Clinically Relevant Reductions in FeNO Reported After 10-day Treatment With KN-002 in Subjects With Mild Asthma

Singh D[1], Young J[2], O'Brien C[2], Burgess G[3], Morgan F[2]

[1] Medicines Evaluation Unit and University of Manchester, Manchester UK, [2] Kinaset Therapeutics Inc. US, [3] GCB Consulting Ltd., UK

Introduction

- The JAK-STAT pathway mediates activity of multiple pro-inflammatory cells, cytokines and chemokines implicated in the pathogenesis of non-eosinophilic and eosinophilic severe asthma.
- KN-002 is a potent and balanced pan-JAK inhibitor[1], formulated as a dry powder for delivery via inhalation.
- Delivery of KN-002 results in high lung bioavailability, low plasma exposure, inhibition of Th1/2/17 pro-inflammatory mediators, attenuation of STAT phosphorylation levels and suppression of house dust mite induced airway hyper-responsiveness[2,3].

Study Objectives

Primary

- Assess the **SAFETY AND TOLERABILITY** of multiple ascending doses of **INHALED** KN-002 in subjects with stable mild asthma

Secondary

- Changes in **FeNO** from Day 1 pre-dose to Day 10 and % subjects with Day 10 FeNO levels <25 ppb
- Characterisation of **PLASMA** pharmacokinetics (PK) via C_{max}, T_{max}, $T_{1/2}$, area under the curve and accumulation ratio for C_{max} and area under the curve

Study Methods

Study Design

- Part 2 of a 4-part Phase 1b, randomized, double-blinded, placebo-controlled study (NCT05006521)
- Subjects in clinic for 12 days; follow-up at Day 17
- **10-DAY TREATMENT PERIOD**

Doses/regimen (6 active:2 placebo)

- 0.6 mg QD, 2 mg QD, 4 mg BID and 8 mg BID

Subjects

- Aged 18-65 years with physician diagnosis of asthma
- **NAÏVE TO ICS THERAPY WITH FeNO ≥30 PPB**
- Body mass index ≥ 18 and ≤ 35 kg/m²
- Pre-bronchodilator FEV_1 ≥ 70% predicted

Study Results

Table 1: Study Demographics

Baseline parameter (mean)	Overall (ITT) Population	
	Active (n=24)	Placebo (n=8)
Age (years)	35.5	47.5
Gender (% male/female)	83/17	88/13
BMI (kg/m²)	26	27
FeNO (ppb)	74.5	48.2
Pre-dose FEV_1 (% pred)	85	80
Blood eosinophil count (10^6/L)	323	229

Safety

- No deaths, serious or severe adverse events (AE), dose changes or withdrawals
- **ALL AES MILD OR MODERATE AND UNRELATED TO TREATMENT.**
- No treatment related or clinically important hematologic or chemistry findings.

Pharmacokinetics

- Dose proportional PK profile (AUC and C_{max})
- **MAXIMUM UNBOUND KN-002 PLASMA CONCENTRATIONS BELOW PHARMACOLOGICALLY ACTIVE LEVELS[1]**
- Steady state PK achieved by Day 3
- Minimal accumulation in plasma after 10-day repeat dose period (C_{max} ratio 1.1 to 1.9, AUC_{TAU} ratio 1.2 to 1.8)
- Main metabolite at 1% of parent in plasma

Table 2: Mean KN-002 Plasma PK Parameters at Day 10

Parameter	0.6 mg QD (n=6)	2 mg QD (n=6)	4 mg BID (n=6)	8 mg BID (n=6)
C_{max} (ng/ml)	2.0	6.5	31.5	60.4
AUC_{0-12} (ng.h/ml)	16.1	55.2	236.8	458.2
T_{max} (hours)	2	3	2	2
$T_{1/2}$ (hours)	9.2	8.4	14.1	20.6

Pharmacodynamics

- **CLINICALLY RELEVANT (>20%) LS MEAN PLACEBO-CORRECTED FeNO REDUCTIONS, FROM DAY 1 PRE-DOSE TO DAY 10 MEAN, ACHIEVED WITH 2 mg QD (-27.9% [95% CI,-56.6, +0.8]), 4 mg BID (-28.4% [95% CI, -55.3,-1.5]) AND 8 mg BID (-24.1% [95% CI, -50.1, +1.9]) REGIMENS**
- 0.6 mg QD established as minimally active dose (LS mean placebo corrected reduction -7.4% [95% CI, -33.5,+18.7])
- Clinically relevant placebo-corrected reductions achieved as quickly as Day 1
- FeNO reduction continues to at least Day 10 and is maintained post 2-day no treatment period
- FeNO values revert to near Day 1 pre-dose levels after a 7-day no treatment period
- **67% OF SUBJECTS RANDOMISED TO 4 mg BID ACHIEVE NORMAL FeNO VALUES (<25 PPB) AT DAY 10**
- Maximal PD effect achieved with 4 mg BID regimen; no additional benefit with 8 mg BID regimen

Figure 1: FeNO change from Day 1 pre-dose to Day 17

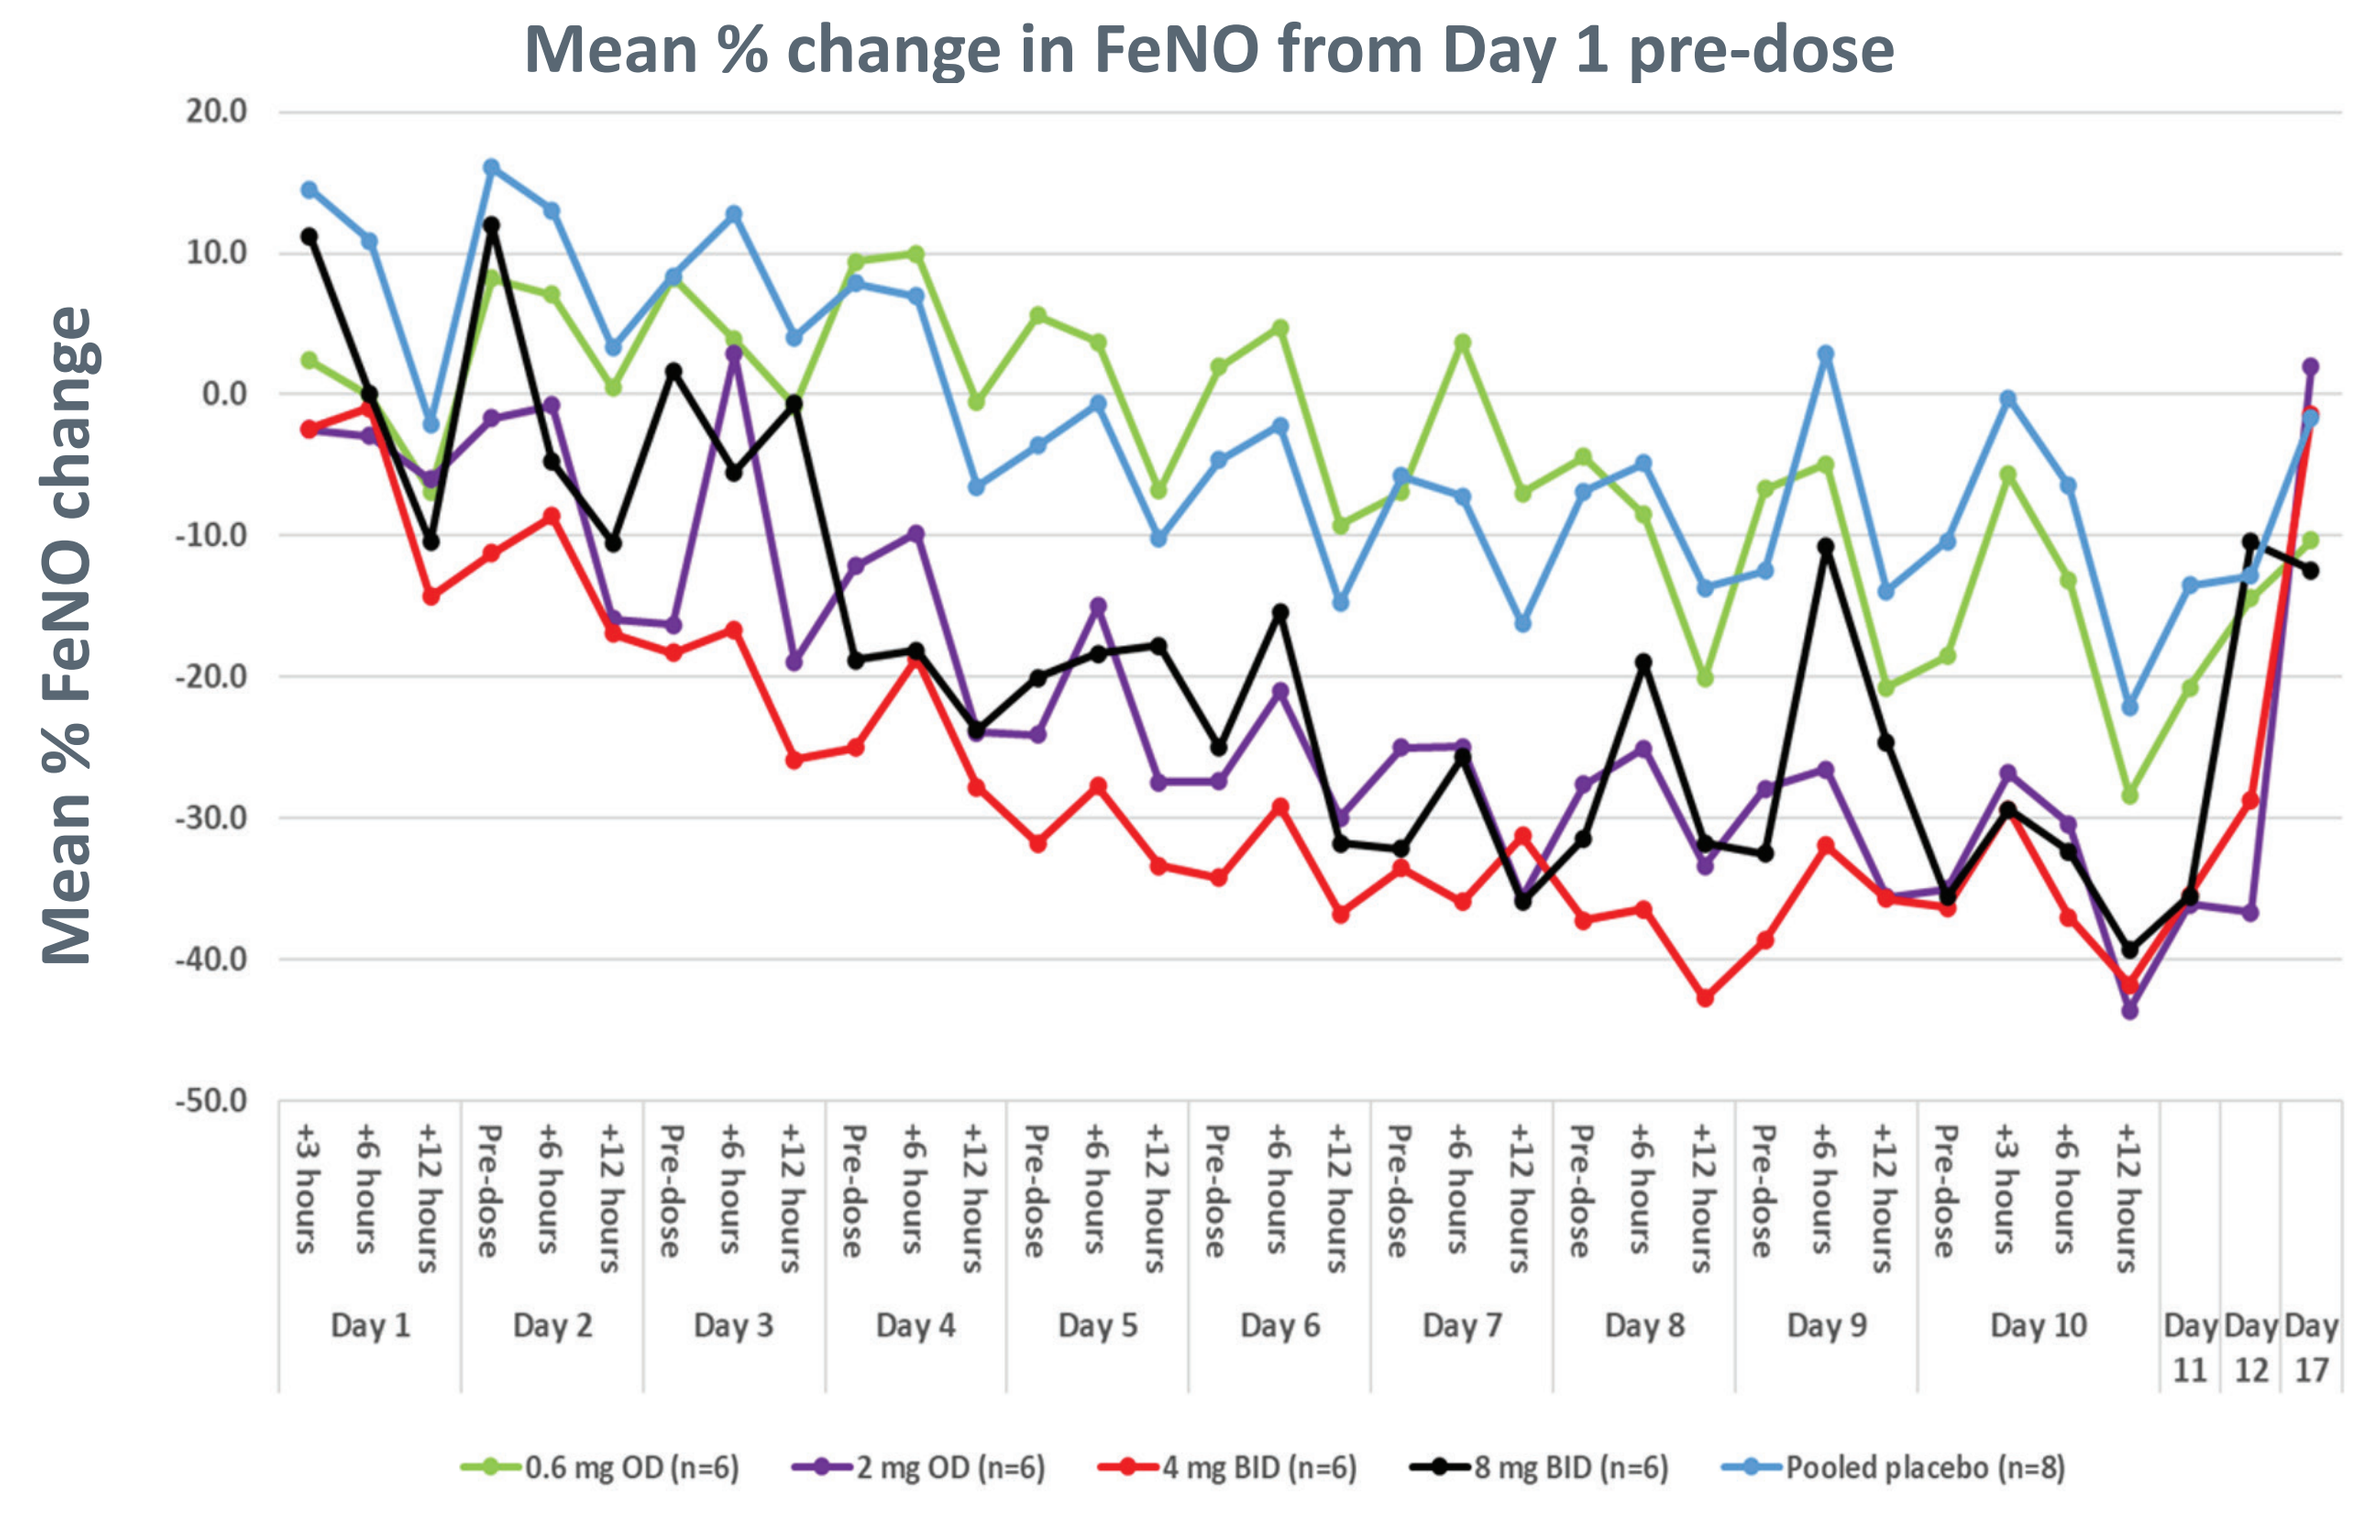

Mean % change in FeNO from Day 1 pre-dose
Mean % FeNO change
20.0
10.0
0.0
-10.0
-20.0
-30.0
-40.0
-50.0
Day 1
Day 2
Day 3
Day 4
Day 5
Day 6
Day 7
Day 8
Day 9
Day 10
Day 11
Day 12
Day 17
0.6 mg OD (n=6)
2 mg OD (n=6)
4 mg BID (n=6)
8 mg BID (n=6)
Pooled placebo (n=8)

Table 3: Table 3: KN-002 responder analysis at Day 10

Dose/regimen	% subjects with FeNO <25 ppb on Day 10
Pooled placebo (n=8)	13
0.6 mg QD (n=6)	17
2.0 mg QD (n=6)	17
4.0 mg BID (n=6)	67
8.0 mg BID (n=6)	50

Conclusions

- **No safety concerns post 10-day administration of daily doses ≤16 mg**
- **Unbound plasma levels below pharmacologically active concentrations**
- **Clinically relevant FeNO reductions achieved with ONCE- AND TWICE DAILY REGIMENS**
- **Maximal FeNO response with 4 mg BID regimen delivered via a SINGLE capsule**
- **Outcomes support evaluation of 4 mg BID in subjects with moderate/severe asthma and COPD**

References: [1] Wiegman C. ERJ 2015; 46: PA2130; [2] Wiegman C. AJRCCM 2015; 191:A6435; [3] Wiegman C. J Aerosol Med Pulm Drug Del 2015: 28(3):A23-A23.

Disclosures: This study is funded by Kinaset Therapeutics Inc. Dave Singh has received consultancy fees from Aerogen, AstraZeneca, Boehringer Ingelheim, Chiesi, Cipla, CSL Behring, EpiEndo, Genentech, GlaxoSmithKline, Glenmark, Gossamer Bio, Kinaset Therapeutics, Menarini, Novartis, Orion, Pulmatritx, Sanofi, Teva, Theravance Biopharma, and Verona Pharma. Gary Burgess has received consultancy fees from Kinaset. Julie Young, Chris O'Brien, and Frazer Morgan are employed by Kinaset.