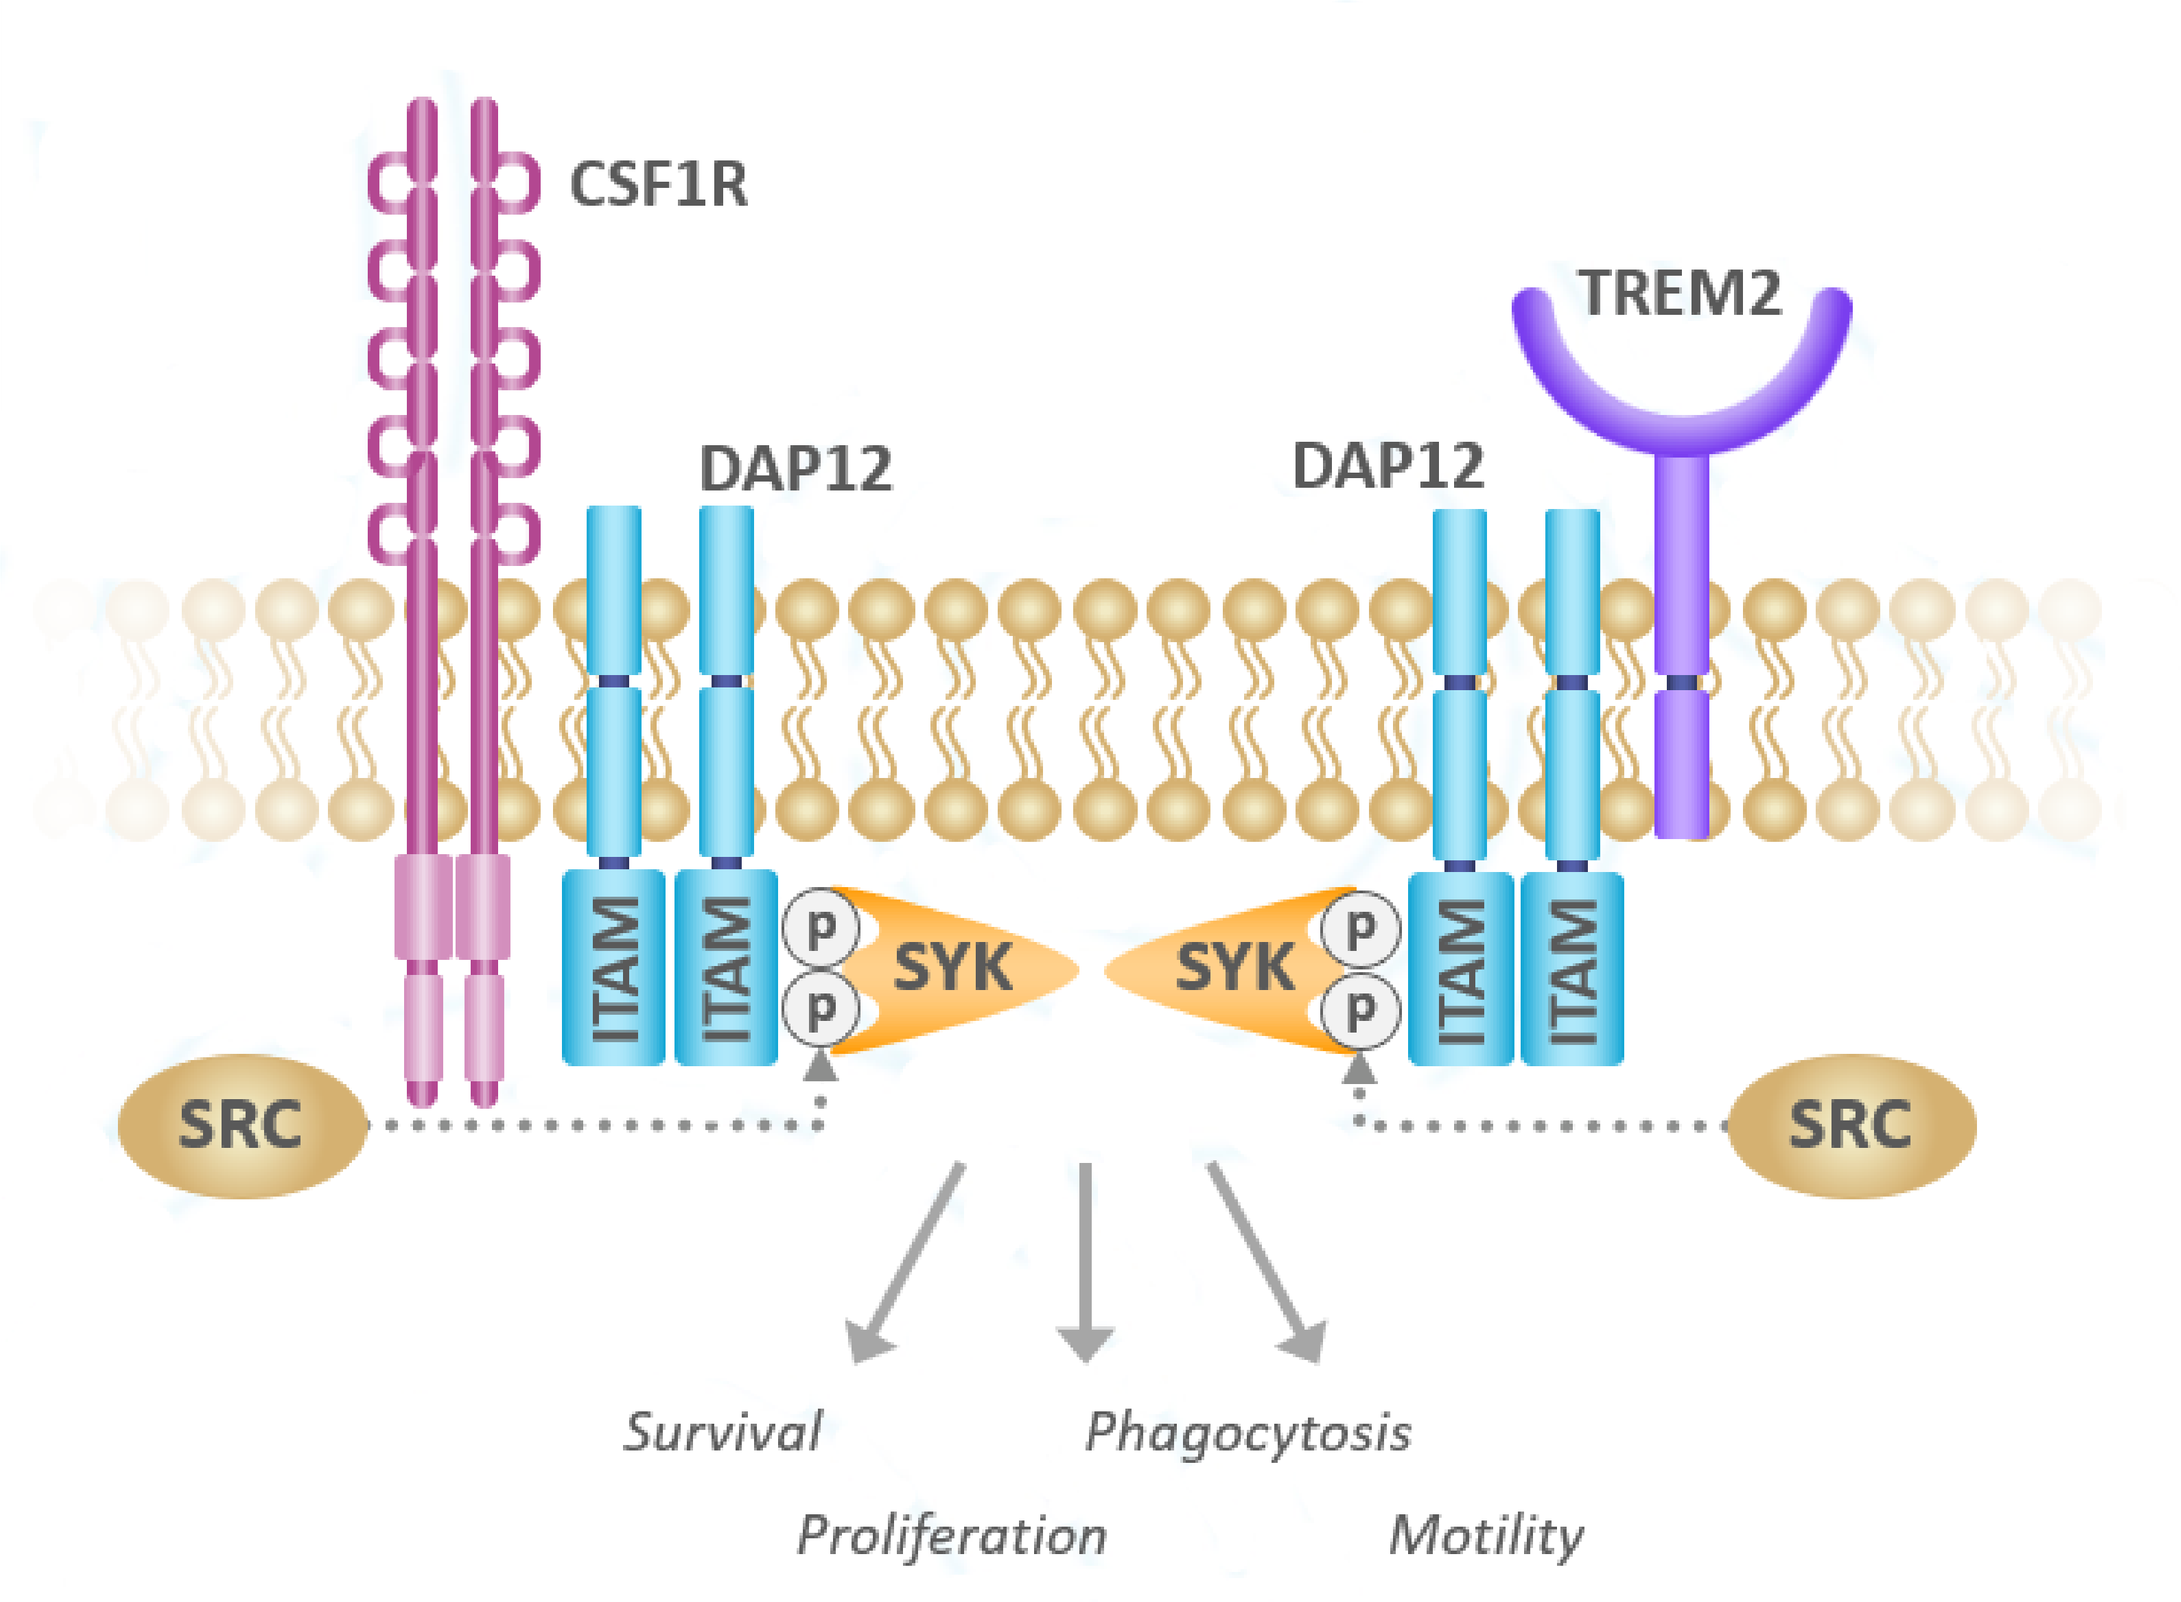

CSF1R
TREM2
DAP12
DAP12
ITAM
ITAM
ITAM
ITAM
p
p
SYK
SYK
p
p
SRC
SRC
Survival
Phagocytosis
Proliferation
Motility

Hypothesis: Agonism of TREM2 signaling will compensate for CSF1R loss of function in ALSP.

Objectives:

- Model ALSP in vitro utilizing healthy, human monocyte derived macrophages (MDM) and induced pluripotent stem cell-derived human microglia (iMGL)
- Model CSF1R haploinsufficiency through pharmacological inhibition (PLX5622) or CSF1R ligands withdrawal
- Determine ability of VGL101, a TREM2 agonist, to ameliorate effects of CSF1R dysfunction in these ALSP models

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com

MATERIALS AND METHODS

hMDM: Peripheral blood mononuclear cells (PBMC) or CD14+ isolated monocytes from Lonza. Cells were replated at 25,000 cells/well in 96 well plates coated with 0.4, 2.0, or 10 µg/mL of VGL101 or matching IgG control. 1 µM PLX5622 was immediately added to the cultures and incubated for 72-96 hours. Cells were imaged for 3 days using an IncuCyte S3 analyzer and assessed changes in morphology/area. Endpoint cell viability was measured using the Promega CellTiter-Glo assay.

iMGL: iCell microglia from FCDI were thawed on Matrigel-coated 6 well plates for 3 days in complete media. iMGLs were re-plated into 96-well plates, pre-coated with VGL101 or IgG isotype matched control, at 15,000 cells/well in three different media formulations: “complete media”, “withdrawal media” comprised of complete media without any growth factors, and “rescue media” containing withdrawal media with supplementation of only CSF1 and IL-34. PLX5622 was added to cells in complete media 24 hours after plating. After another 24 hours, apoptosis was assessed via caspase 3/7 dye in the IncuCyte SX3. Cell viability was measured at assay endpoint using CellTiter-Glo.

.

pSYK assay was performed using PerkinElmer Phospho-SYK (TYR 525/526) AlphaLISA kit according to manufacturer’s instructions. MDMs were treated with half-log increments of VGL101 or IgG control for 45 minutes after 6 days of differentiation, and iMGL were treated for 5 minutes, 2 days post re-plating into 96 well plates.

sCSF1R/sTREM2. For sCSF1R assay, cell culture plates were pre-coated with VGL101 or human IgG isotype control. Cell microglia were platted unto antibody pre-coated plates and incubated for 24 hrs. For sTREM2 assay, antibody was spiked into cell suspension. Soluble CSF1R and soluble TREM2 levels were measured using Abcam CSF1R and sTREM2 ELISA kits, respectively, per manufacturer protocol.

RESULTS

Figure 1: VGL101 is a Nanomolar Agonist of Phospho-SYK in Both iMGL and hMDM

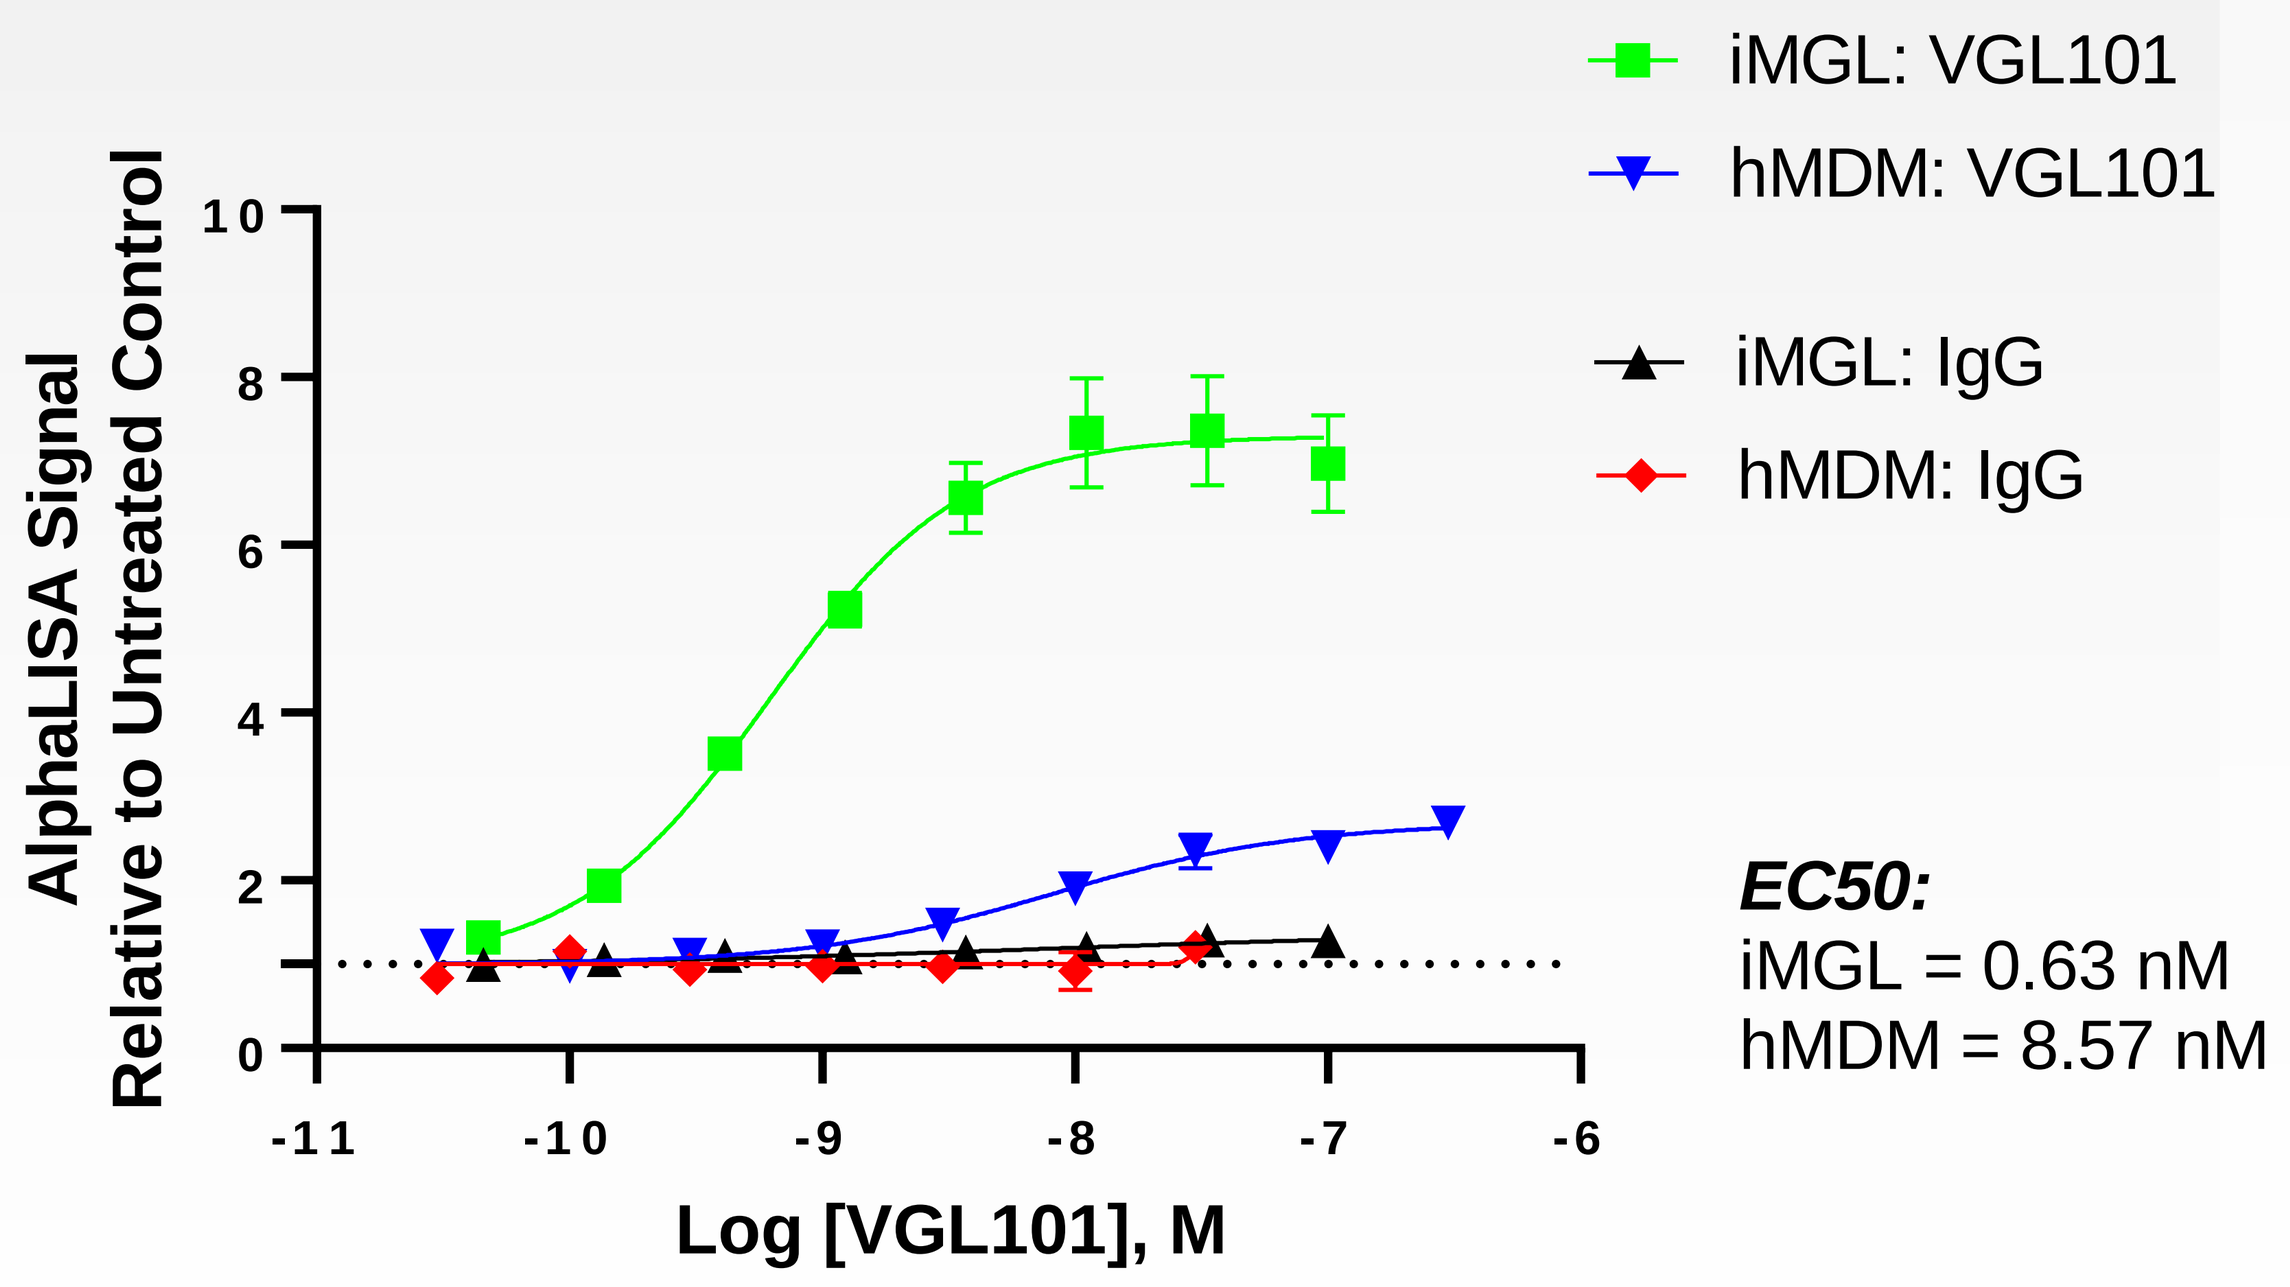

iMGL: VGL101
hMDM: VGL101
iMGL: IgG
hMDM: IgG
AlphaLISA Signal
Relative to Untreated Control
Log [VGL101], M
EC50:
iMGL = 0.63 nM
hMDM = 8.57 nM

iMGL: Data shown is mean of 6 independent experiments,
hMDM: Data shown is mean of 2 independent experiments.
All data shown +/- SEM and normalized to average of matched IgG control

RESULTS

Figure 2. VGL101 Rescues Cellular Morphology in iMGL in Response to CSF1R Inhibition by PLX5622(A) or CSF1/IL34 Withdrawal (B)

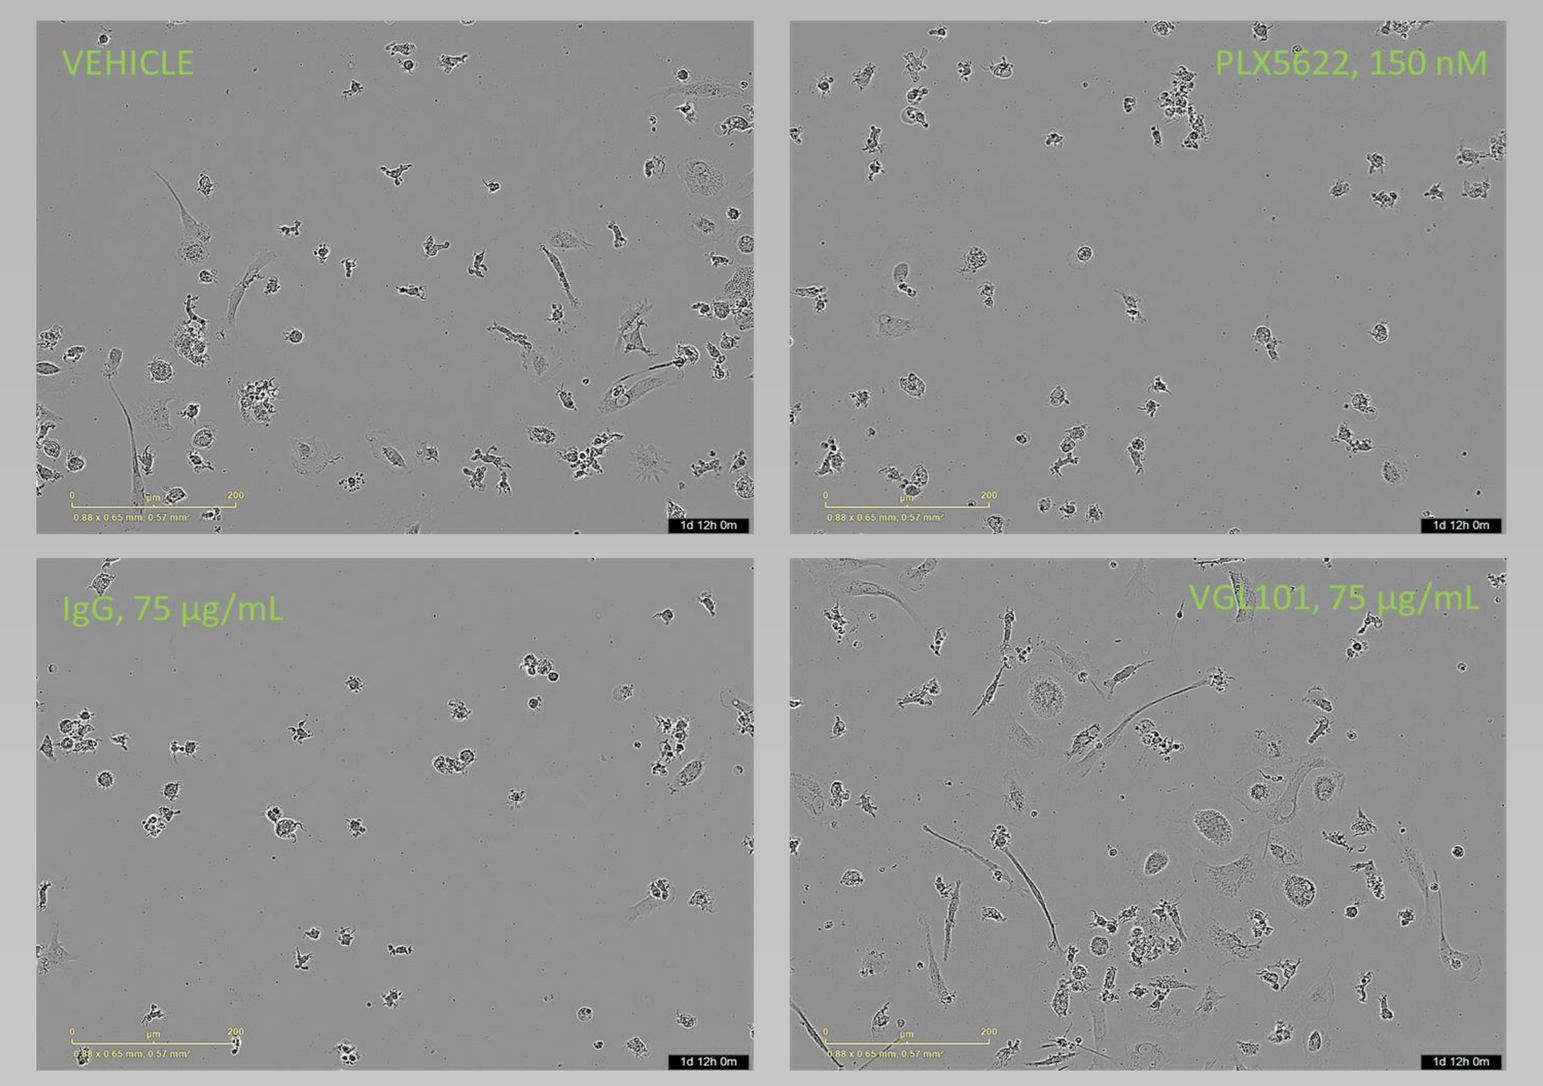

VEHICLE
PLX5622, 150 nM
IgG, 75 µg/mL
VGL101, 75 µg/mL

Representative Images (20X)

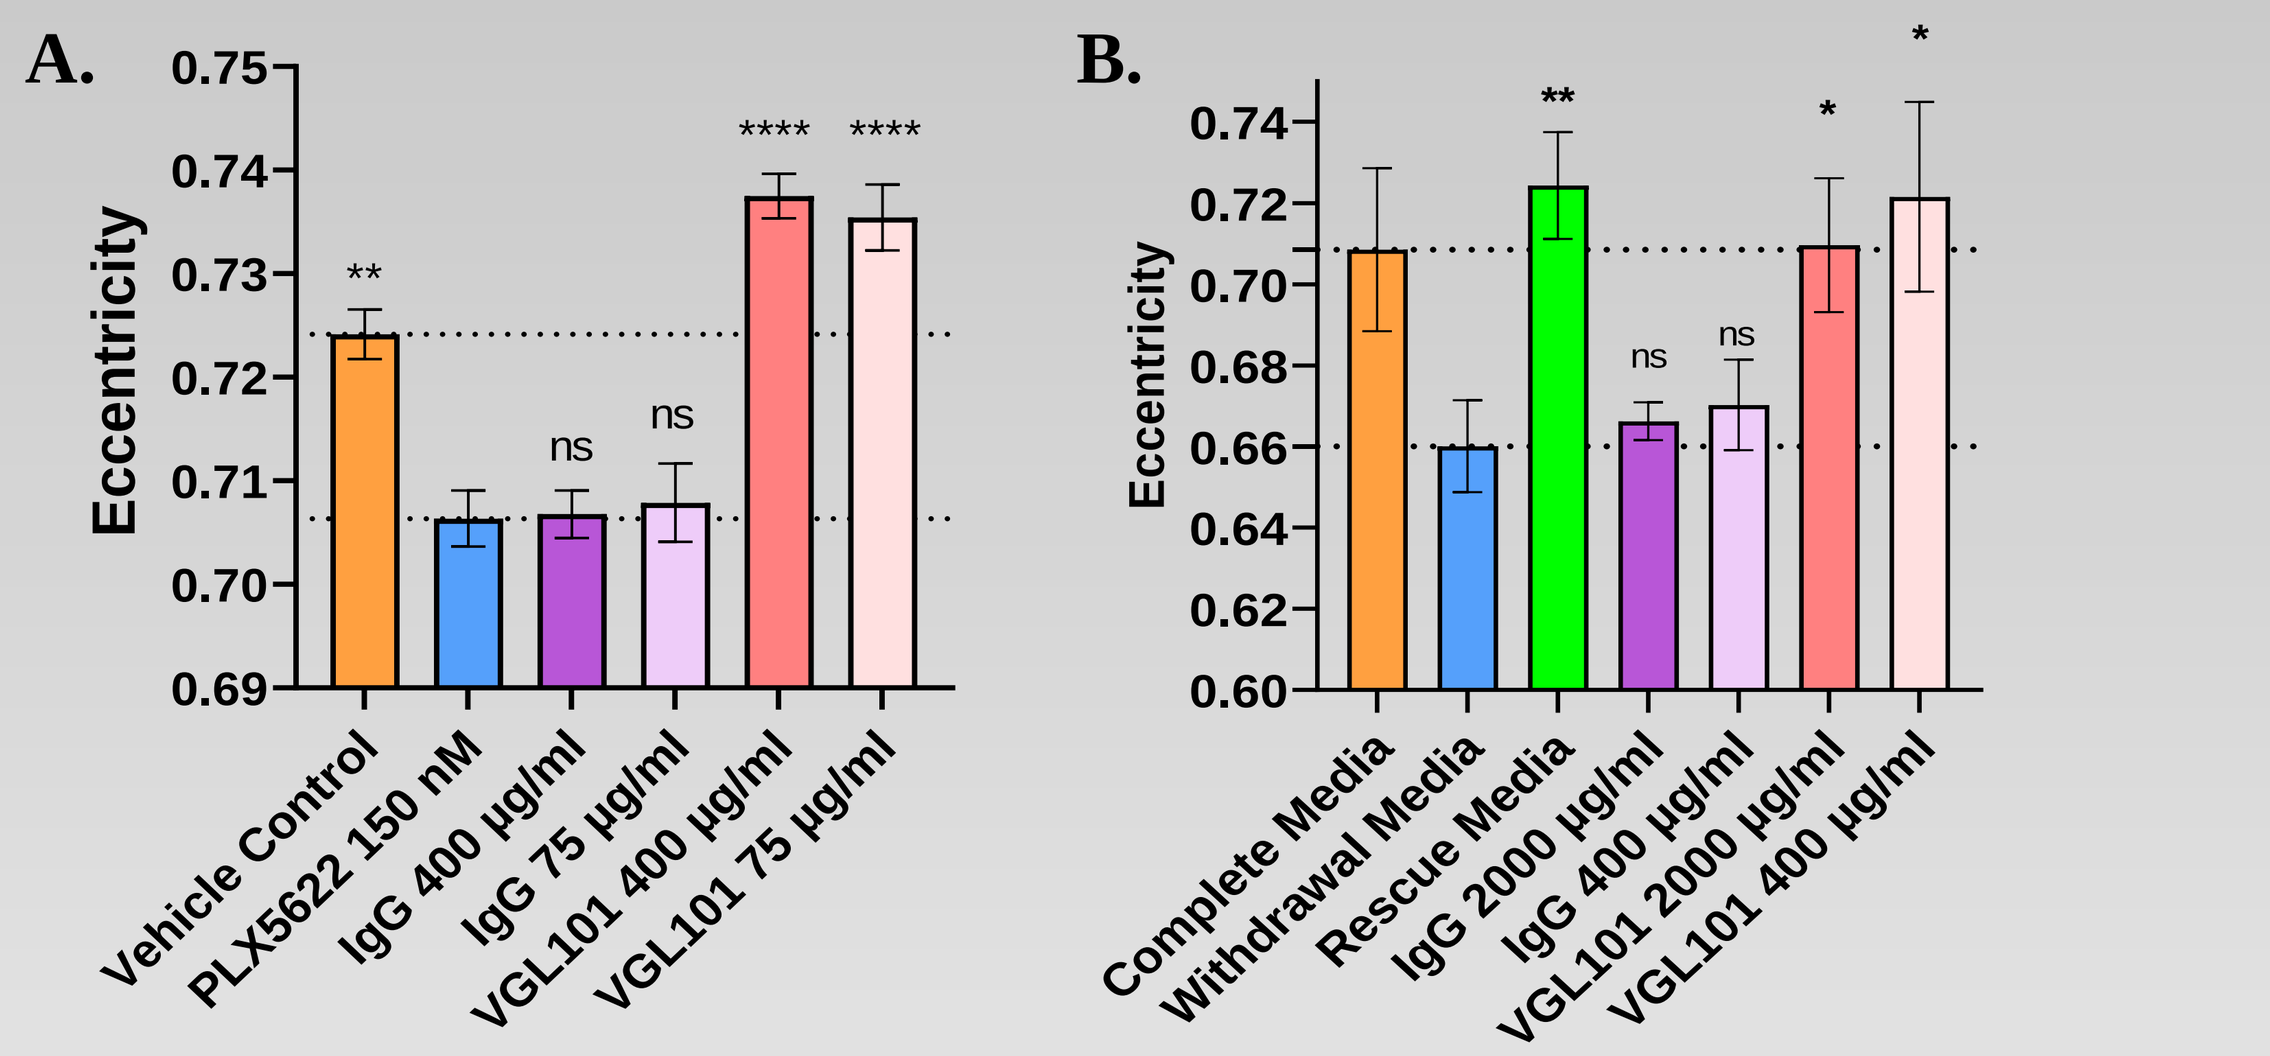

A.
Eccentricity
Vehicle Control
PLX5622 150 nM
IgG 400 µg/ml
IgG 75 µg/ml
VGL101 400 µg/ml
VGL101 75 µg/ml
B.
Eccentricity
Complete Media
Withdrawal Media
Rescue Media
IgG 2000 µg/ml
IgG 400 µg/ml
VGL101 2000 µg/ml
VGL101 400 µg/ml

Data are shown as mean eccentricity values +/- SEM from 3-4 independent experiments. P-values are as determined either using Ordinary One-Way ANOVA with multiple comparisons, or using two-tailed, paired T-tests
**** $p < 0.0001$ ** $p < 0.01$, * $p < 0.05$

Figure 3. VGL101 Rescues Apoptosis in iMGL in Response to CSF1R Inhibition by PLX5622 (A) or CSF1/IL34 Withdrawal (B)

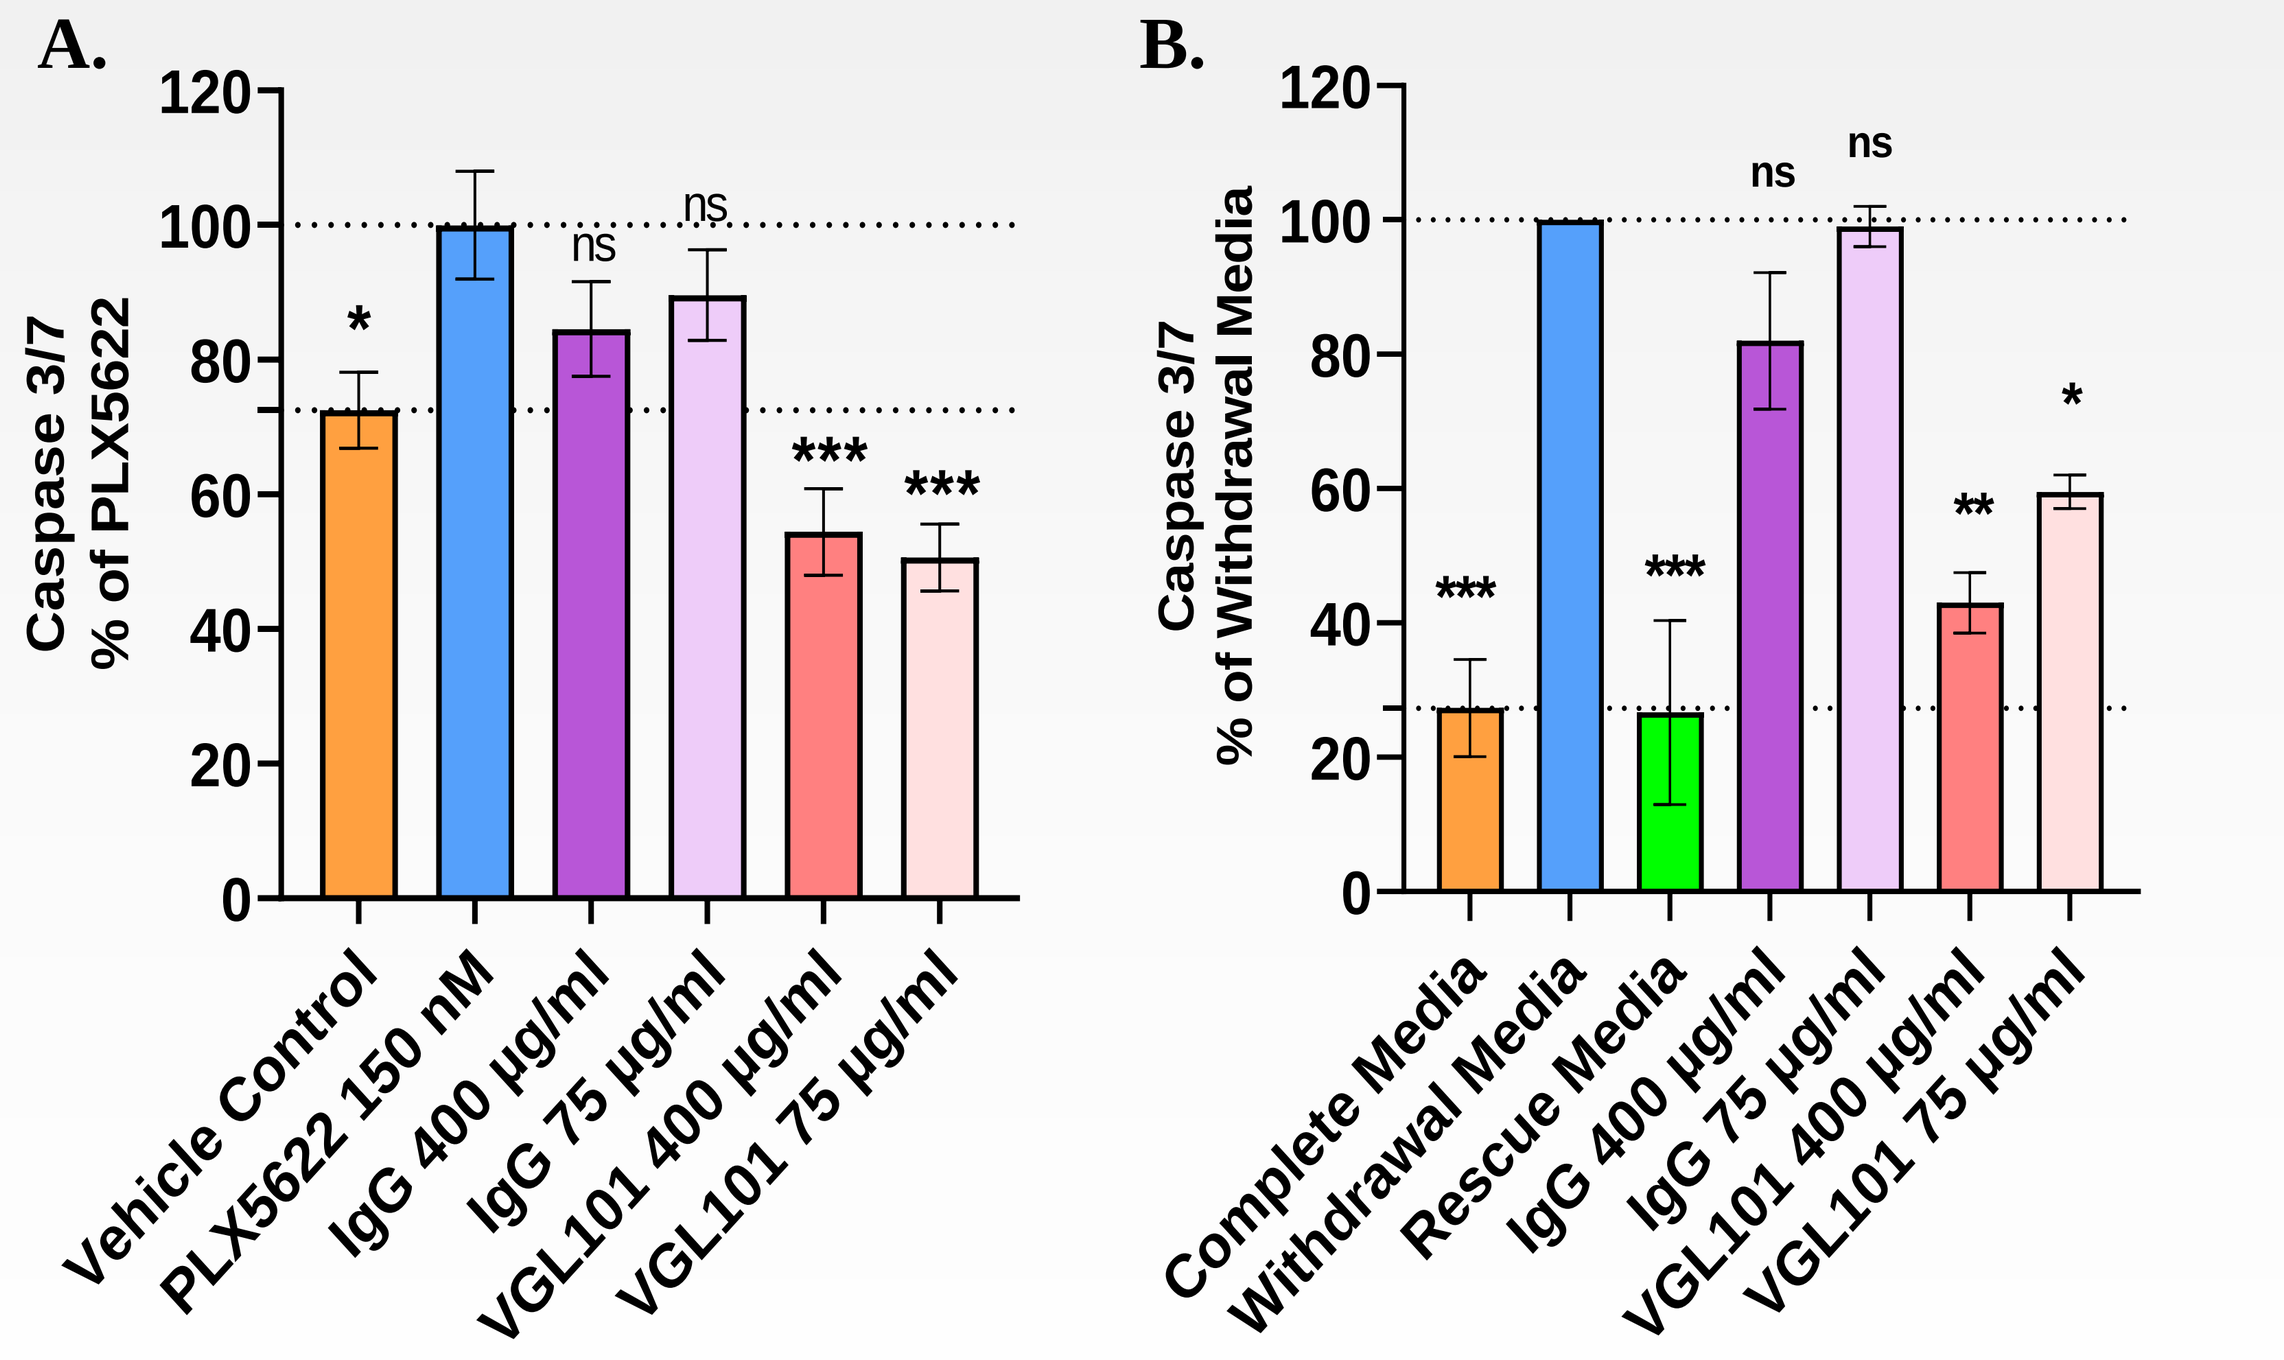

A.
Caspase 3/7
% of PLX5622
Vehicle Control
PLX5622 150 nM
IgG 400 µg/ml
IgG 75 µg/ml
VGL101 400 µg/ml
VGL101 75 µg/ml
B.
Caspase 3/7
% of Withdrawal Media
Complete Media
Withdrawal Media
Rescue Media
IgG 400 µg/ml
IgG 75 µg/ml
VGL101 400 µg/ml
VGL101 75 µg/ml

Each sample was normalized to its matched control. Data are shown as mean +/- SEM from 3-4 independent experiments. P-values are determined by Ordinary One-Way ANOVA with Multiple Comparisons. *** $p < 0.001$, ** $p < 0.01$, *$p < 0.05$

RESULTS

Figure 4: VGL101 Rescues Viability in iMGL in Response to CSF1R Inhibition by PLX5622 (A) or CSF1/IL34 Withdrawal (B)

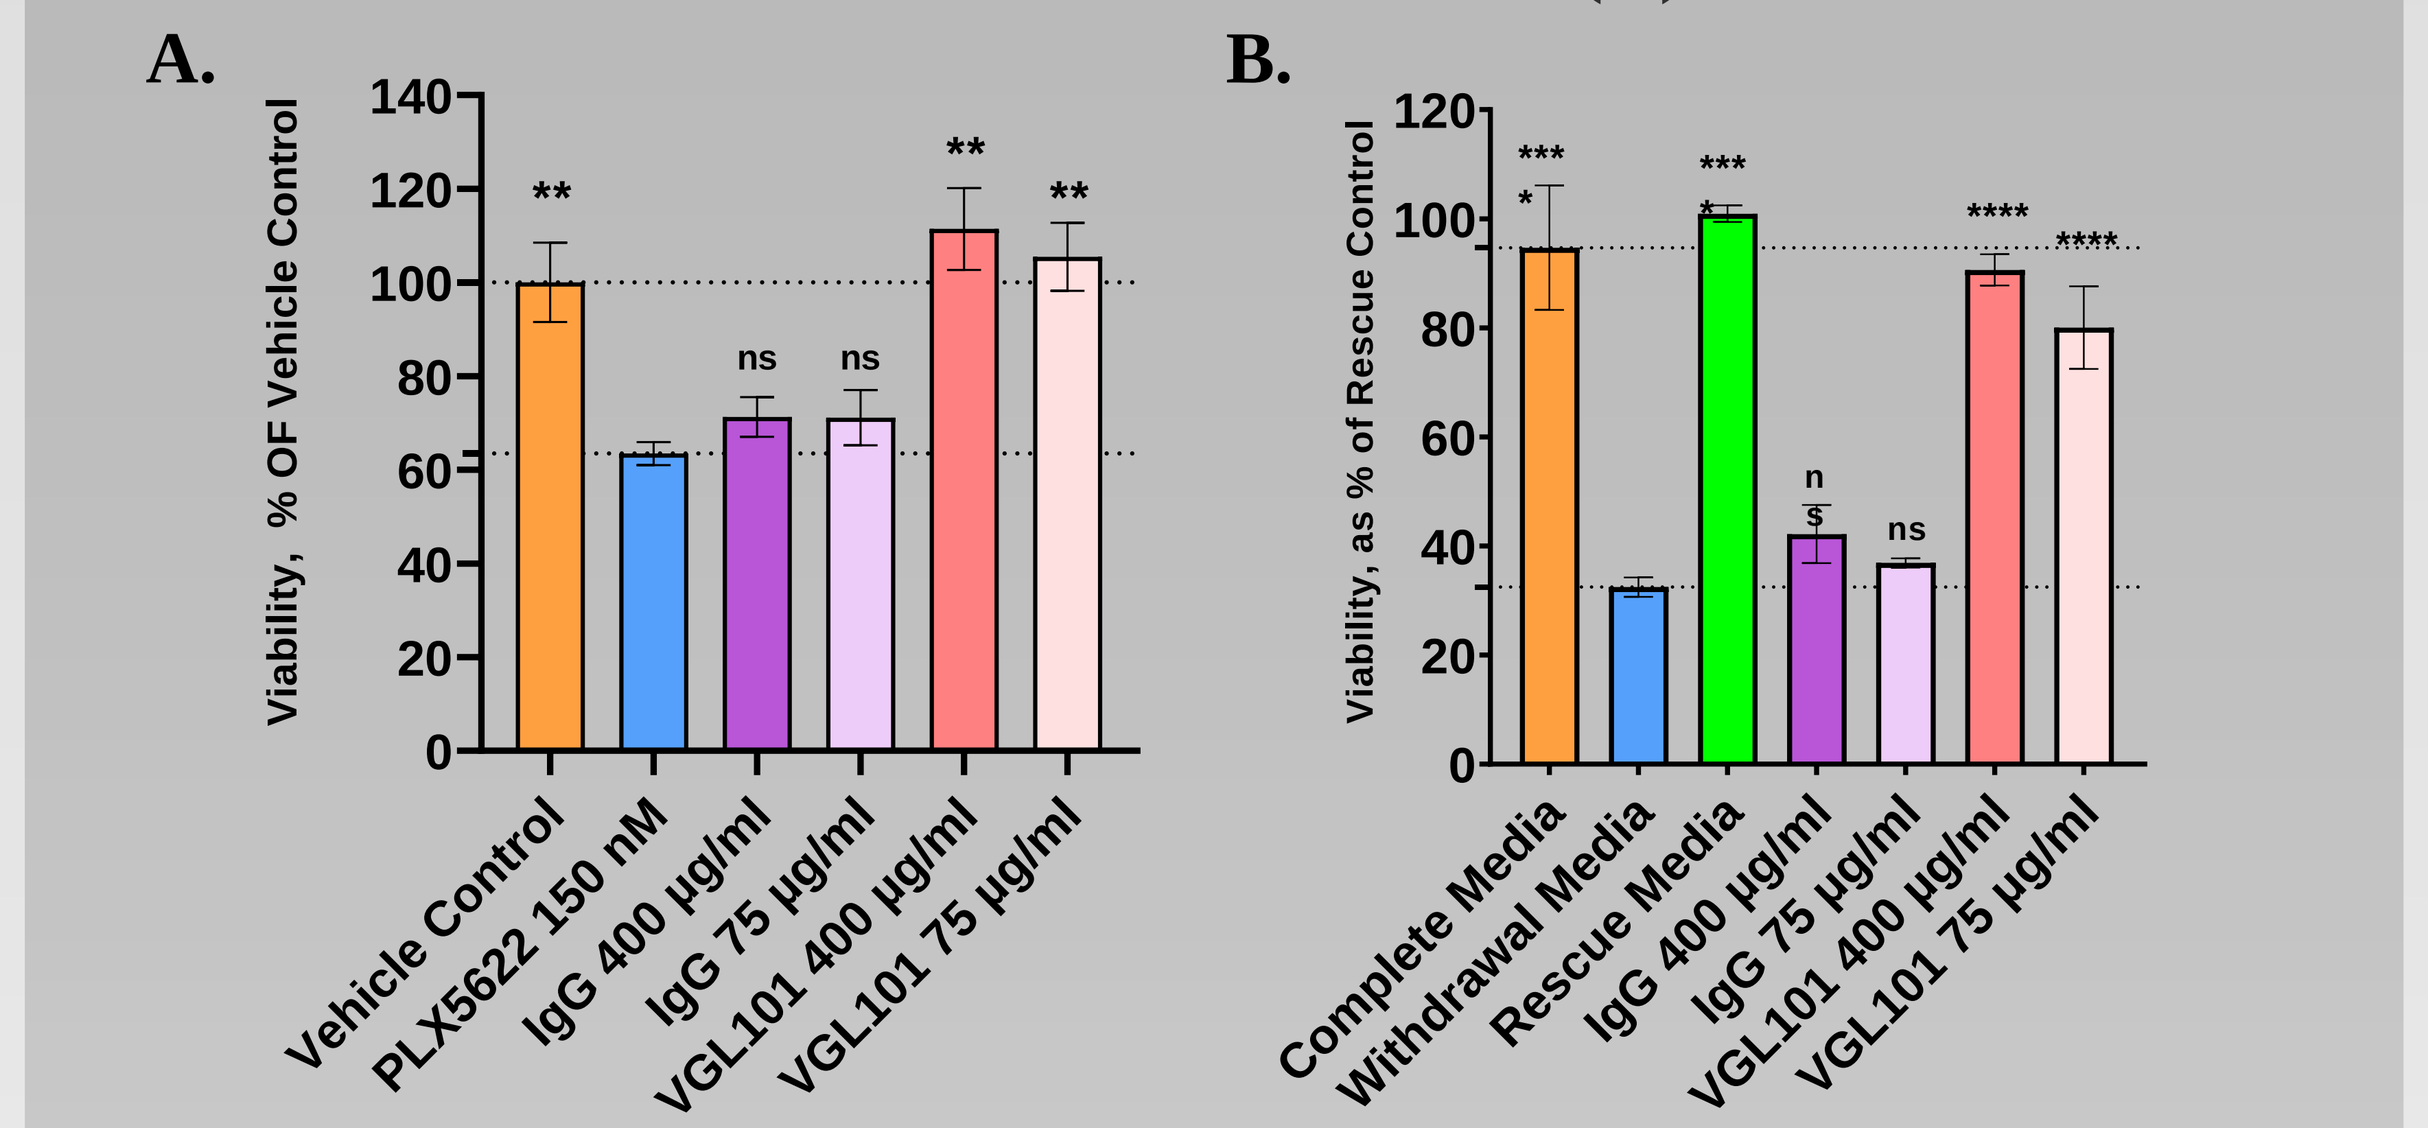

A.
Viability, % OF Vehicle Control
Vehicle Control
PLX5622 150 nM
IgG 400 µg/ml
IgG 75 µg/ml
VGL101 400 µg/ml
VGL101 75 µg/ml
B.
Viability, as % of Rescue Control
Complete Media
Withdrawal Media
Rescue Media
IgG 400 µg/ml
IgG 75 µg/ml
VGL101 400 µg/ml
VGL101 75 µg/ml

Each sample was normalized to its matched control. Data are shown as mean +/- SEM from 3-4 independent replicates. P-values are as determined by Ordinary One-Way ANOVA with Multiple Comparisons. **** $p < 0.0001$, ** $p < 0.01$

Figure 5: VGL101 Rescues PLX5622 Mediated CSF1R Kinase Inhibition Induced Cell Death (A) and Morphology (B) in hMDM

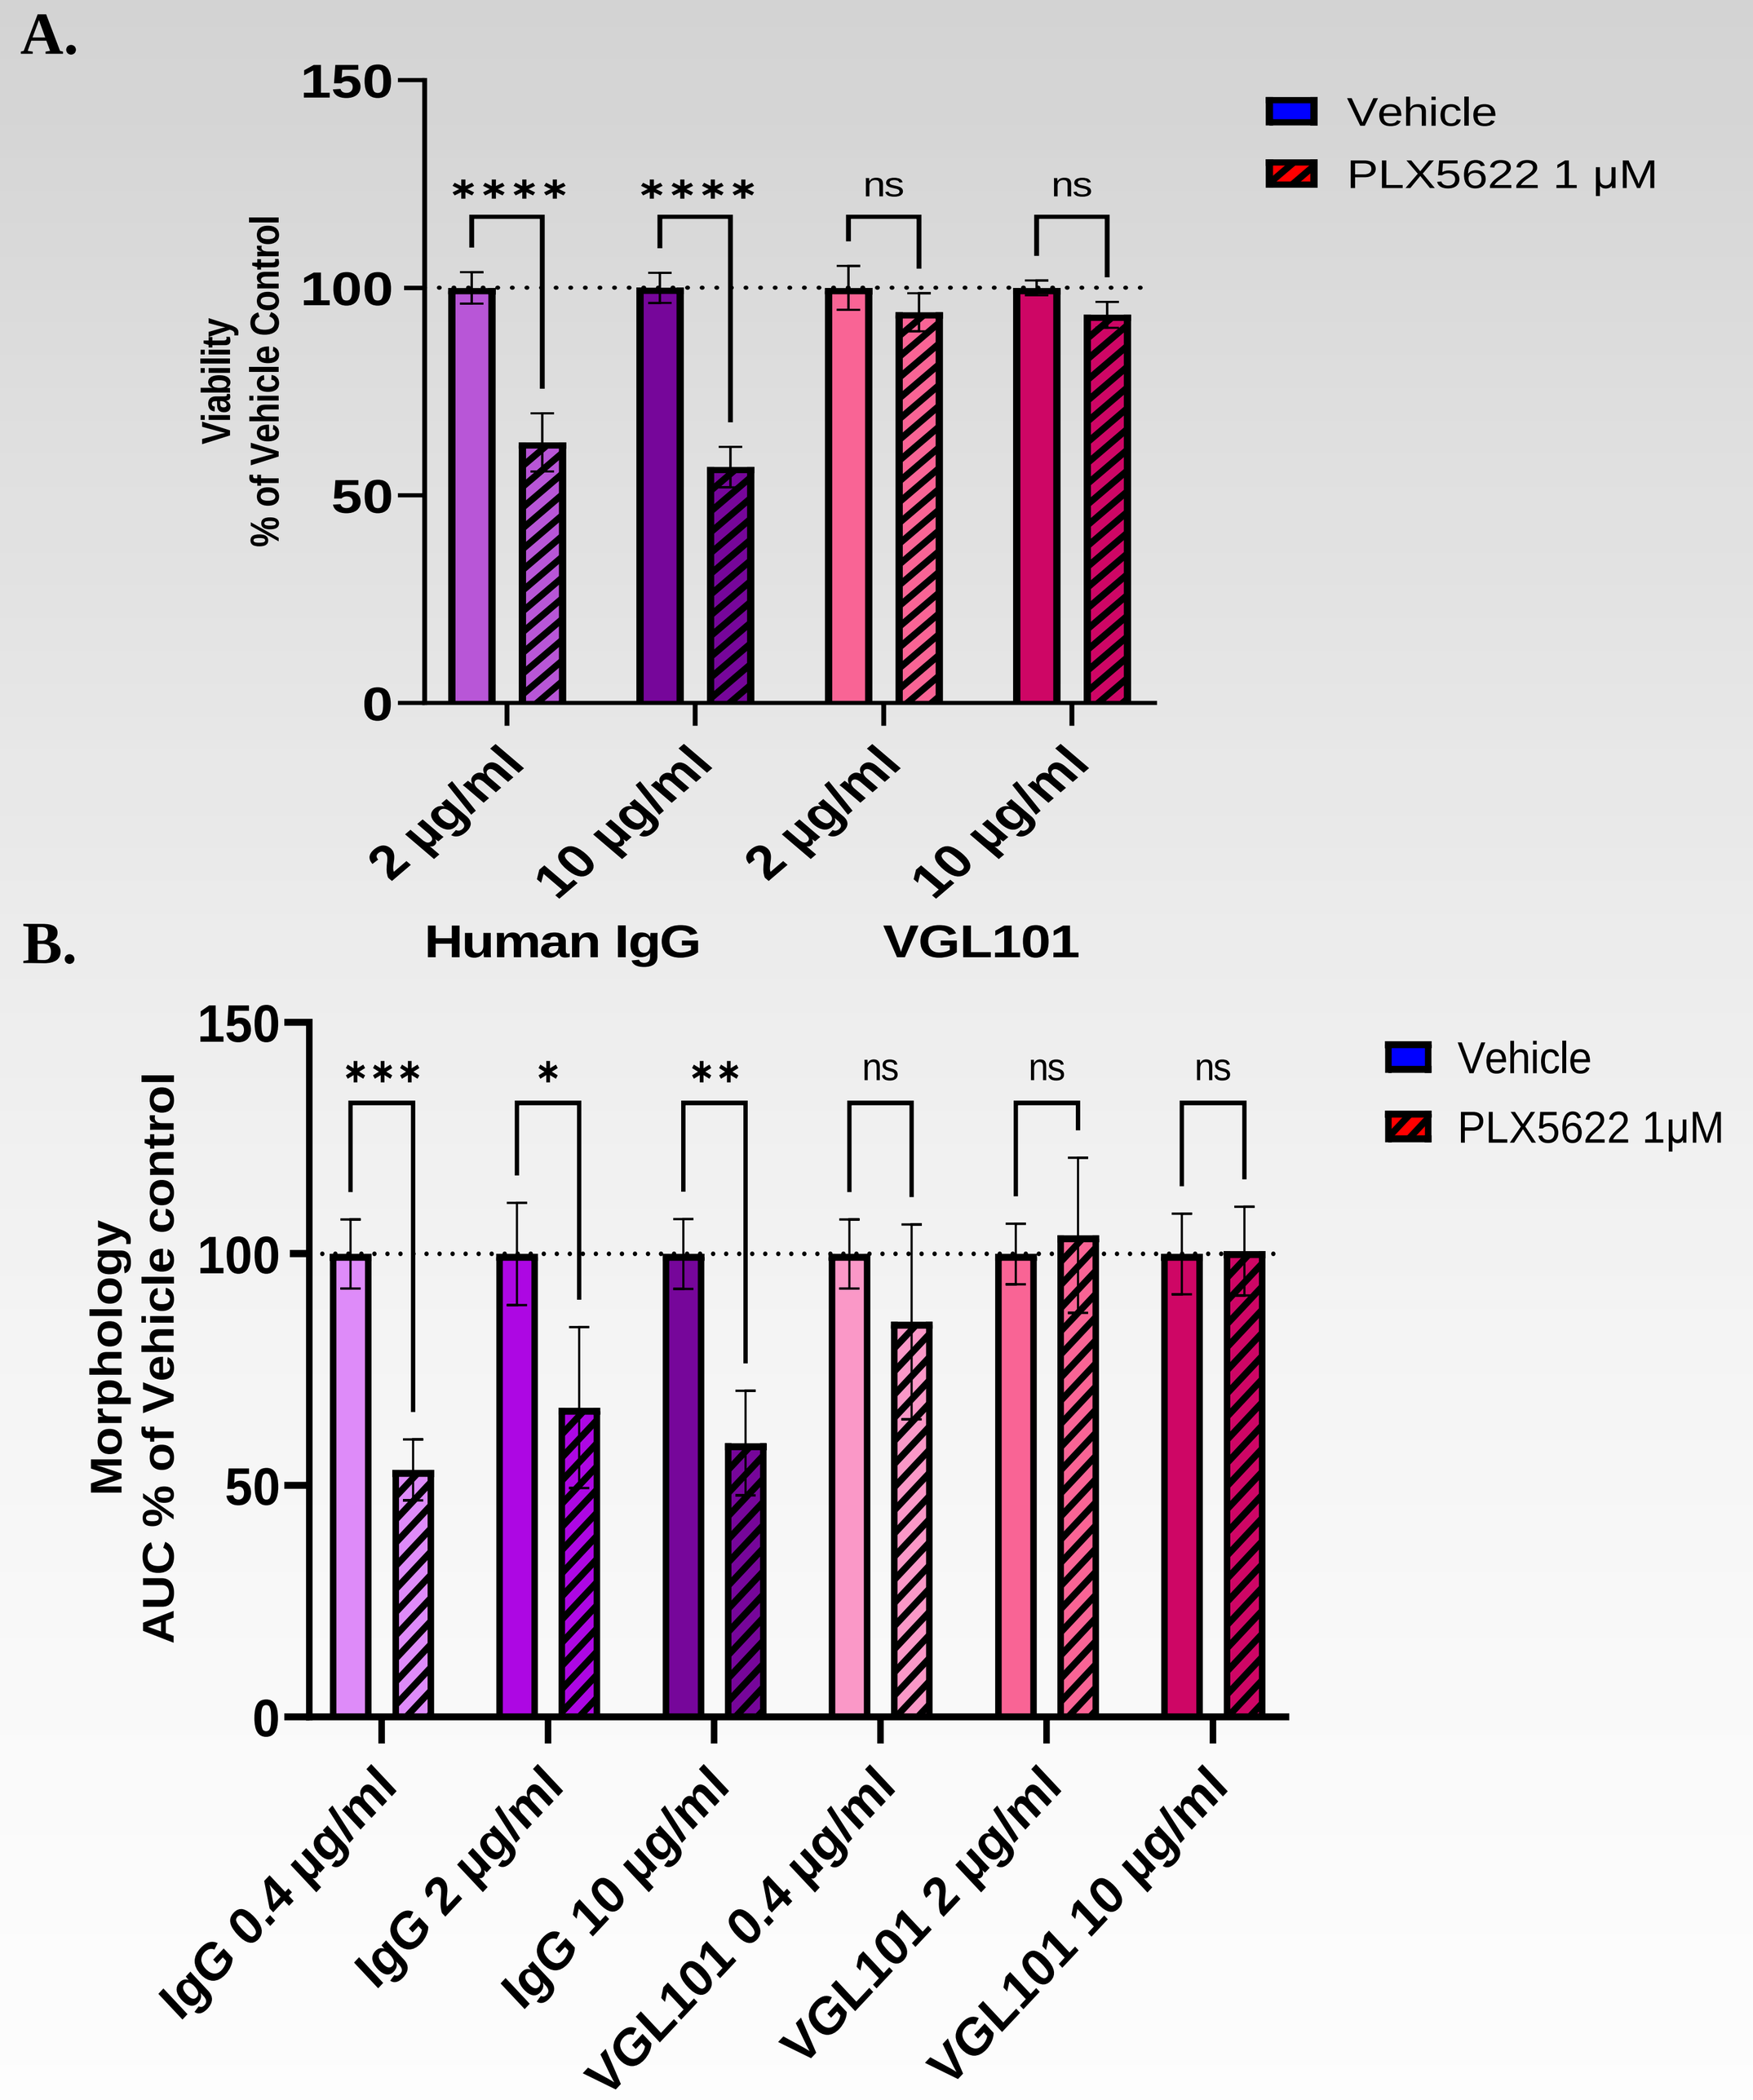

A.
Vehicle
PLX5622 1 µM
Viability
% of Vehicle Control
2 µg/ml
10 µg/ml
2 µg/ml
10 µg/ml
Human IgG
VGL101
B.
Vehicle
PLX5622 1µM
Morphology
AUC % of Vehicle control
IgG 0.4 µg/ml
IgG 2 µg/ml
IgG 10 µg/ml
VGL101 0.4 µg/ml
VGL101 2 µg/ml
VGL101 10 µg/ml

A) CellTiterGlo viability results. Data are shown as mean of raw signal +/- SEM from 3 wells per condition from one representative experiment.
B) Data are shown as area under the curve for percent high area (>1000µm^2), high eccentricity (>0.83) cells measured every 8 hours for 3 days. Mean +/- SD from 3 images/well, 3 wells/condition. P-values are determined by two-way ANOVA and posthoc Sidak’s test. * $p < 0.05$, ** $p < 0.01$, *** $p < 0.001$, **** $p < 0.0001$

RESULTS

Figure 6. VGL101 Increases sCSF1R and Decreases sTREM2

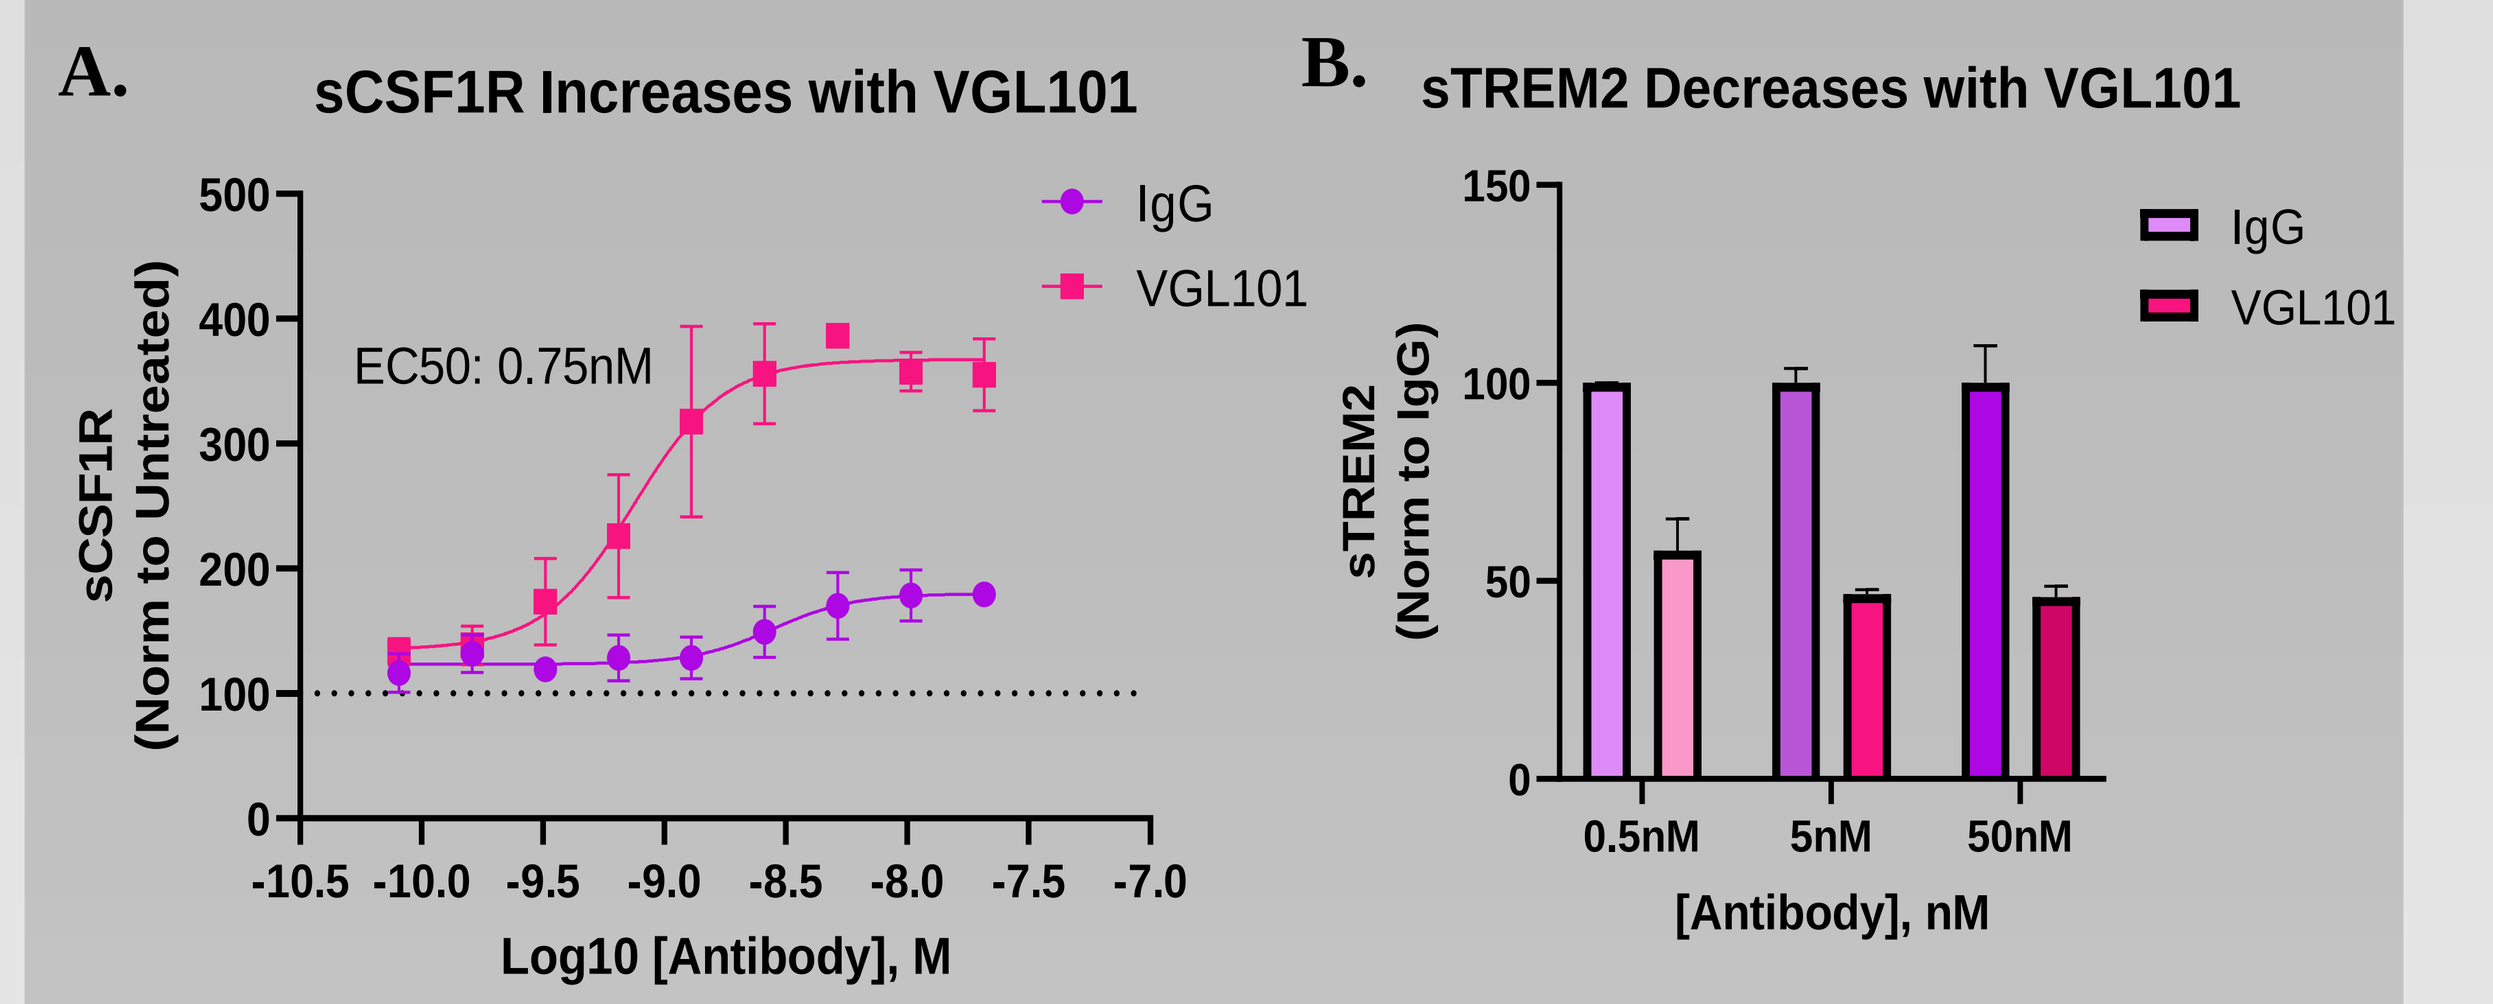

A.
sCSF1R Increases with VGL101
IgG
VGL101
EC50: 0.75nM
sCSF1R
(Norm to Untreated)
Log10 [Antibody], M
B.
sTREM2 Decreases with VGL101
IgG
VGL101
sTREM2
(Norm to IgG)
0.5nM
5nM
50nM
[Antibody], nM

Each sample was normalized to IgG control.
Data are shown as mean +/- SEM from 2 independent experiments.

CONCLUSIONS

Vigil Neuro was able to model a CSF1R-dependent leukodystrophy in two independent human cell systems: hMDM and iMGL.

Key findings include:

- CSF1R inhibition resulted in decreased viability, increased caspase 3/7 activation, and an altered morphology.
- VGL101 treatment induced phosphorylation of SYK in both models, demonstrating agonism of the TREM2 receptor.
- VGL101 rescued disease phenotypes in both cell types and in two models of CSF1R inhibition.
- Increases in sCSF1R and decreases in sTREM2 upon VGL101 treatment in iMGLs may inform target engagement studies

Together, these data demonstrate that TREM2 agonism is a viable mechanism to compensate for CSF1R dysfunction in vitro, supporting further evaluation of VGL101 as a viable therapeutic for ALSP.

REFERENCES

1. Stanley ER, Chitu V. CSF1 receptor signaling in myeloid cells. Cold Spring Harb Perspect Biol. 2014;6(6):a021857. Published 2014 Jun 2. doi:10.1101/cshperspect.a021857
2. Ellwanger DC, Wang S, Brioschi S, Shao Z, Green L, Case R, et al. Prior activation state shapes the microglia response to antihuman TREM2 in a mouse model of Alzheimer's disease. Proc Natl Acad Sci U S A. 2021 Jan 19;118(3):e2017742118. doi: 10.1073/pnas.2017742118. PMID: 33446504; PMCID: PMC7826333.
3. Schlepckow K, Monroe KM, Kleinberger G, Cantuti-Castelvetri L, Parhizkar S, Xia D, et al. Enhancing protective microglial activities with a dual function TREM2 antibody to the stalk region. EMBO Mol Med. 2020;12(4):e11227. doi:10.15252/emmm.201911227
4. Otero K, Turnbull IR, Poliani PL, et al. Macrophage colony-stimulating factor induces the proliferation and survival of macrophages via a pathway involving DAP12 and beta-catenin. Nat Immunol. 2009;10(7):734-743. doi:10.1038/ni.1744

CONTACT

Sarah Carmody, VP Investor Relations and Corporate Communications,
scarmody@vigilneuro.com

Daria Tchessalova, Senior Scientist at Vigil Neuroscience,
dtchessalova@vigilneuro.com