IO-202, A First-in-Class LILRB4 Antagonist Antibody, Activates Dendritic Cells and Inhibits Solid Tumor Growth in Preclinical Studies

AACR 2021 Poster # 1629

Maria José Costa[1], Tao Huang[1], Jing-Tyan Ma[1], Caroline Bonnans[1], Azita Tabrizi[1], X. Charlene Liao[1], Paul Woodard[1], Benjamin P. Chen[2], Ching-Cheng Hsu[2], Mi Deng[2], Cheng Cheng Zhang[2], and An Song[1]

[1]Immune-Onc Therapeutics, Inc., Palo Alto, CA, USA,[2]The University of Texas Southwestern Medical Center, Dallas, TX, USA

1 Abstract

LILRB4, also known as ILT3, is an immune inhibitory transmembrane protein found on monocytes and antigen presenting cells, including dendritic cells (DCs). LILRB4 upregulation in DCs induces a tolerogenic phenotype that facilitates the generation of antigen-specific T regulatory cells [1,2]. LILRB4 is also expressed on acute myeloid leukemia (AML) with monocytic differentiation, in which it promotes T cell suppression and tumor infiltration [3,4]. IO-202 is a first-in-class LILRB4 antagonist antibody that is being evaluated in a Phase I trial (NCT04372433) for the treatment of AML and chronic myelomonocytic leukemia (CMML). RNA-seq data from TCGA database indicates that *LILRB4* expression is upregulated in many solid tumor types. Therefore, the therapeutic potential of IO-202 in solid tumors was investigated in this study.

Using computational biology approaches, we found that high *LILRB4* expression in solid tumors from TCGA database is associated with macrophage infiltration in the tumor microenvironment. Flow cytometric analysis of cancer patients' blood and tumor samples confirmed LILRB4 expression on tumor-associated macrophages (TAMs) and on monocytic myeloid-derived suppressor cells (M-MDSCs) and DCs in tumor and periphery. These data raised the hypothesis that LILRB4 functions as a myeloid checkpoint and contributes to tumor immune evasion, not only in AML, but also in many solid tumor types.

***LILRB4* is not present in rodents. As such, the functional activity and anti-tumor efficacy of IO-202 were evaluated using human primary immune cells and immunocompetent *LILRB4* transgenic mice, respectively. In healthy donor monocyte-derived DCs, IO-202 treatment promoted DC maturation, activation and an antigen presenting phenotype, as well as enhanced their ability to activate allogeneic T cells. In the radiation therapy (RT)-resistant Lewis lung carcinoma (LLC) syngeneic model, IO-202 in combination with RT resulted in tumor growth inhibition, which was associated with changes in the relative abundance of tumor-infiltrating immune cells, evidenced by a reduction in M-MDSCs and an increase in lymphocytes.**

In conclusion, our data suggest that LILRB4 functions as a myeloid checkpoint in multiple cancer types and that IO-202, the first-in-class LILRB4 antagonist antibody, enhances dendritic cell function and T cell activation *in vitro* and anti-tumor immunity in a solid tumor model *in vivo*. These data demonstrate the therapeutic potential of IO-202 as a myeloid checkpoint inhibitor for solid tumors.

2 Therapeutic hypothesis for IO-202 in solid tumor indications

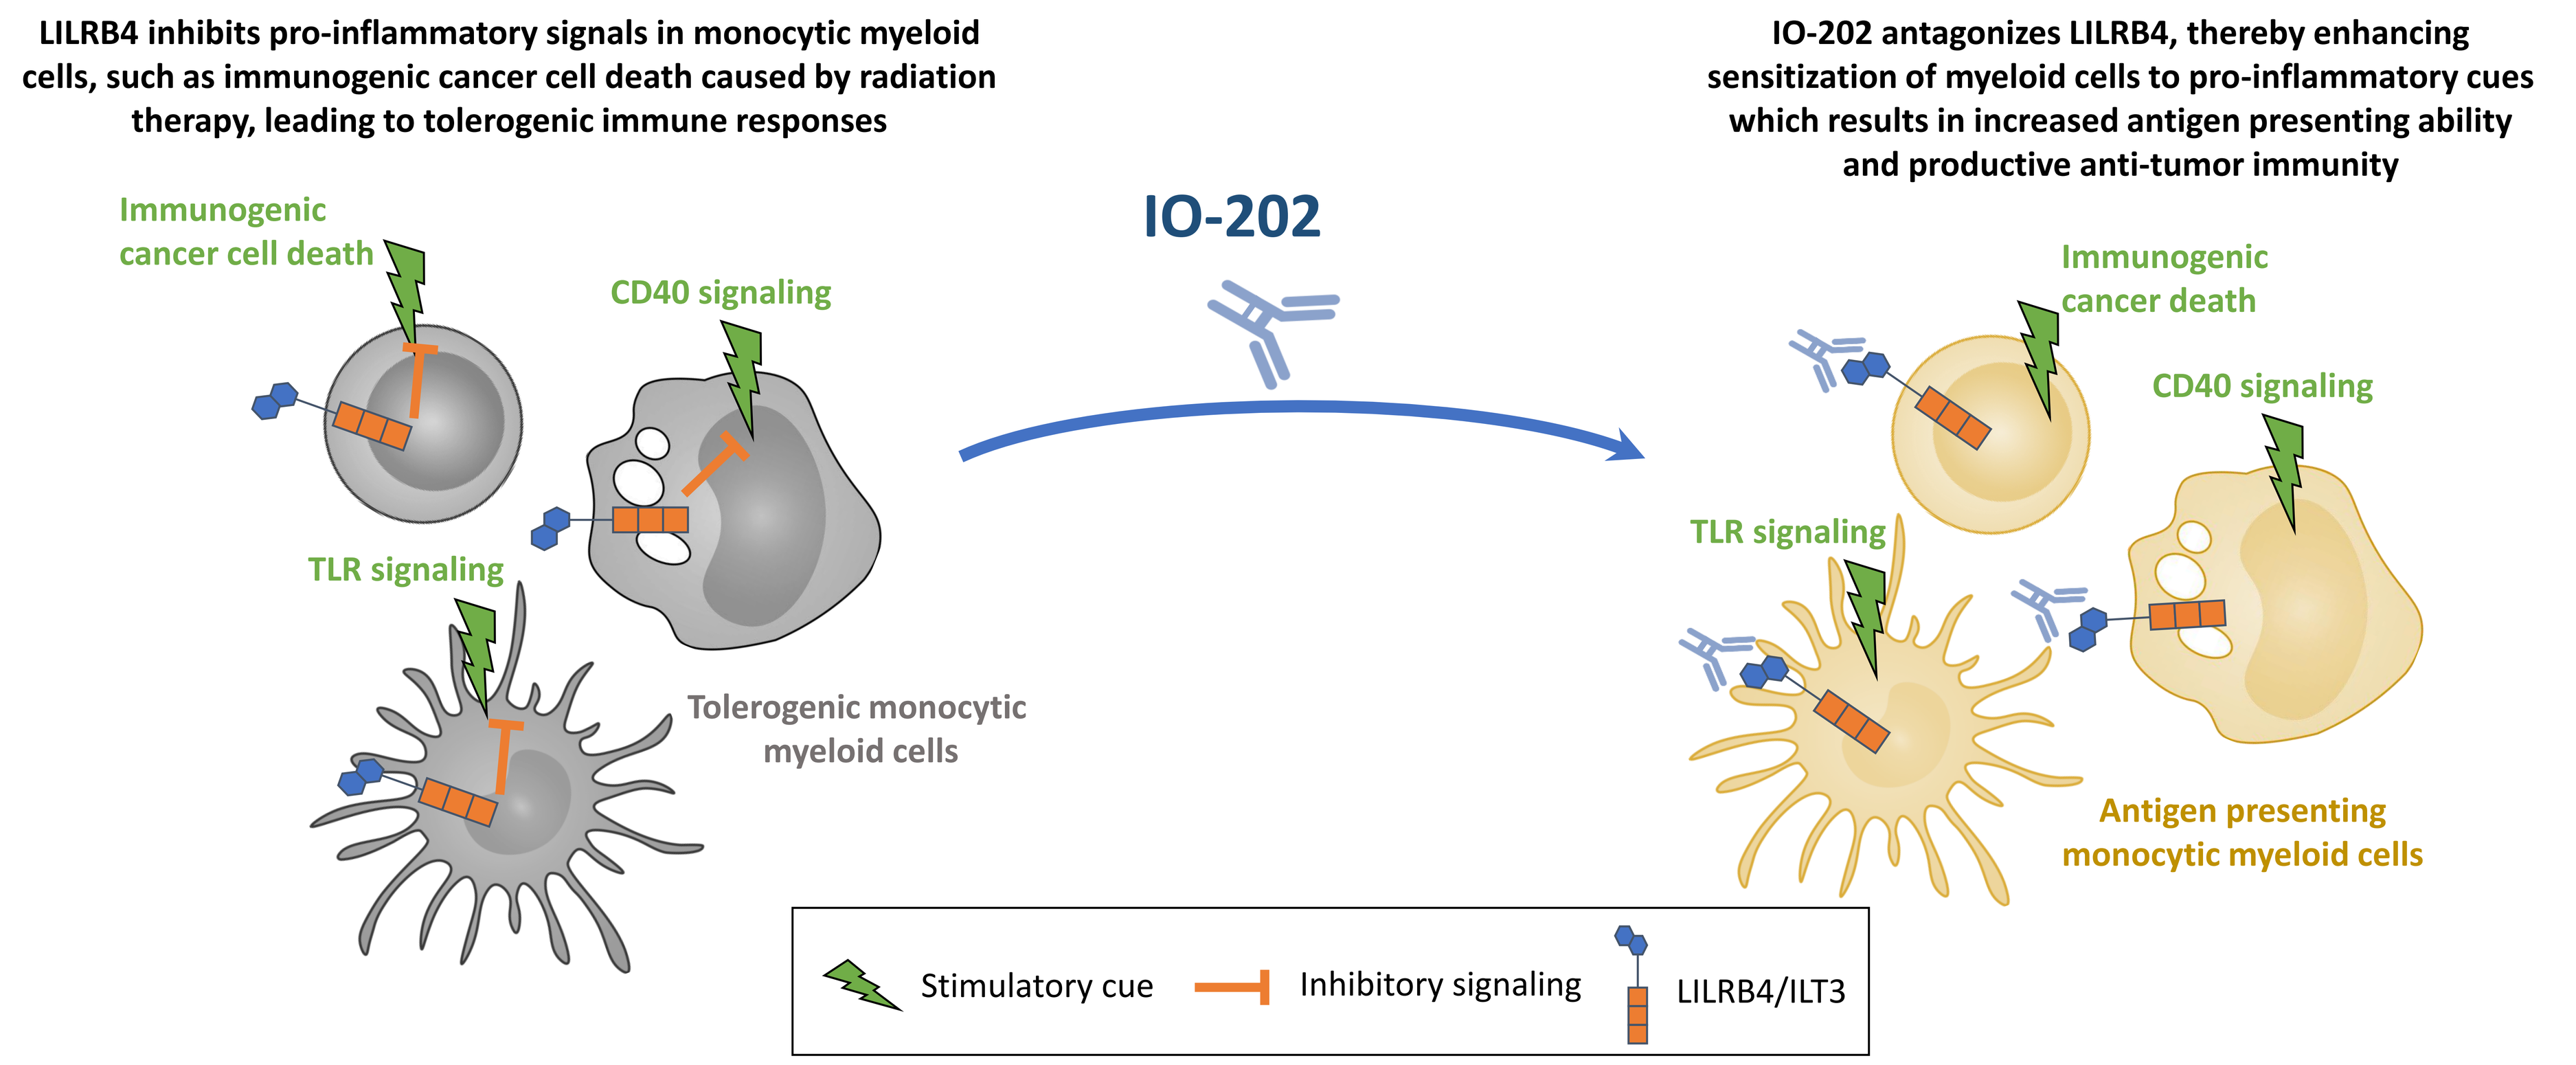
LILRB4 inhibits pro-inflammatory signals in monocytic myeloid cells, such as immunogenic cancer cell death caused by radiation therapy, leading to tolerogenic immune responses
IO-202 antagonizes LILRB4, thereby enhancing sensitization of myeloid cells to pro-inflammatory cues which results in increased antigen presenting ability and productive anti-tumor immunity
Immunogenic cancer cell death
CD40 signaling
TLR signaling
Tolerogenic monocytic myeloid cells
IO-202
Immunogenic cancer death
CD40 signaling
TLR signaling
Antigen presenting monocytic myeloid cells
Stimulatory cue
Inhibitory signaling
LILRB4/ILT3

3 Expression of *LILRB4* in solid tumors is associated with macrophage infiltration

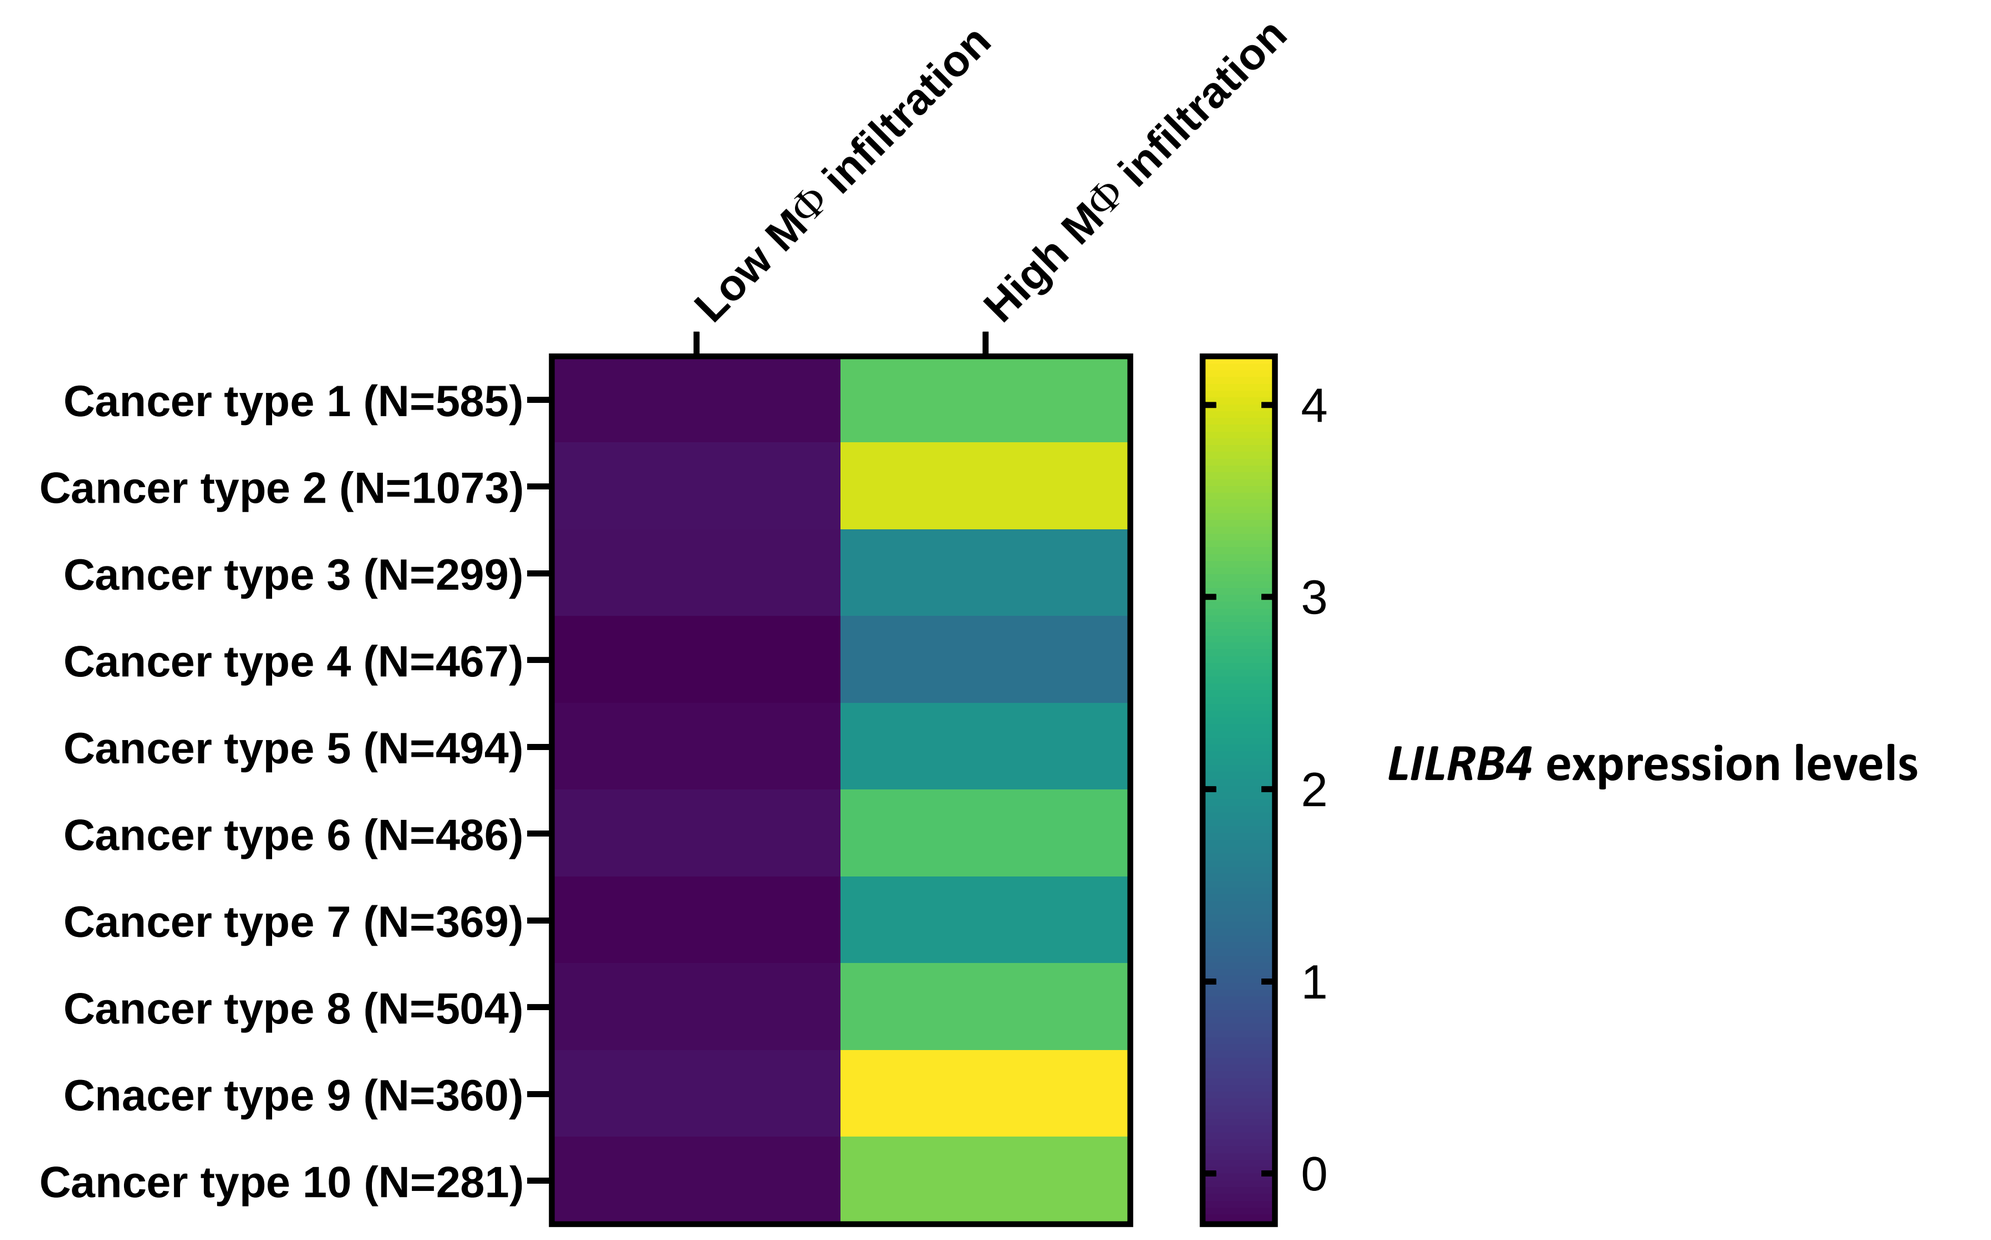
Low MΦ infiltration
High MΦ infiltration
Cancer type 1 (N=585)
Cancer type 2 (N=1073)
Cancer type 3 (N=299)
Cancer type 4 (N=467)
Cancer type 5 (N=494)
Cancer type 6 (N=486)
Cancer type 7 (N=369)
Cancer type 8 (N=504)
Cnacer type 9 (N=360)
Cancer type 10 (N=281)
4
3
2
1
0
LILRB4 expression levels

Macrophages are the most abundant immune cell type in many solid tumors [5]. By segregating solid tumor samples from the TCGA RNA seq database based on the presence or not of a distinct macrophage gene expression "signature", we demonstrate the association of increased expression of *LILRB4* with high macrophage infiltration. The results shown here are in part based upon data generated by the TCGA Research Network: https://www.cancer.gov/tcga.

4 IO-202 binds monocytic myeloid cells in peripheral blood and solid tumor tissue

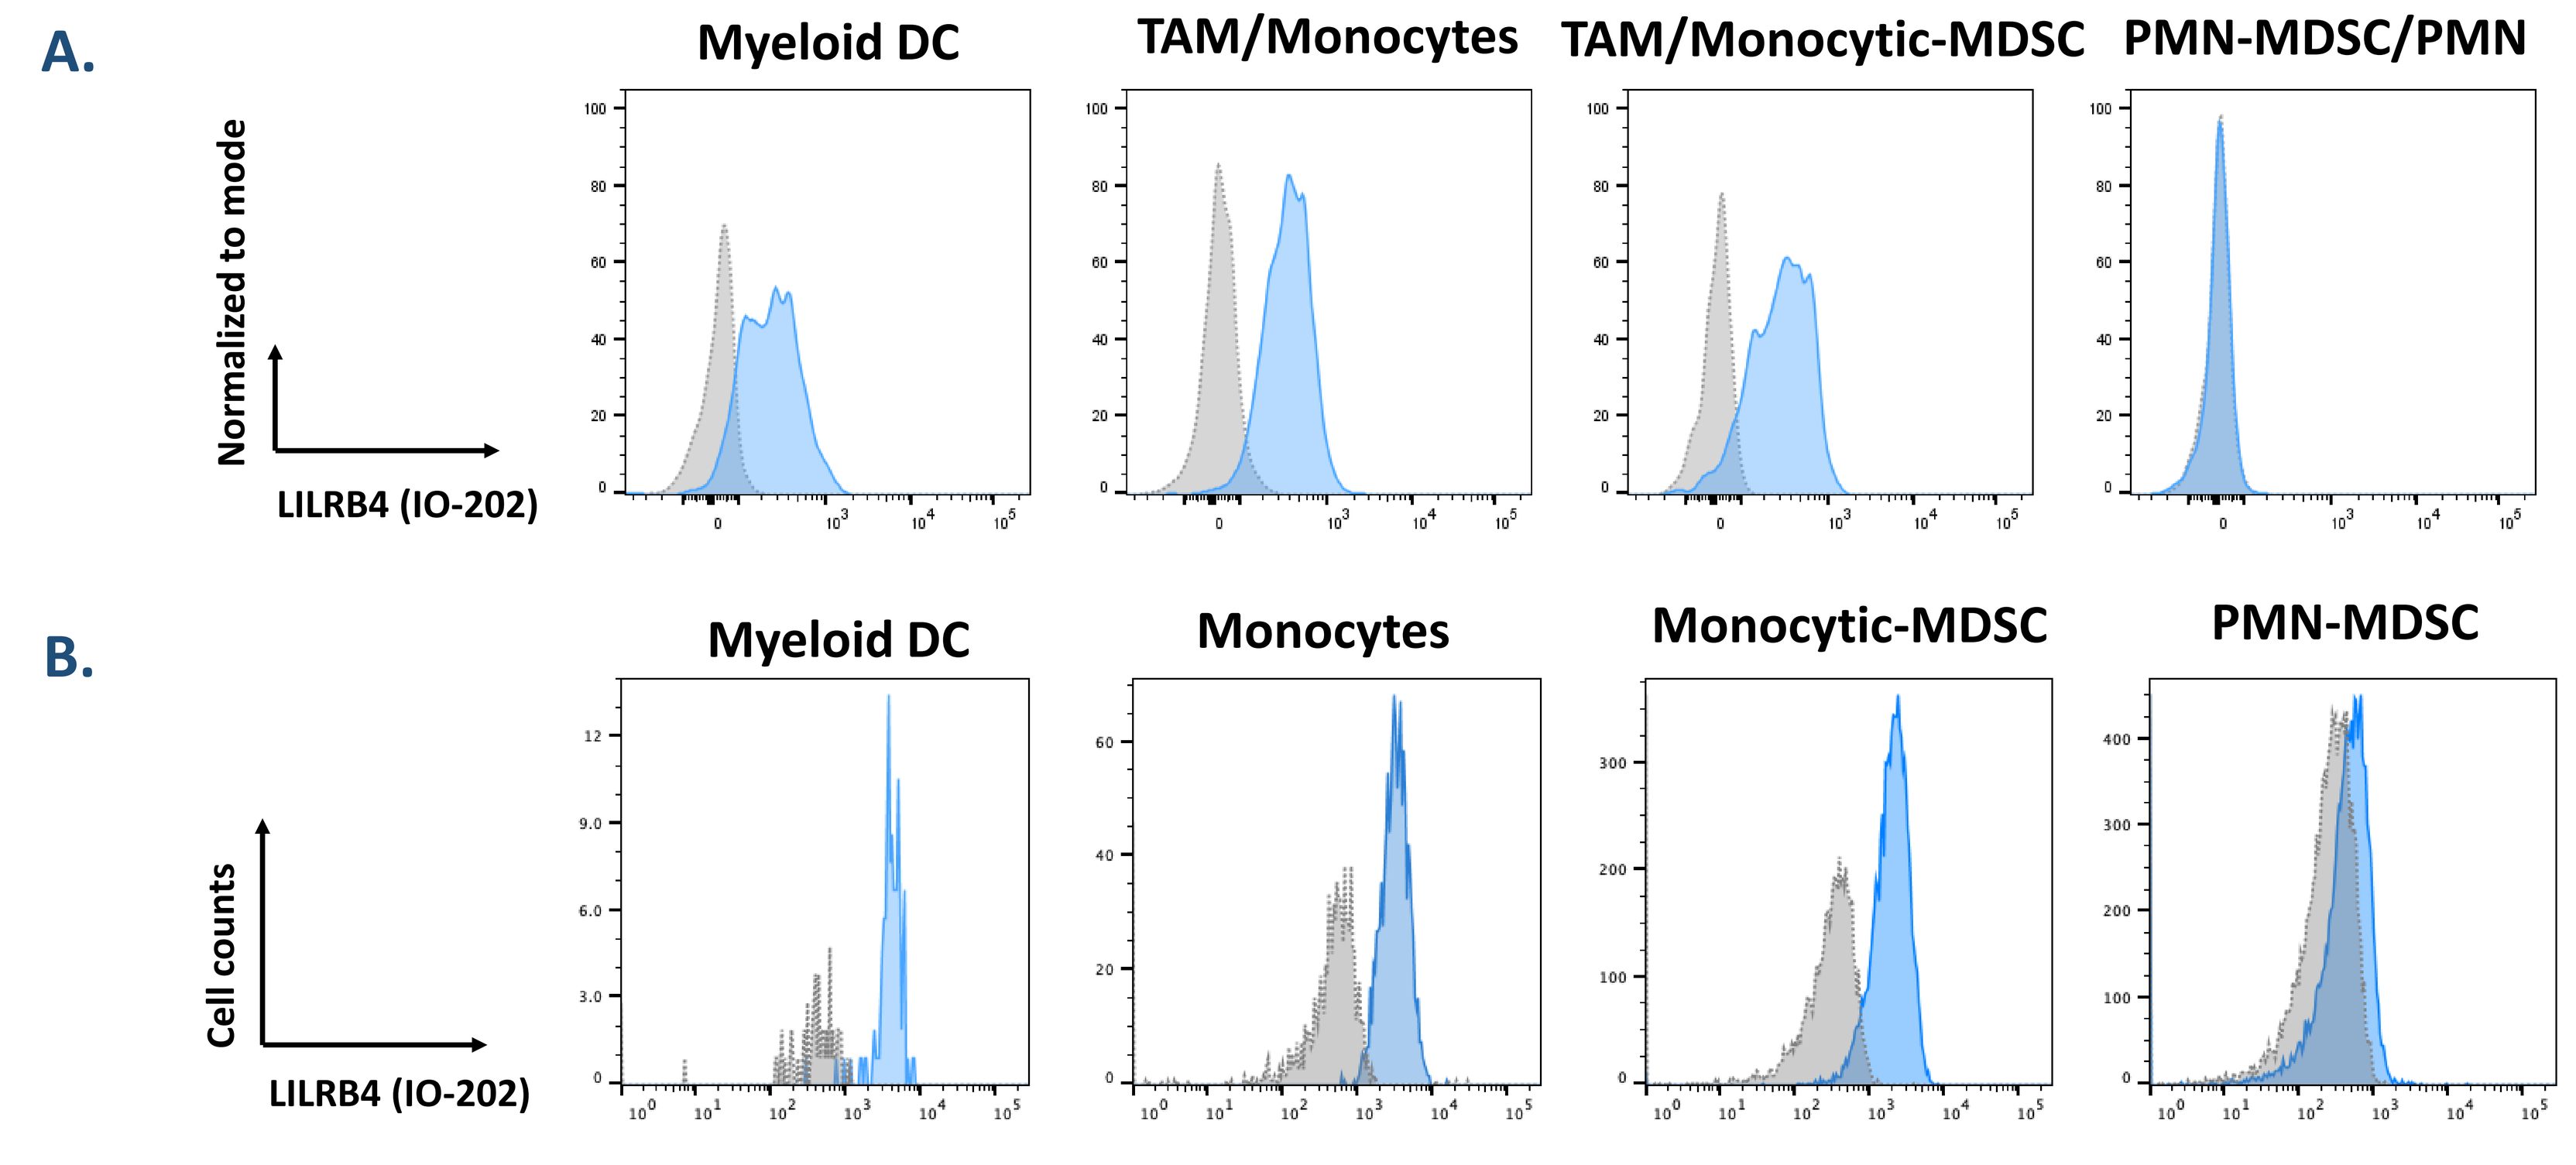
A.
Myeloid DC
TAM/Monocytes
TAM/Monocytic-MDSC
PMN-MDSC/PMN
Normalized to mode
LILRB4 (IO-202)
B.
Myeloid DC
Monocytes
Monocytic-MDSC
PMN-MDSC
Cell counts
LILRB4 (IO-202)

Representative results from flow cytometric analysis showing that IO-202 (blue histogram) specifically binds to all monocytic myeloid cells in tumor tissue and peripheral blood from solid tumor cancer patients. Grey histogram corresponds to sample incubated with IgG1 isotype control. Samples were stained with antibody cocktails for cell surface markers and IO-202 or isotype control antibody. **A.** Tumor tissue was dissociated into single cell suspensions using non-enzymatic methods. Myeloid cell populations were gated as follows: myeloid DC ($CD11b^+CD15^-CD14^-HLA\text{-}DR^+CD11c^+$), TAM/monocytes ($CD11b^+CD15^-CD14^+HLA\text{-}DR^+$), TAM/M-MDSC ($CD11b^+CD15^-CD14^+HLA\text{-}DR^-$), PMN-MDSC/PMN ($CD11b^+CD15^+CD14^-$). **B.** Peripheral blood from the same patient as in A. Myeloid cell populations were gated as follows: myeloid DC ($CD11b^+CD14^-CD11c^+CD1c^+$), monocytes ($CD11b^+CD14^+HLA\text{-}Dr^{hi}$), Monocytic-MDSC ($CD11b^+CD14^+HLA\text{-}DR^{Lo}$), PMN-MDSC ($CD11b^+CD15^+CD14^-HLA\text{-}DR^-Lox\text{-}1^+$).

5 IO-202 enhances dendritic cell activation in response to TLR signaling

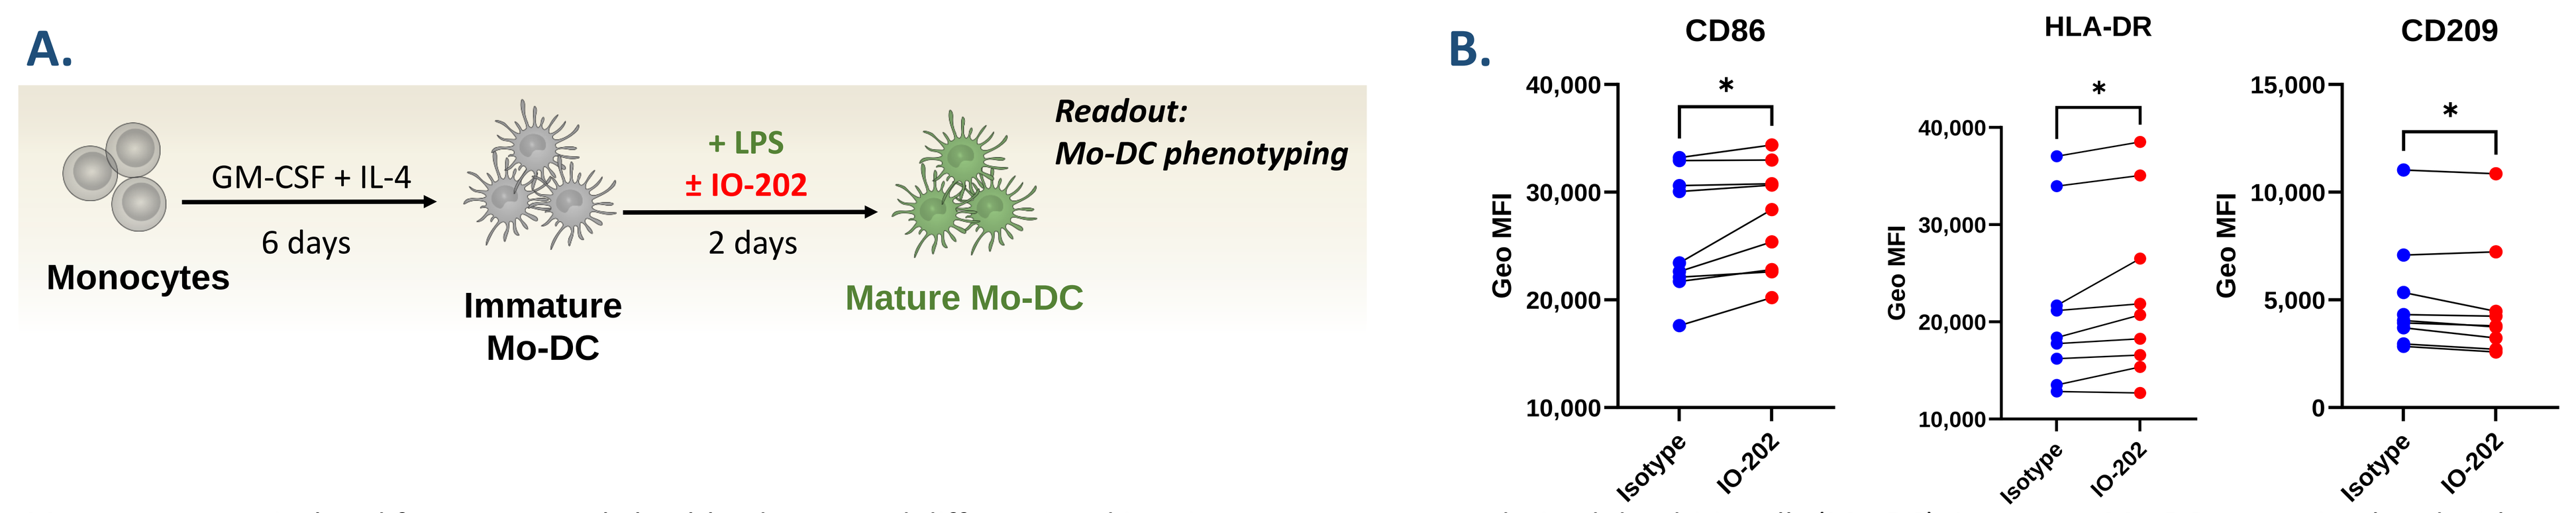
A.
Monocytes
GM-CSF + IL-4
6 days
Immature Mo-DC
+ LPS
± IO-202
2 days
Mature Mo-DC
Readout:
Mo-DC phenotyping
B.
CD86
HLA-DR
CD209
Geo MFI
Isotype
IO-202

A. Monocytes were isolated from apparently healthy donors and differentiated into immature monocyte-derived dendritic cells (Mo-DC). Immature Mo-DC were stimulated with 100 ng/mL LPS for 2 days in the presence of 15 µg/mL IO-202 or isotype control, to assess the effect of IO-202 on Mo-DC in response to activation of TLR4 signaling. **B.** DC were harvested and the expression of key dendritic cell activation (CD86 and HLA-DR) and tolerogenic (CD209) markers was analyzed by flow cytometry. Each line represents the result from a different donor, *$p < 0.05$ (paired t test).

6 IO-202 potentiates CD40L-induced dendritic cell maturation/activation, leading to increased T cell activation

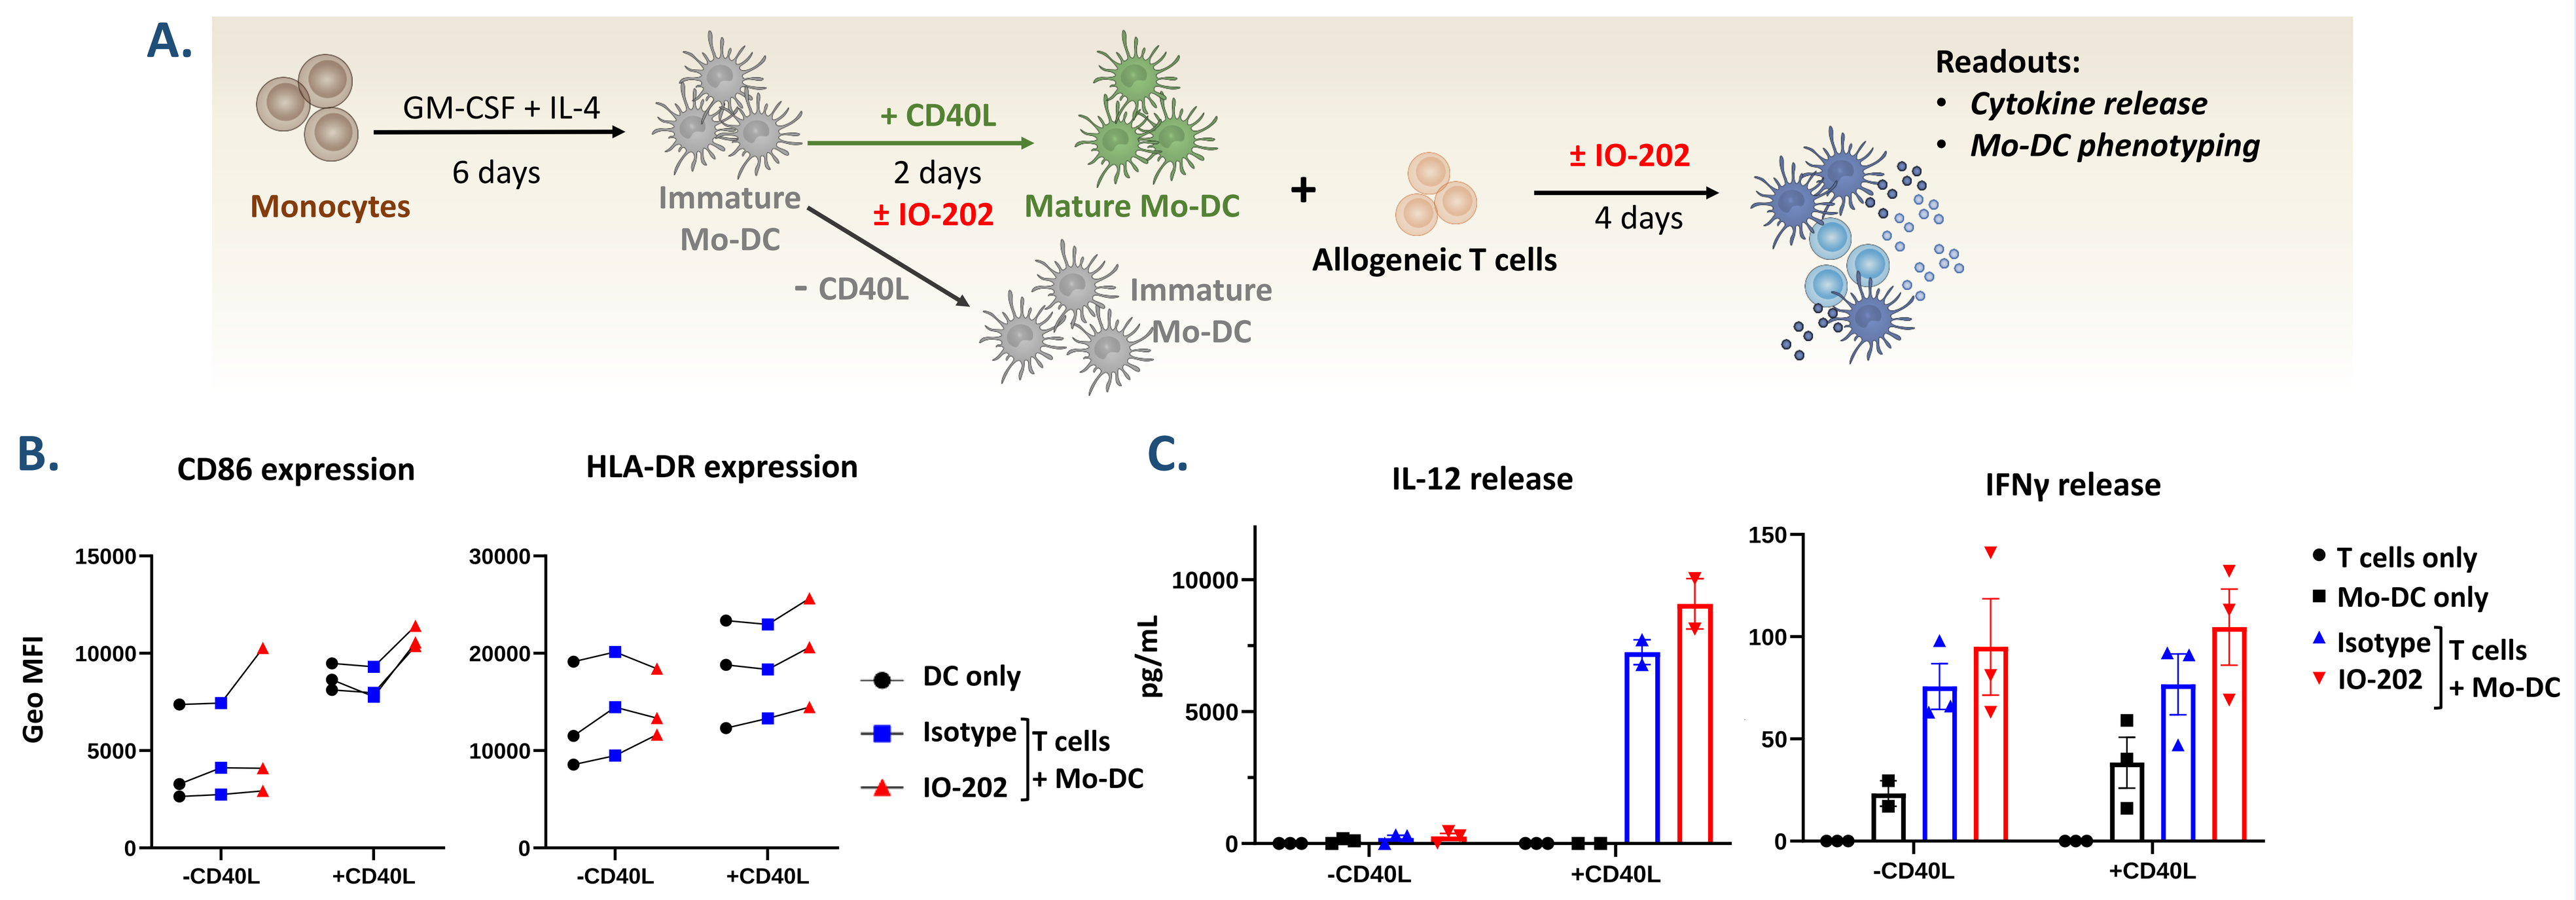
A.
Monocytes
GM-CSF + IL-4
6 days
Immature Mo-DC
+ CD40L
2 days
± IO-202
Mature Mo-DC
- CD40L
Immature Mo-DC
+
Allogeneic T cells
± IO-202
4 days
Readouts:
Cytokine release
Mo-DC phenotyping
B.
CD86 expression
HLA-DR expression
Geo MFI
-CD40L
+CD40L
DC only
Isotype
IO-202
T cells + Mo-DC
C.
IL-12 release
IFNγ release
pg/mL
T cells only
Mo-DC only
Isotype
IO-202
T cells + Mo-DC

A. Immature monocyte-derived dendritic cells (Mo-DC) were generated in vitro and stimulated with 5 µg/mL CD40 ligand (CD40L) to induce Mo-DC maturation/activation through CD40 signaling or left untreated (immature Mo-DC). During both culture conditions, either IO-202 or isotype control (15 µg/mL) was added to the media. At the end of the treatment, Mo-DC were co-cultured with allogeneic T cells to assess the effect of IO-202 on the phenotype and function of mature and immature Mo-DC. **B-C.** Each line or data point represents the result from a different donor at the end of the co-culture. IO-202 enhanced the antigen presentation phenotype and functionality of CD40L-matured/activated Mo-DC but not of immature Mo-DC. **B.** The expression of key dendritic cell activation (CD86 and HLA-DR) markers was analyzed by flow cytometry. **C.** Cytokine levels at the end of the Mo-DC+T cell co-culture, as determined by ELISA of media supernatant samples. Data shown are mean ± standard error of the mean. IO-202 increased the levels of IL-12 (a cytokine that promotes Th1 responses) and IFNγ (an effector cytokine indicative of T cell activation) in co-cultures specifically bearing Mo-DC activated/matured by CD40L.

7 LILRB4 antagonism sensitizes solid tumor syngeneic model to radiation therapy

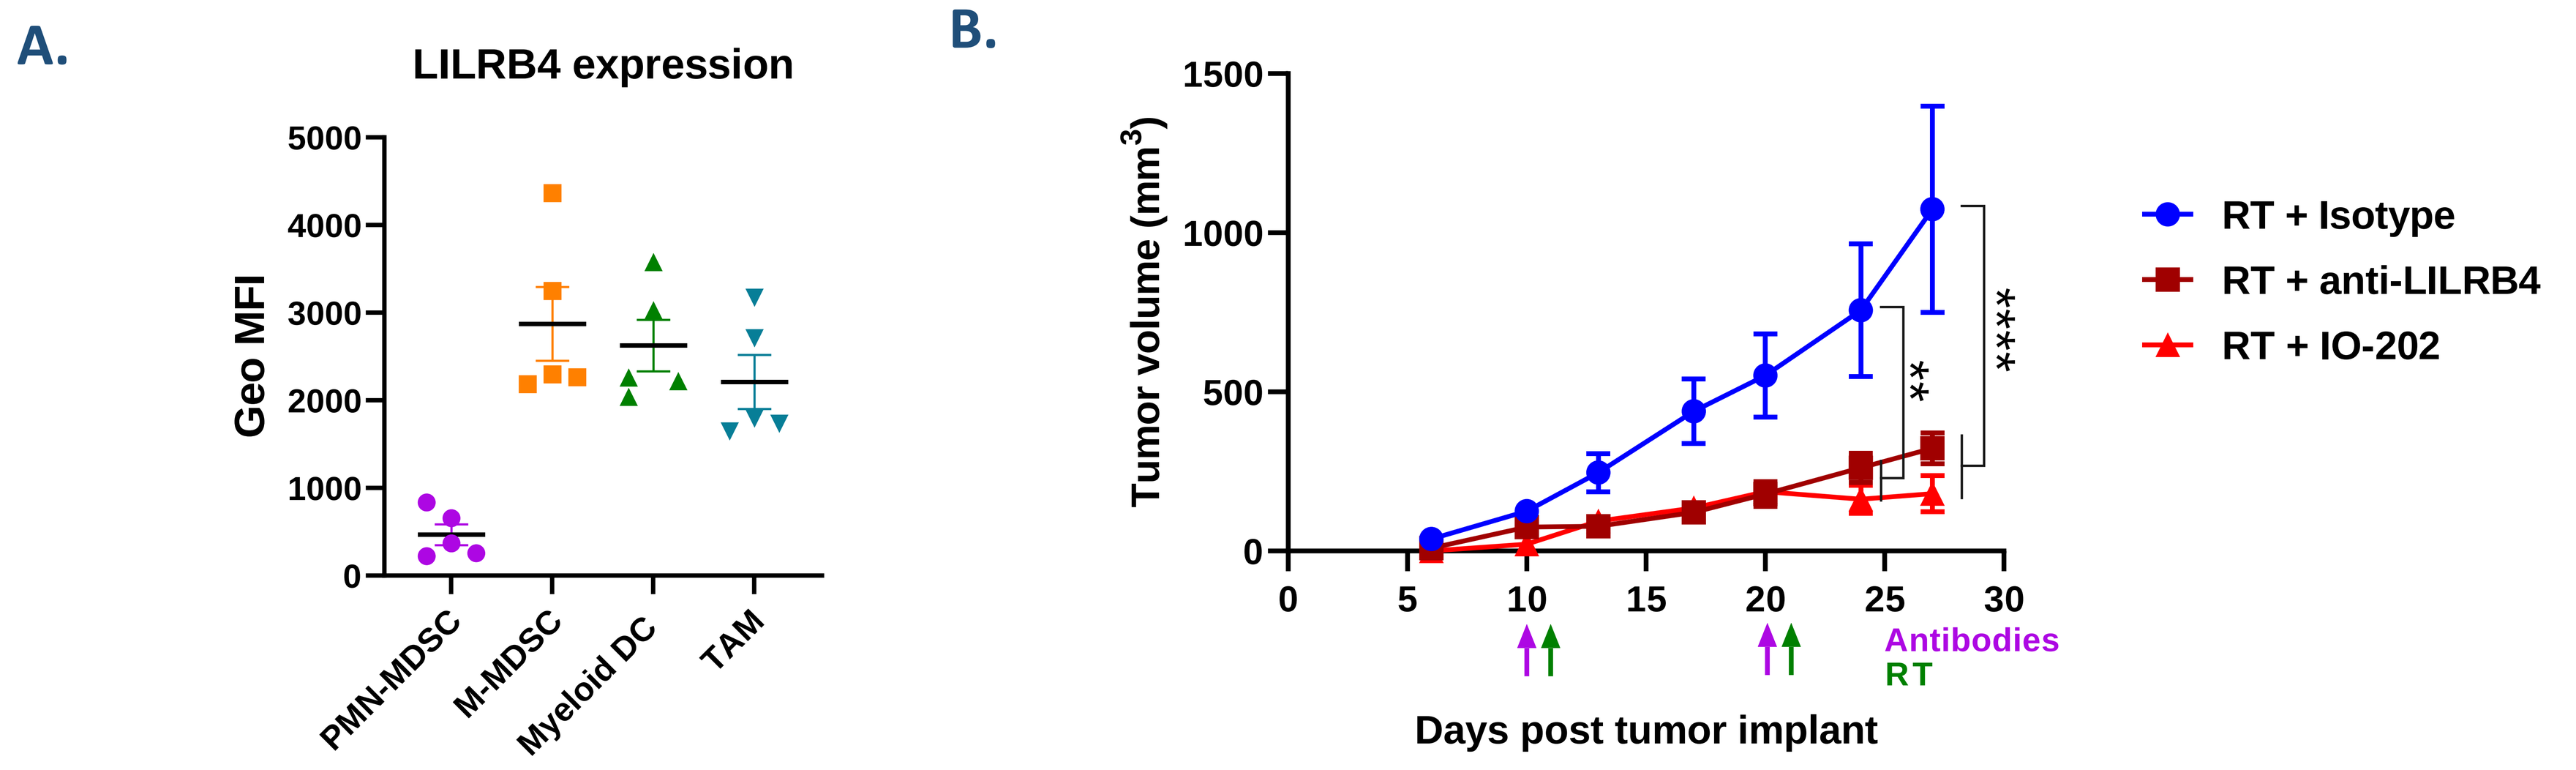
A.
LILRB4 expression
Geo MFI
PMN-MDSC
M-MDSC
Myeloid DC
TAM
B.
Tumor volume (mm³)
Days post tumor implant
Antibodies
RT
**

RT + Isotype
RT + anti-LILRB4
RT + IO-202

A. Expression of LILRB4 in various myeloid cells from the immune infiltrate of Lewis lung carcinoma tumors implanted in syngeneic *Cx3cr1*-Cre/*Rosa26-LILRB4* transgenic mice.
B. The radiation therapy (RT) -resistant Lewis lung carcinoma model [6] was subcutaneously implanted in LILRB4 transgenic mice. On day 10 post tumor implant, mice were randomized into 3 treatment groups and received 10 mg/kg human IgG1 (hIgG1) isotype control, or IO-102 or a research tool anti-LILRB4 hIgG1 antibody. On the following day, the mice received the first dose (10 Gy) of RT. This treatment protocol was repeated on days 20-21 post tumor implant (arrows indicate treatment days). Data shown are mean ± standard error of the mean, with n = 3-5 mice per group, **$p < 0.01$, ****$p < 0.0001$ (2-way ANOVA with Tukey's multiple comparisons test).

8 Combining radiation therapy with LILRB4 antagonist antibody treatment rebalances the tumor immune infiltrate and promotes a pro-inflammatory phenotype in TAM

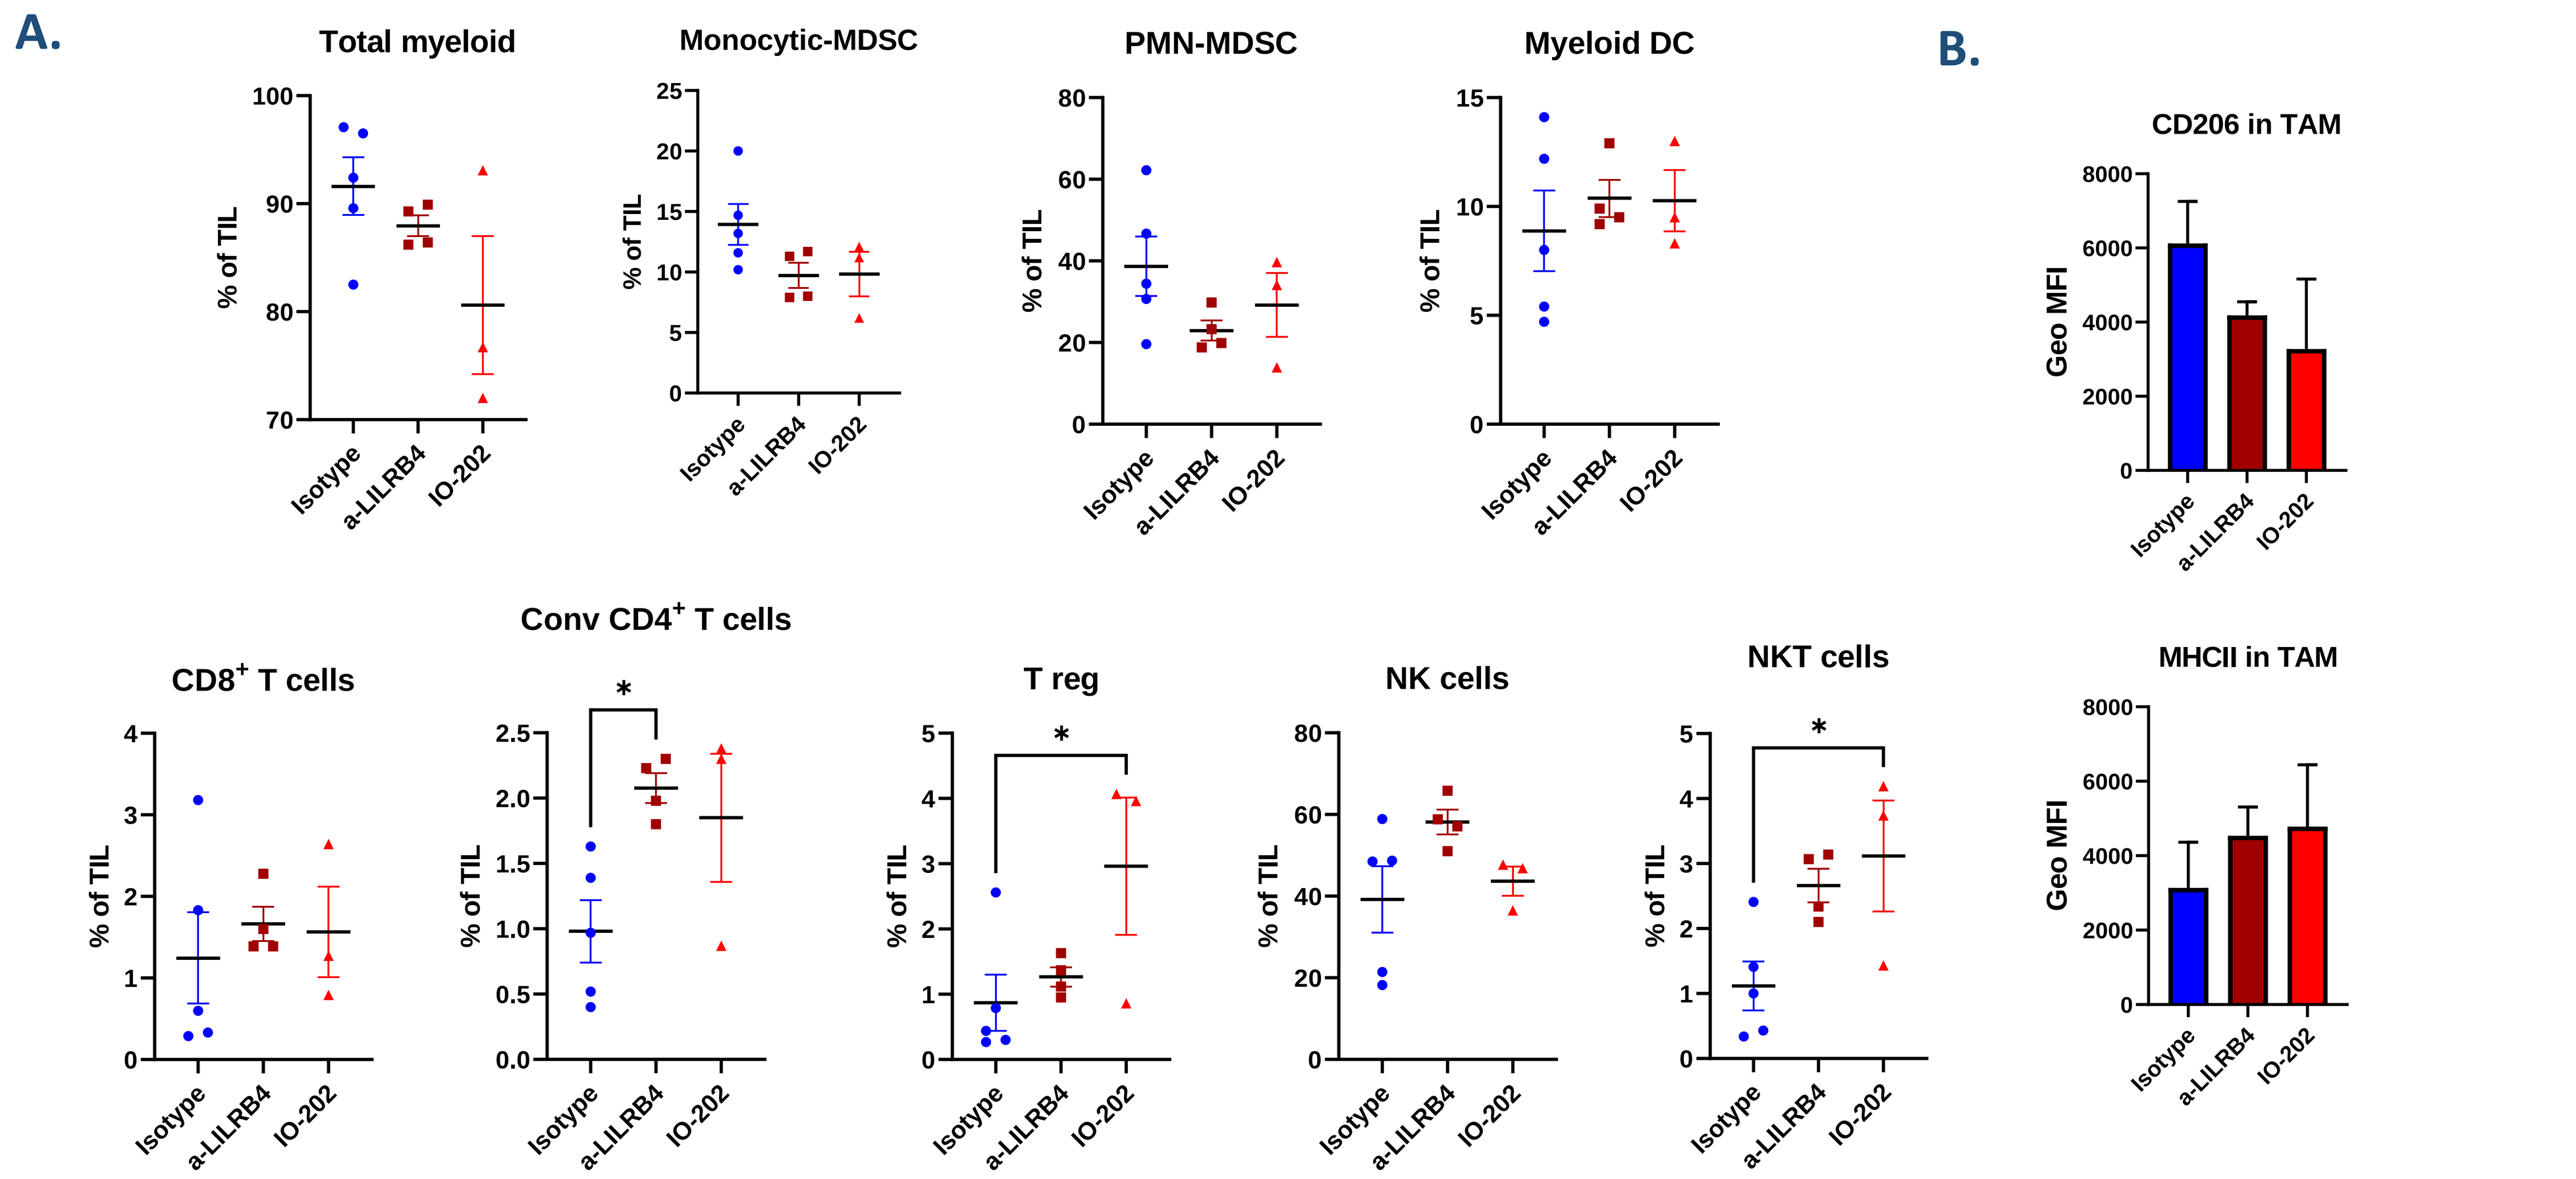
A.
Total myeloid
Monocytic-MDSC
PMN-MDSC
Myeloid DC
CD8+ T cells
Conv CD4+ T cells
T reg
NK cells
NKT cells
% of TIL
Isotype
a-LILRB4
IO-202
B.
CD206 in TAM
MHCII in TAM
Geo MFI

A-B. Effects of the combination treatment radiation therapy (RT) + anti-LILRB4 antagonism in the tumor immune infiltrate of Lewis lung carcinoma model implanted in syngeneic *Cx3cr1*-Cre/*Rosa26-LILRB4* transgenic mice. Tumors were analyzed by flow cytometry at the end of the study shown in panel 7B (day 27 post tumor implant). Compared to RT + isotype control, the combination of RT + LILRB4 antagonist antibodies induced immune changes indicative of an enhanced anti-tumor response. Data shown are mean ± standard error of the mean. **A.** The combination of RT + LILRB4 antagonist antibodies resulted in a decrease in the frequencies of myeloid-derived suppressive cells (MDSC), whereas the frequencies of lymphocytes were increased. Each data point represents an individual mouse. TIL = tumor-infiltrating leukocytes, MDSC = myeloid-derived suppressor cells, PMN = polymorphonuclear, conv = conventional, T reg = T regulatory cells. *$p < 0.05$ (One-way ANOVA). **B.** The combination of RT + LILRB4 antagonist antibody treatments resulted in a decrease in the expression of the anti-inflammatory marker CD206, whereas it increased the expression of MHCII on TAM. These changes suggest that ~~the~~ combination treatment induced a repolarization of TAM from pro-tumorigenic/anti-inflammatory to pro-inflammatory, potentially contributing to the anti-tumor response. There were no detectable changes in the frequency of tumor-associated macrophages (TAM) (data not shown).

9 Conclusions

- ***LILRB4* mRNA expression is associated with macrophage infiltration in many solid tumor types from TCGA database**
- **IO-202 specifically binds to monocytic myeloid cells from human tumor tissue and peripheral blood, confirming LILRB4 expression in cancer-associated monocytic myeloid cells**
- **IO-202 antagonism of LILRB4 activity enhances the pro-inflammatory effect and antigen-presenting phenotype of DC induced by various activation signals (ex. TLR, CD40) and thereby promotes their ability to activate allogeneic T cells**
- **Both IO-202 and a research tool LILRB4 antagonist antibody sensitize the Lewis lung carcinoma model to radiation therapy in syngeneic *LILRB4* transgenic mouse.**
- **The anti-tumor efficacy of the IO-202 + radiation therapy combination correlates with a rebalance of the tumor immune infiltrate, with shifts in lymphocyte and MDSC frequencies and the emergence of a pro-inflammatory TAM phenotype**
- **Data presented herein provide rationale for evaluating the clinical activity of IO-202 as an immunotherapy in solid tumors**

10 References

[1] Manavalan JS, Rossi PC, Vlad G *et al.* High expression of ILT3 and ILT4 is a general feature of tolerogenic dendritic cells. *Transpl Immunol* 2003;11:245–58.[2] Suciu-Foca N, Feirt N, Zhang Q-Y *et al.* Soluble Ig-like transcript 3 inhibits tumor allograft rejection in humanized SCID mice and T cell responses in cancer patients. *J Immunol Baltim Md 1950* 2007;178:7432–41.[3] Deng M, Gui X, Kim J *et al.* LILRB4 signalling in leukaemia cells mediates T cell suppression and tumour infiltration. *Nature* 2018;562:605–9. [4] Li Z, Deng M, Huang F *et al.* LILRB4 ITIMs mediate the T cell suppression and infiltration of acute myeloid leukemia cells. *Cell Mol Immunol* 2020;17:272–82. [5] Cassetta L & Pollard JW, *Nature Reviews Drug Discovery*, 2018 Dec;17(12):887-904. [6] Yokouchi H *et al*. Combination tumor immunotherapy with radiotherapy and Th1 cell therapy against Lewis lung carcinomas. *Proc Amer Assoc Cancer Res*, Volume 45, 2004.