Preclinical pharmacology of low molecular weight GLP-1 receptor agonist XW014

W. GUO[1], X. WU[1], H. ZOU[1], B. WANG[1], R. ZHU[1], C. L. JONES[2], M. FENAUX[2]
[1]Sciwind Biosciences, China [2]Sciwind Biosciences, USA

INTRODUCTION

Glucagon-like peptide-1 (GLP-1) is a peptide hormone produced by the gut in response to food intake and then rapidly degraded. GLP-1 has multiple beneficial effects on metabolism, including delaying gastric emptying and increasing satiety, stimulating insulin secretion, lowering blood sugar, reducing liver fat, and promoting weight loss. Peptide agonists of the GLP-1 receptor (GLP-1R) have shown promise for treating obesity, type 2 diabetes, and nonalcoholic steatohepatitis (NASH)[1]. XW014 is a small molecule, orally bioavailable GLP-1R agonist that is in preclinical development.

AIM

To evaluate the activity of small molecule GLP-1R agonist XW014 *in vitro* and *in vivo.*

METHOD

GLP-1R agonism was evaluated *in vitro* by assessing cAMP production in HEK-293 cells overexpressing GLP-1R from human or cynomolgus monkey. cAMP production in cells treated with XW014 for 30 minutes was quantified by HTRF-based assay (Cisbio).

Recombinant C57BL/6 mice (n=8/group) expressing humanized GLP-1R were given a single oral dose of vehicle (10% solutol + 90% saline) or test compounds (XW014 at 3, 10, or 30 mg/kg, or comparator GLP-1R agonist danuglipron [PF-06882961][2] at 10 mg/kg). Intraperitoneal glucose tolerance test (IPGTT) was performed 30 min after dosing.

Cynomolgus monkeys (n=5/group) were given a single oral dose of XW014 (10, 20, or 40 mg/kg) or IV dose of danuglipron (2 mg/kg). Intravenous glucose tolerance test (IVGTT) was performed at baseline and 7 min (IV group) or 2 h (PO groups) after dosing.

C57BL/6 diet-induced obesity (DIO) mice expressing humanized GLP-1R were dosed with XW014 (30 or 60 mg/kg PO BID) or vehicle for 15 days (n=4/group for vehicle; n=5/group for treatment). Weight, cumulative food intake, post-prandial glucose, and oral glucose tolerance on Day 15 were measured.

RESULTS

XW014 is a potent GLP-1R agonist *in vitro*

XW014 showed potent agonism of cAMP pathway activation in HEK-293 cells expressing GLP-1R from human and cynomolgus monkey.

	XW014 EC_{50} (mean +/- SD) [nM]
Human	0.44 +/- 0.20
Cynomolgus monkey	0.79 +/- 0.25

XW014 reduces blood glucose in a humanized GLP-1R mouse model

Humanizing GLP-1R is required to test small molecule agonists in mice, as this class of compounds does not activate the rodent receptor[3].

We tested single oral doses of 3, 10, and 30 mg/kg XW014, as well as a comparator small molecule GLP-1R agonist danuglipron (10 mg/kg), in the hGLP-1R mouse model.

Blood glucose levels over time

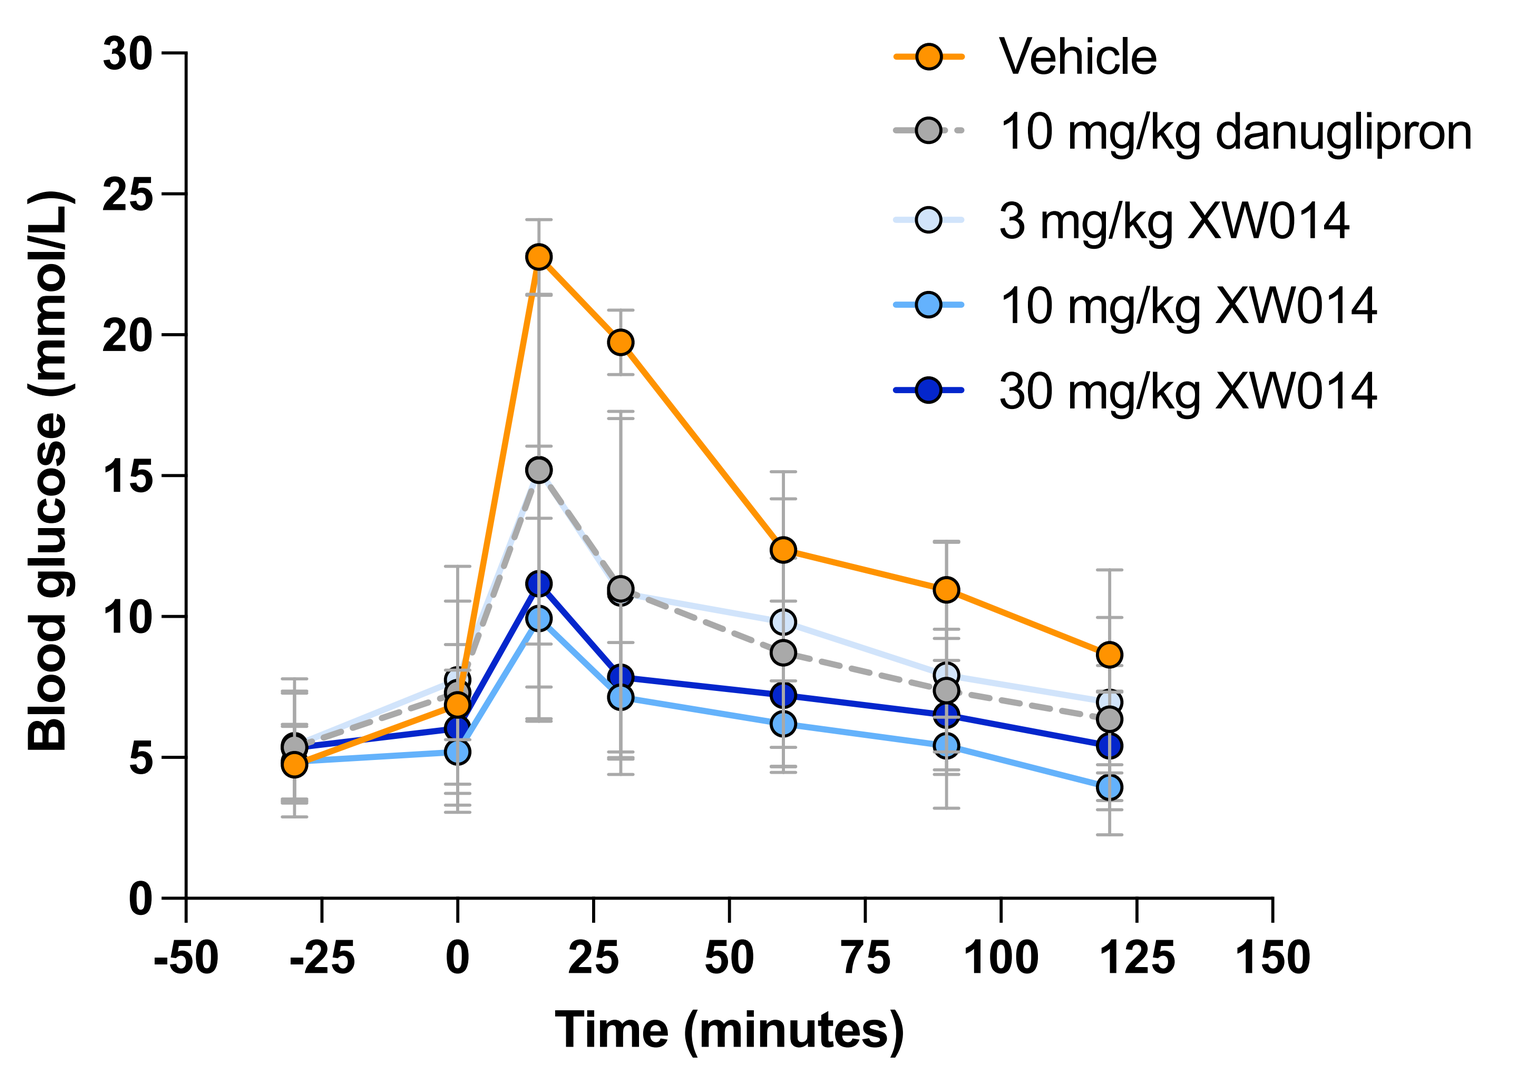
Vehicle
10 mg/kg danuglipron
3 mg/kg XW014
10 mg/kg XW014
30 mg/kg XW014
Blood glucose (mmol/L)
Time (minutes)

Blood glucose $AUC_{(0-120min)}$

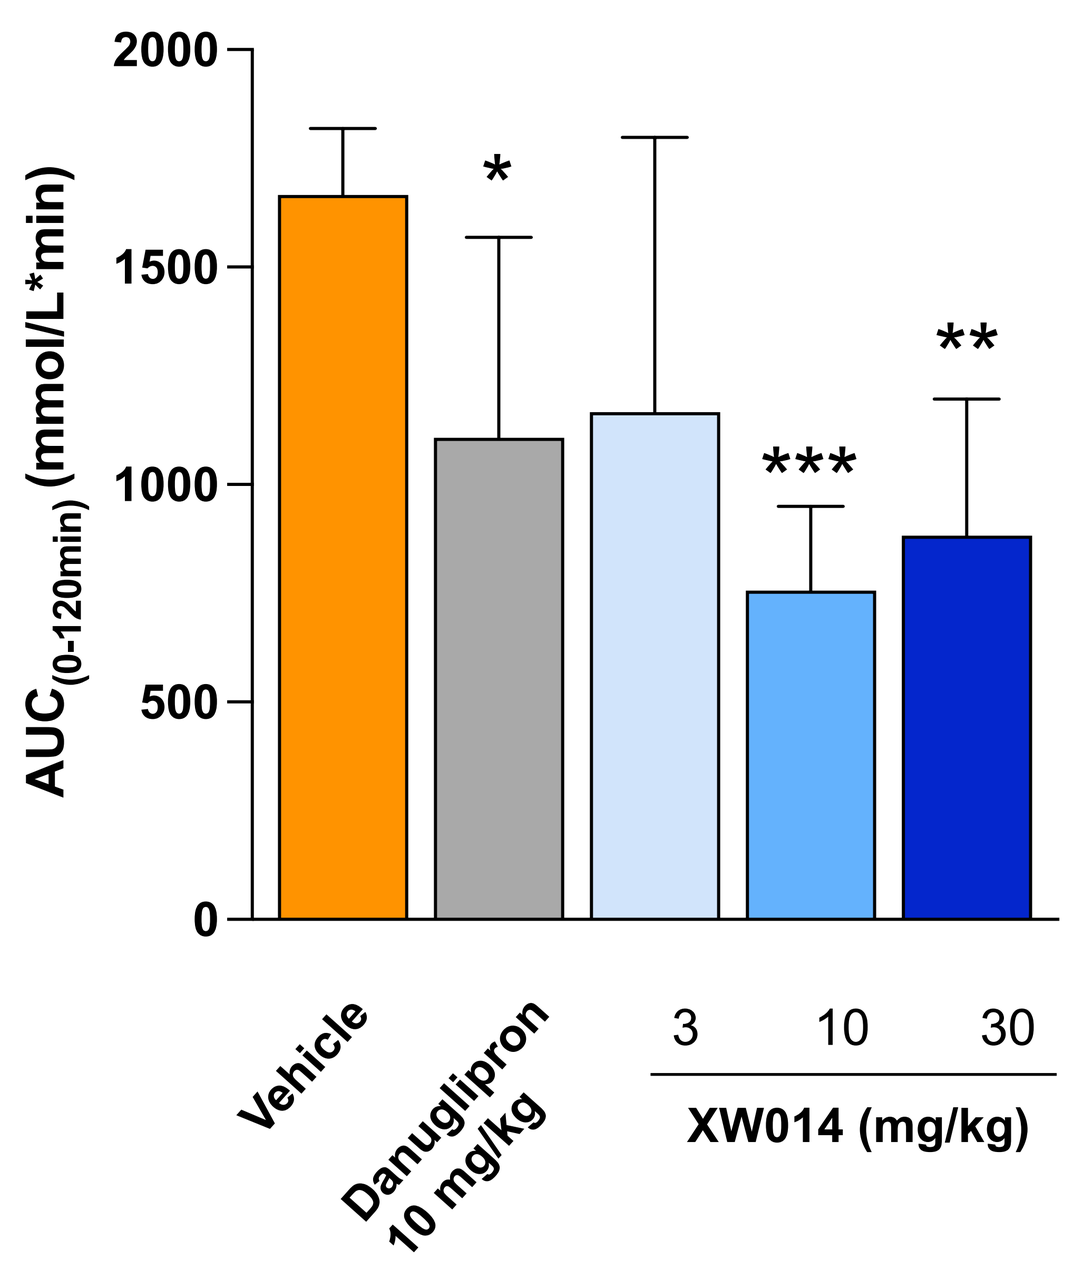
$AUC_{(0-120min)}$ (mmol/L*min)
Vehicle
Danuglipron 10 mg/kg
3
10
30
XW014 (mg/kg)
*

**

Blood glucose levels over time are presented as mean +/- standard deviation (SD). Blood glucose AUC(0-120 min) are presented mean and standard deviation (SD). One-way ANOVA analysis was used to evaluate significance: *p<0.05, **p<0.01, ***p<0.001. n=8 animals per group.

Escalating doses of XW014 at 3, 10, and 30 mg/kg, as well as danuglipron (10 mg/kg), lowered blood glucose levels following IPGTT. Both the 10 mg/kg and 30 mg/kg doses of XW014 significantly reduced blood glucose $AUC_{0-120min}$ compared to vehicle.

There was no difference between vehicle and XW014 treatment groups in plasma insulin levels collected 30 min post glucose challenge (data not shown).

XW014 reduces blood glucose and increases insulin in cynomolgus monkeys

Compared to baseline IVGTT, treatment of cynomolgus monkeys with a single dose of XW014 at 10, 20, or 40 mg/kg PO suppressed plasma glucose ($AUC_{0-60min}$) and increased the glucose disposal rate ($Kd_{3-40min}$).

Plasma glucose $AUC_{(0-60min)}$

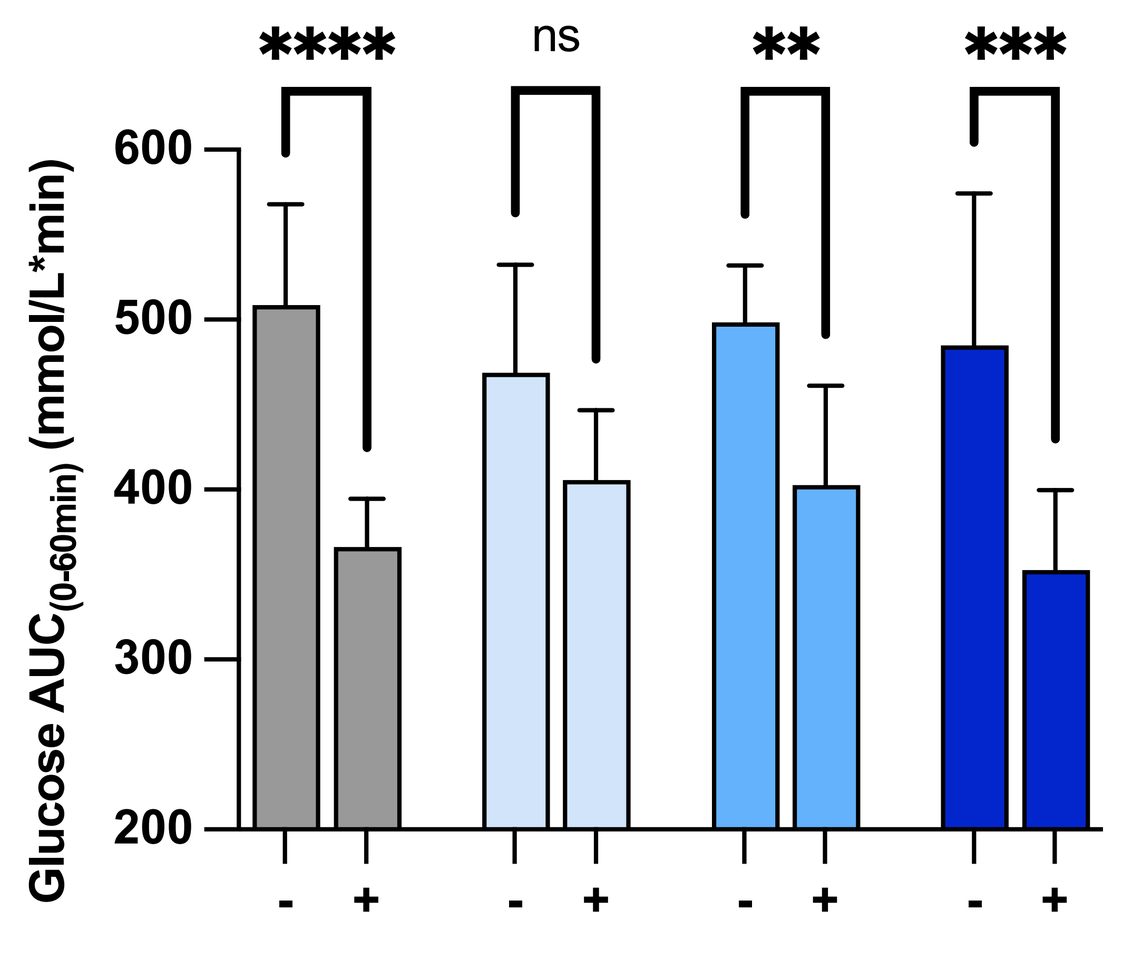
Glucose $AUC_{(0-60min)}$ (mmol/L*min)

ns
**

Glucose disposal rate ($Kd_{3-40min}$)

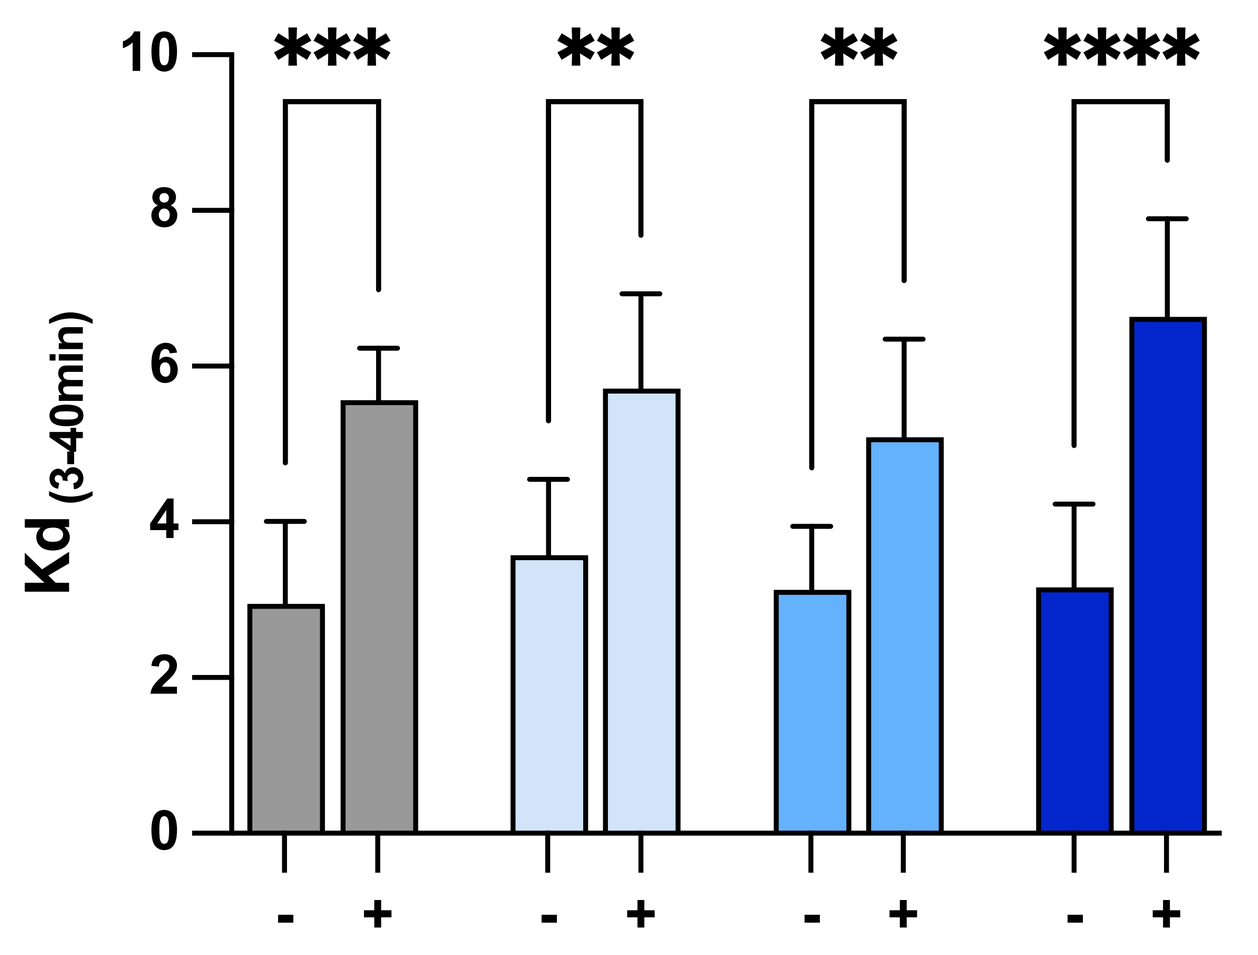
$Kd_{(3-40min)}$

**
**

Insulin $AUC_{(0-60min)}$

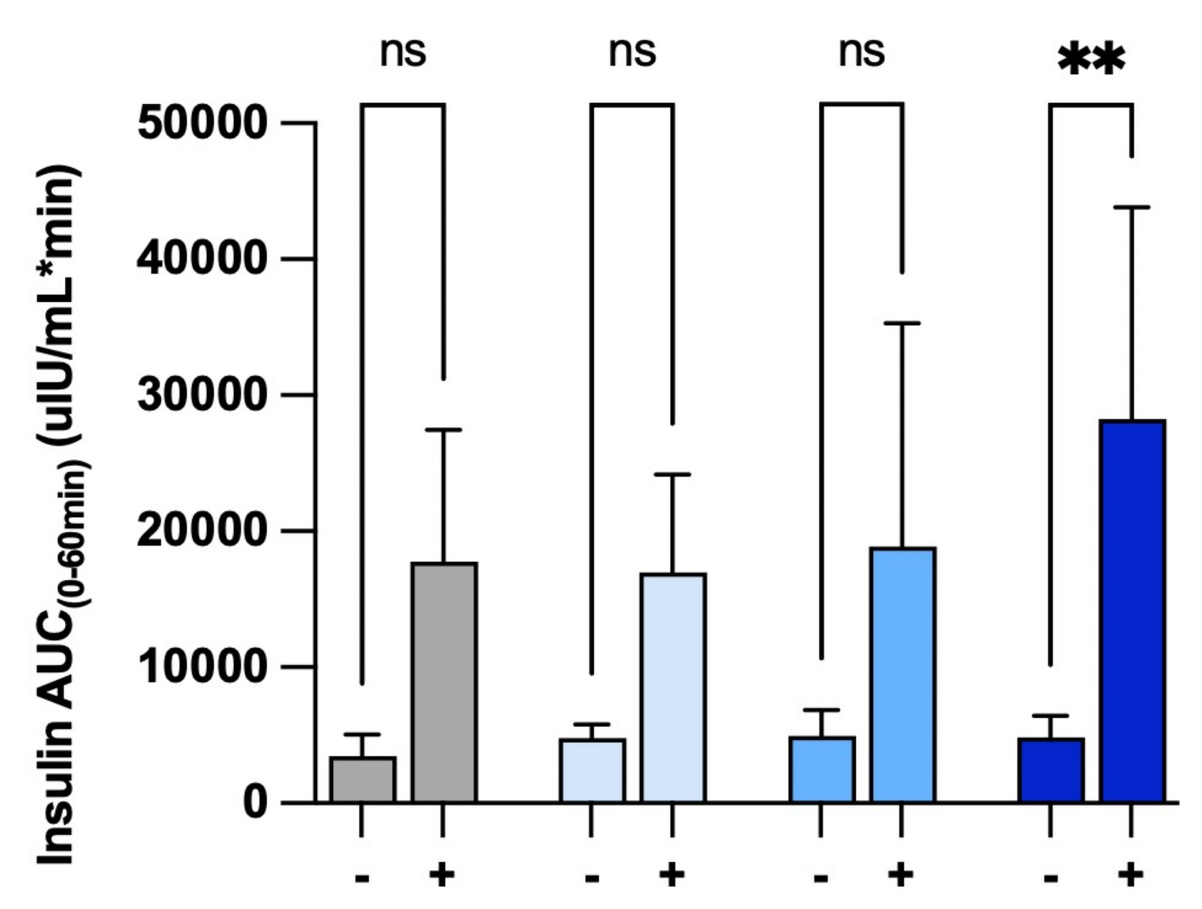
Insulin $AUC_{(0-60min)}$ (uIU/mL*min)
ns
ns
ns
**

2 mg/kg IV Danuglipron
10 mg/kg PO XW014
20 mg/kg PO XW014
40 mg/kg PO XW014

IVGTT response at baseline (-) and post-treatment (+) are shown for each parameter. Data are presented as mean and SD. Two-way ANOVA analysis was used to evaluate significance: ns, not significant, *p<0.05, **p<0.01, ***p<0.001, ****p<0.0001. n=5 animals per group.

A single dose of XW014 40 mg/kg PO significantly increased insulin secretion ($AUC_{0-60min}$) after glucose challenge compared to baseline.

XW014 leads to weight loss and glycemic control in a humanized GLP-1R obese mouse model

In C57BL/6 diet-induced obesity (DIO) mice expressing the humanized GLP-1R, treatment with XW014 (50 mg/kg BID) for 4 days resulted in reduced blood glucose, food intake, and body weight (data not shown). Following these preliminary results, we extended the study to 15 days of dosing with XW014 (30 or 60 mg/kg) or vehicle PO BID.

Multiple dose treatment with XW014 was well tolerated, and all mice survived to the end of study. Significant body weight loss and reduced food intake were observed at doses of 30 and 60 mg/kg BID XW014 compared to vehicle.

Reductions in post-prandial plasma glucose and improved glycemic control following oral glucose challenge were also seen in mice treated with XW014 compared to vehicle.

XW014 treatment in a hGLP-1R DIO mouse model

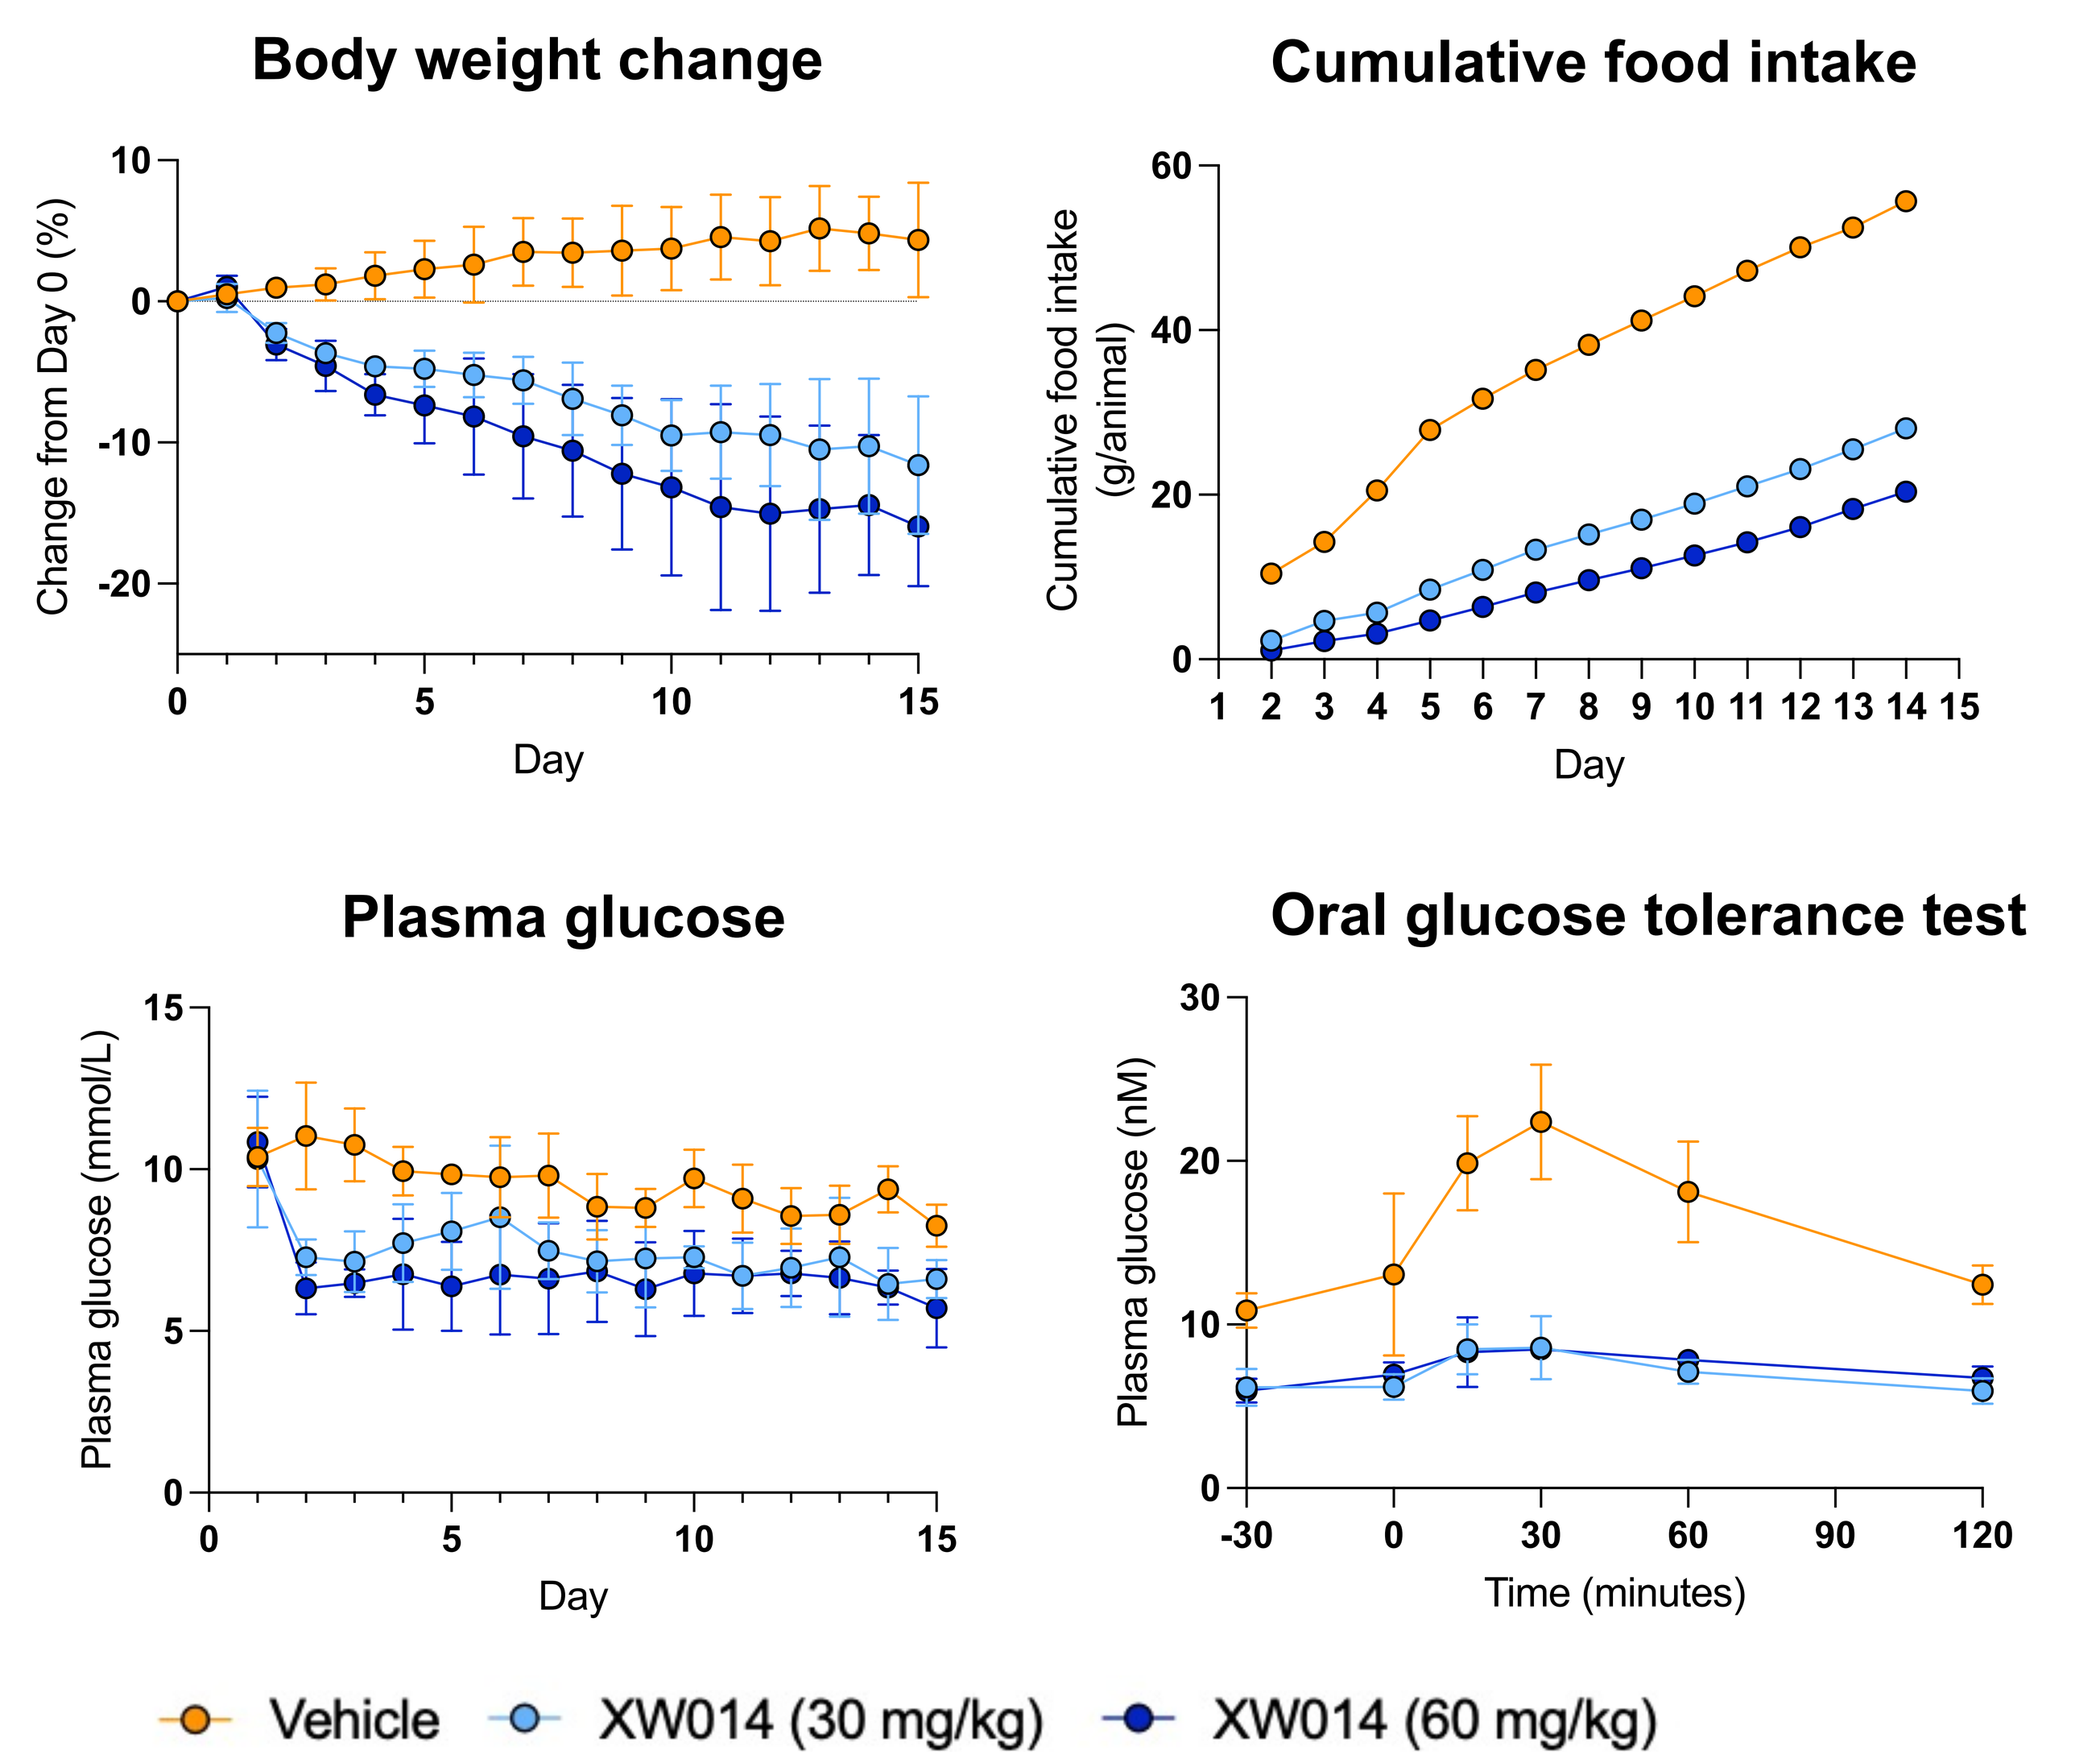
Body weight change
Change from Day 0 (%)
Day
Cumulative food intake
Cumulative food intake (g/animal)
Day
Plasma glucose
Plasma glucose (mmol/L)
Day
Oral glucose tolerance test
Plasma glucose (nM)
Time (minutes)
Vehicle
XW014 (30 mg/kg)
XW014 (60 mg/kg)

Data are presented as mean and SD. n=4 animals in the vehicle group and n=5 animals per treatment group. For the oral glucose tolerance test, test compound or vehicle were administered at T = -30 min and glucose was administered PO at T = 0 min.

CONCLUSIONS

XW014 is a potent small molecule agonist of the GLP-1 receptor *in vitro* and induces effective target engagement in two preclinical models, humanized GLP-1R mice and cynomolgus monkeys. The results support the continued development of XW014 for the treatment of metabolic disease.

REFERENCES

1. **Drucker DJ**. Mechanisms of Action and Therapeutic Application of Glucagon-like Peptide-1. *Cell Metabolism* 2018; 27: 740-756

2. **Saxena AR et al.** Danuglipron (PF-06882961) in type 2 diabetes: a randomized, placebo-controlled, multiple ascending-dose phase 1 trial. *Nat Med* 2021;27:1079-87.

3. **Zhang X et al.** Differential GLP-1R Binding and Activation by Peptide and Non-peptide Agonists. Molecular Cell 2020; 80: 485-500.E7

CONTACT INFORMATION

martijn.fenaux@sciwindbio.com